ARC-4: Efficacy and Safety of Etrumadenant (AB928) plus Carboplatin, Pemetrexed and a PD-1 Antibody in Participants with Metastatic Non-Small Cell Lung Cancer

Poster # 1309P

A Spira[1], P Conkling[2], A Chaudhry[3], DE Colburn[4], HN Gilbert[4], M Scharville[4], OS Gardner[4], K Krishnan[4], MC Paoloni[4], and M Johnson[5]

[1]US Oncology, Fairfax, VA; [2]US Oncology, Norfolk, VA; [3]Medical Oncology Associates, Spokane, WA; [4]Arcus Biosciences, Inc., Hayward, CA; [5]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN

INTRODUCTION

The Adenosine Axis in Cancer

- Dying tumor cells release high levels of ATP into the tumor microenvironment (TME) where CD39 and CD73 convert it to adenosine[1,2] (**Figure 1**)
- By binding adenosine receptors 2a and 2b ($A_{2a}R$ and $A_{2b}R$) expressed on immune cells, adenosine promotes immunosuppression by inhibiting critical components of the antitumor immune response, ultimately enabling tumors to evade destruction[2]
- Additionally, $A_{2a}R$ signaling impairs the activation, proliferation, and cytotoxic activity of effector T cells[3]
- Initial research focused on $A_{2a}R$ as the most relevant adenosine receptor in cancer physiology; however, $A_{2b}R$ signaling mediates unique functions, such as dendritic cell activation and function[4]
- Thus, adenosine receptor blockade may be necessary to overcome adenosine-dependent immunosuppression and lead to enhanced therapeutic efficacy of some chemotherapeutic agents

Figure 1. Critical Role of Adenosine Pathway in Immunosuppressive Tumor Microenvironment

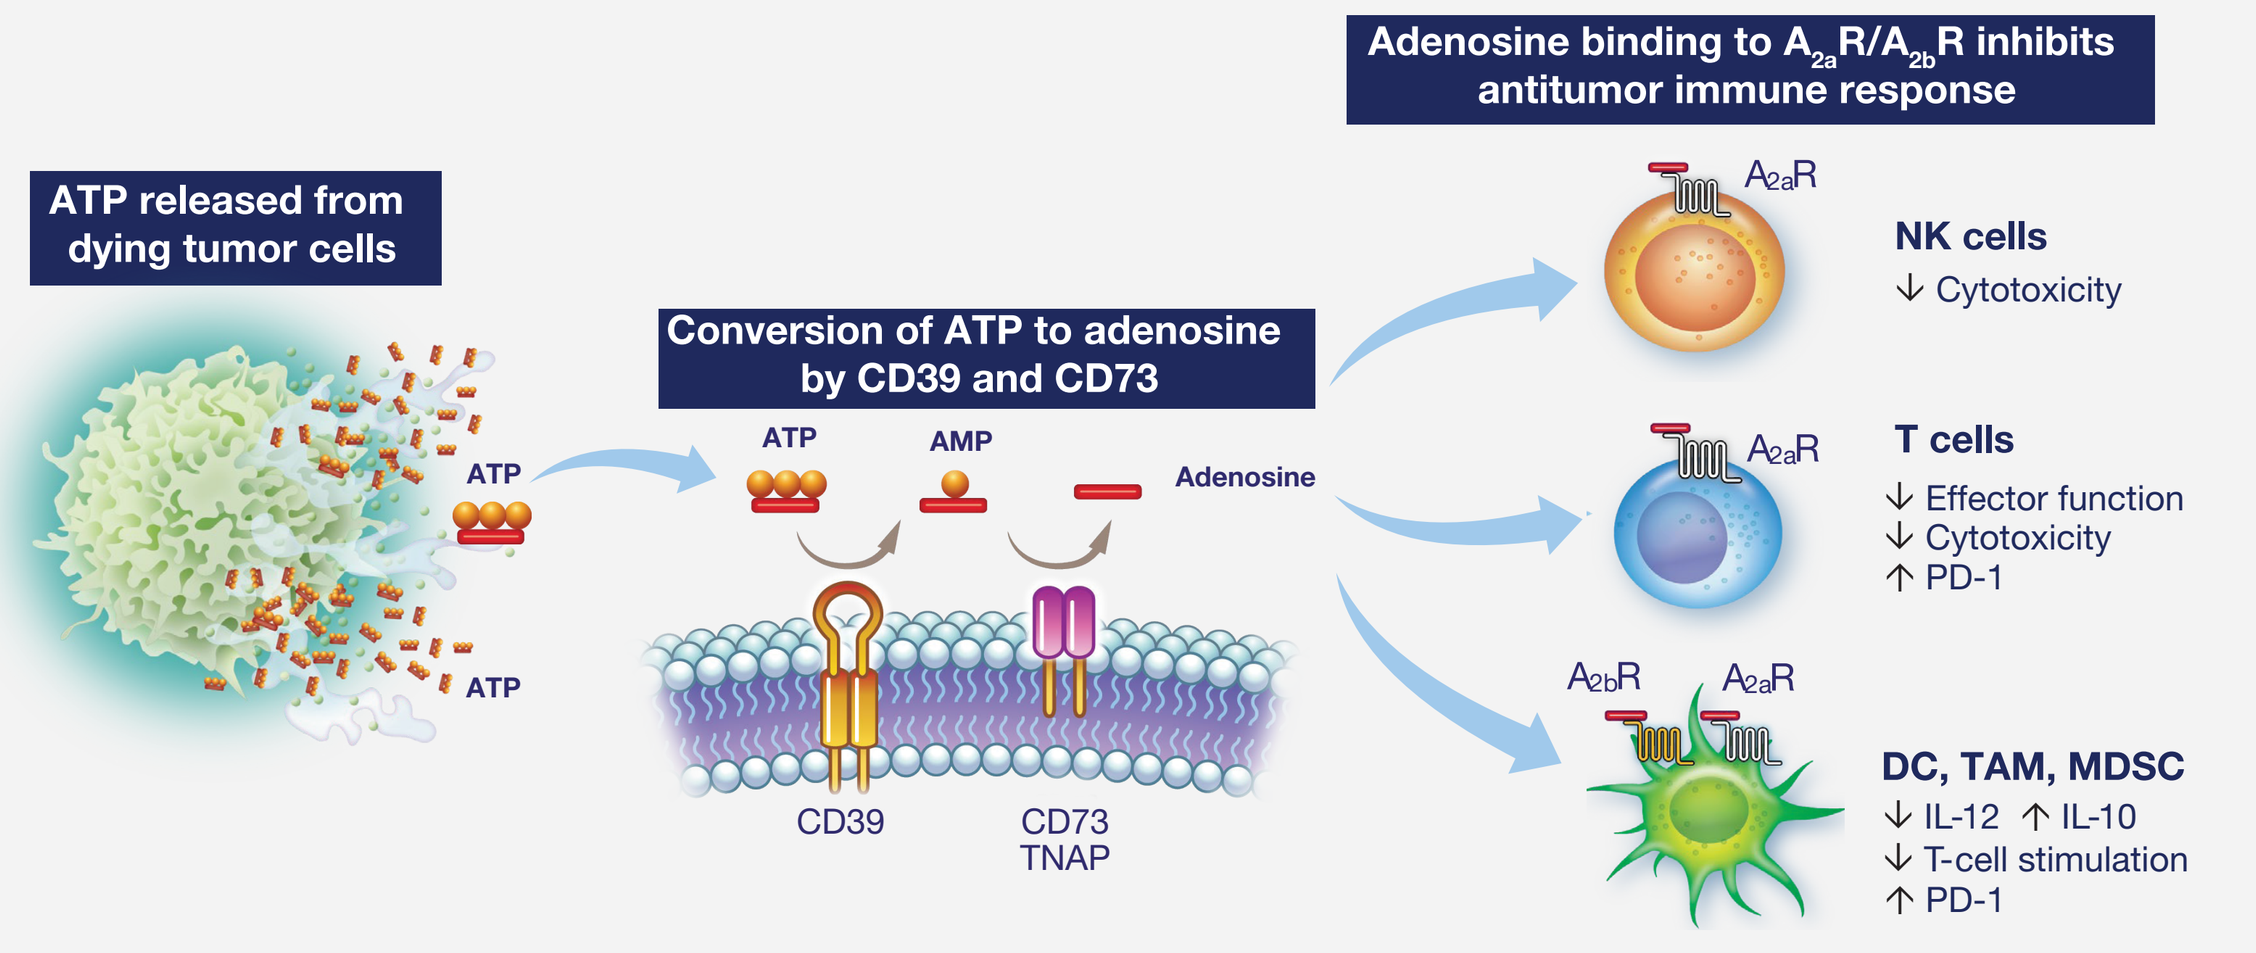
ATP released from dying tumor cells
ATP
ATP
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
TNAP
Adenosine binding to A2aR/A2bR inhibits antitumor immune response
A2aR
NK cells
↓ Cytotoxicity
A2aR
T cells
↓ Effector function
↓ Cytotoxicity
↑ PD-1
A2bR
A2aR
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation
↑ PD-1

AMP, adenosine monophosphate; ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; IL, interleukin; MDSC, myeloid-derived suppressor cell; NK, natural killer; PD-1, programmed cell death protein-1; TAM, tumor-associated macrophage; TNAP, tissue nonspecific alkaline phosphatase.

- Etrumadenant (AB928) is an orally bioavailable, small-molecule, selective dual antagonist of $A_{2a}R$ and $A_{2b}R$ that was specifically designed to block the immunosuppressive effects associated with high adenosine concentration within the TME; it is the only adenosine receptor antagonist in active clinical trials that potently blocks $A_{2b}R$
- Currently, there are 4 ongoing global phase 1/1b disease-specific platform studies to assess safety, tolerability, pharmacokinetics (PK), pharmacodynamics, and preliminary clinical activity of etrumadenant in combination with chemotherapy and/or anti–PD-1 antibody[5]
 - Based on dose escalation data from these studies, etrumadenant 150 mg once daily (QD) was selected as the recommended dose for expansion (RDE) based on PK, PK/pharmacodynamics correlation, and a well-tolerated safety profile of etrumadenant + chemo/immunotherapy

ARC-4 Study Rationale

- For locally advanced or metastatic non-small cell lung cancer (NSCLC), first-line treatment can include platinum-containing chemotherapy. Median overall survival and 5-year survival rates associated with these regimens are low[6]
- For non-squamous NSCLC with sensitizing mutations in epidermal growth factor receptor (*EGFR*) tyrosine kinase inhibitors (TKIs) are commonly used as first-line therapy[7]; however, only a subset of tumors have these mutations and development of TKI-resistance is common[8]
- Addition of PD-1/PD-L1-targeted immunotherapy to platinum chemotherapy has improved outcomes for some patients with NSCLC[9,10]; however, many either do not initially respond or responses are short-lived, so an urgent unmet need remains for these patients
- Combination therapy that includes antagonism of the adenosine and PD-1/PD-L1 pathways with platinum chemotherapy may hold promise for enhancing treatment efficacy without additional toxicity
 - Lung adenocarcinomas have high expression of $A_{2b}R$, CD73, and PD-1, suggesting that the adenosine and PD-1 pathways may be particularly important for tumor growth and persistence[11,12]
 - Additionally, tumors with mutations in *EGFR*, *KRAS*, and *BRAF* have higher CD73 expression compared with tumors without those mutations (**Figure 2**)
 - In patients with NSCLC, the adenosine pathway is a potential mechanism of resistance to anti–PD-1 therapy[13,14] and high tumor expression of CD73 is associated with poor prognosis[15], which may indicate a therapeutic advantage to combination $A_{2a}R/A_{2b}R$ and PD-1 blockade
 - In mice, etrumadenant monotherapy had minimal effects on tumor growth, but enhanced the antitumor efficacy of an anti–PD-L1 antibody or chemotherapy without additive toxicity (data not shown)

Figure 2. Correlation of *EGFR*, *KRAS*, and *BRAF* Mutations with Elevated CD73 Expression

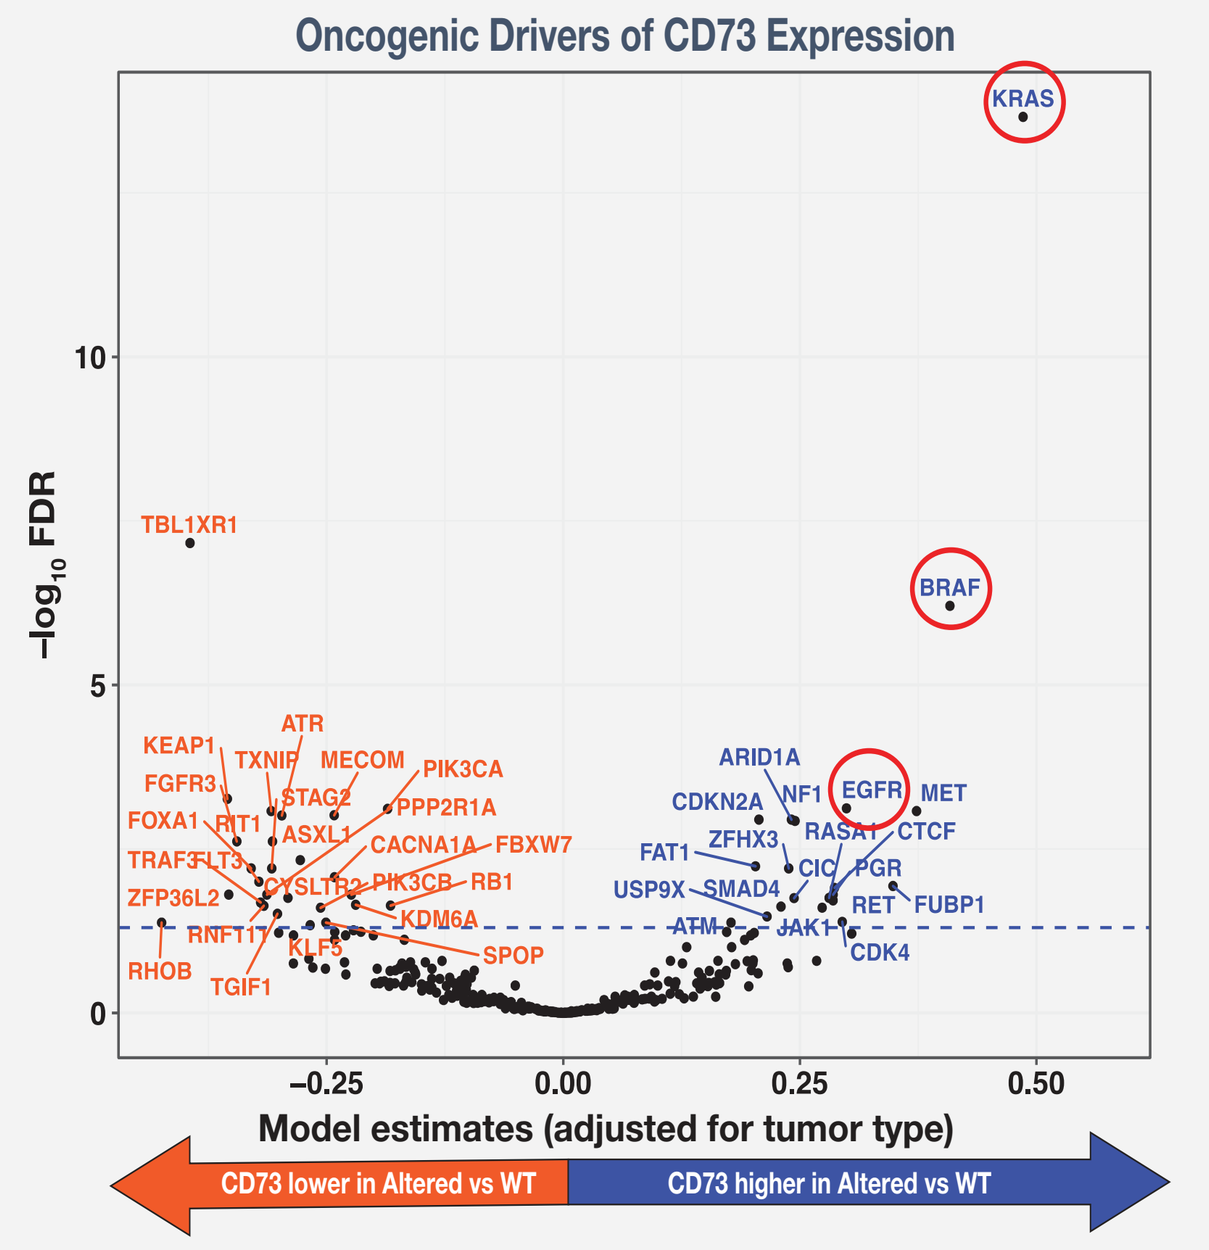
Oncogenic Drivers of CD73 Expression
–log10 FDR
KRAS
BRAF
EGFR
TBL1XR1
Model estimates (adjusted for tumor type)
CD73 lower in Altered vs WT
CD73 higher in Altered vs WT

FDR, false discovery rate; WT, wild type.

METHODS

Study Design

- ARC-4 is an ongoing phase 1/1b, multicenter, open-label, dose escalation and dose expansion study to evaluate safety, tolerability, PK, pharmacodynamics, and clinical activity of etrumadenant + carboplatin/pemetrexed + pembrolizumab or zimberelimab (anti–PD-1 antibody) in patients with locally advanced, metastatic, or recurrent NSCLC (**Figure 3**)
 - **Dose escalation:** Etrumadenant (75 or 150 mg) orally (PO) once daily (QD) was administered with standard intravenous (IV) doses of carboplatin (AUC 5 mg/mL/min), pemetrexed (500 mg/m²), and pembrolizumab (200 mg) once every 3 weeks (Q3W) based on a 3+3 design
 - **Dose expansion:** Patients with sensitizing *EGFR* mutations were enrolled and received etrumadenant (150 mg PO QD) with standard IV doses of carboplatin, pemetrexed, and zimberelimab (360 mg Q3W)

Figure 3. ARC-4 Study Design

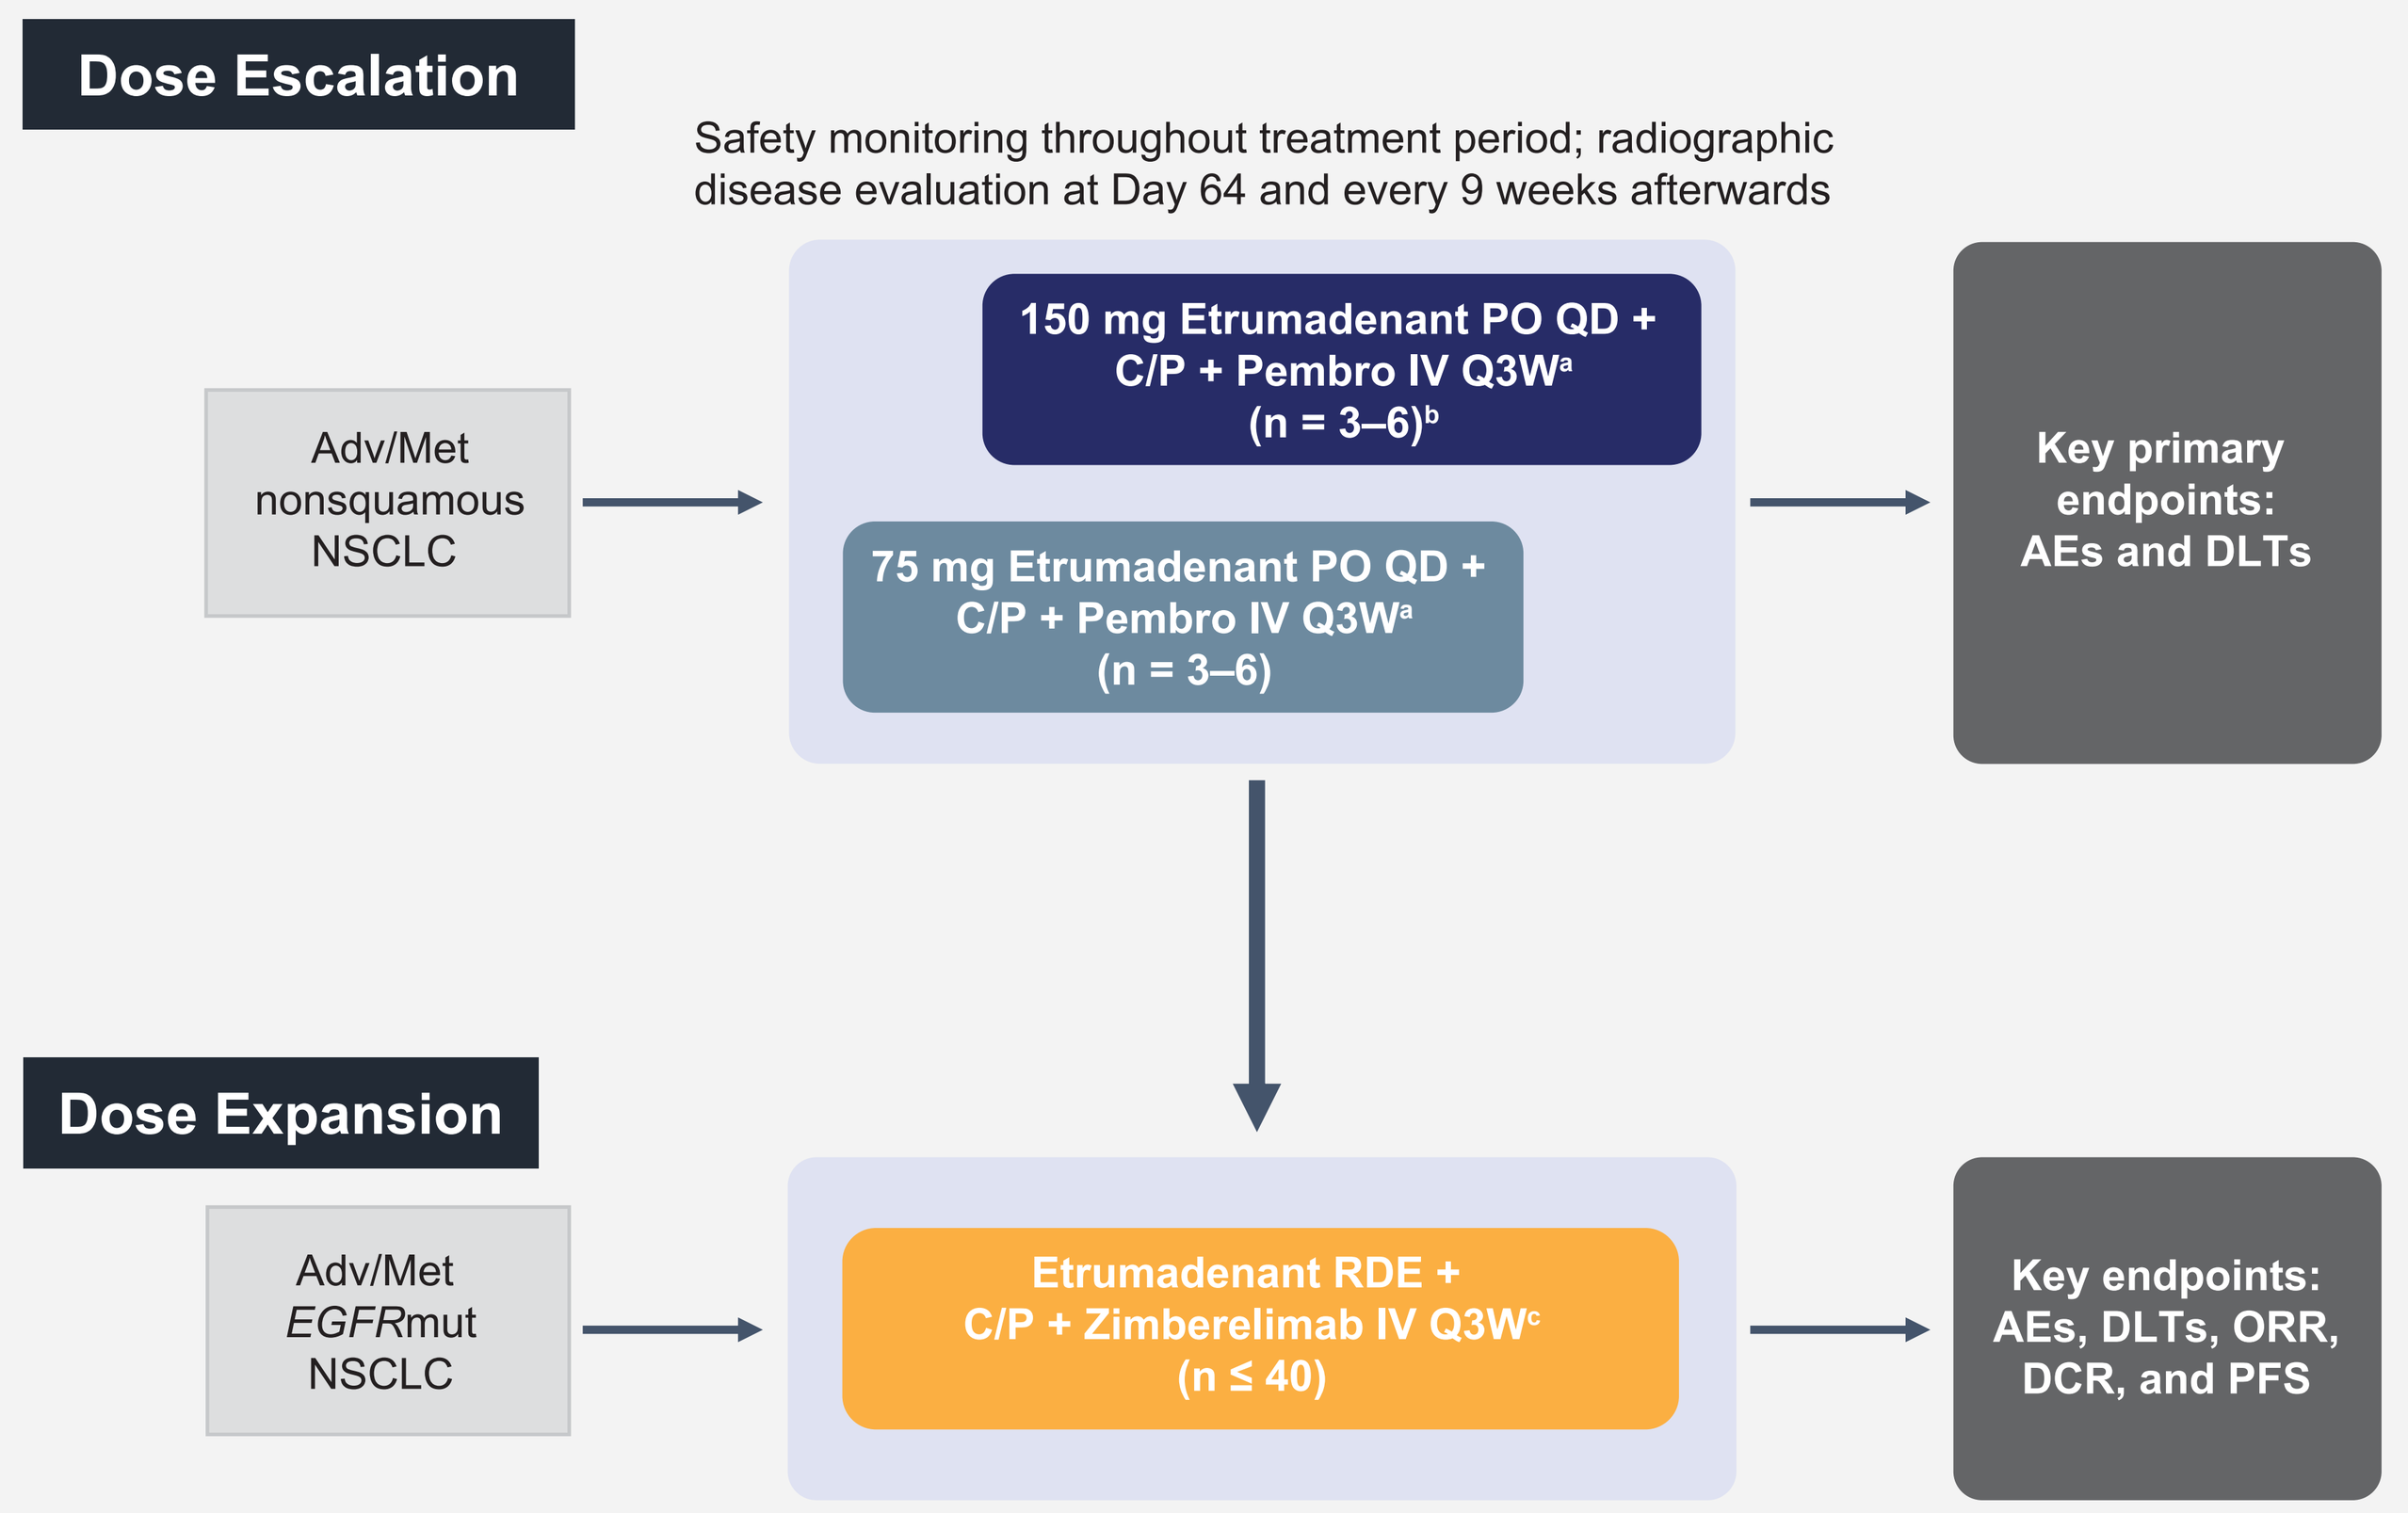
Dose Escalation
Safety monitoring throughout treatment period; radiographic disease evaluation at Day 64 and every 9 weeks afterwards
Adv/Met nonsquamous NSCLC
150 mg Etrumadenant PO QD + C/P + Pembro IV Q3Wa (n = 3–6)b
75 mg Etrumadenant PO QD + C/P + Pembro IV Q3Wa (n = 3–6)
Key primary endpoints: AEs and DLTs
Dose Expansion
Adv/Met EGFRmut NSCLC
Etrumadenant RDE + C/P + Zimberelimab IV Q3Wc (n ≤ 40)
Key endpoints: AEs, DLTs, ORR, DCR, and PFS

[a] Carboplatin AUC 5 mg/mL/min, pemetrexed 500 mg/m², pembrolizumab 200 mg IV Q3W; [b] When a minimum of 3 patients available for toxicity completed the DLT evaluation period (1 chemotherapy cycle) for a given etrumadenant dose, subsequent patients were enrolled at the higher etrumadenant dose; [c] Zimberelimab 360 mg. Adv, advanced; AE, adverse event; AUC, area under the curve; C, carboplatin; DLT, dose-limiting toxicity; *EGFR*, epidermal growth factor receptor; EOT, end of treatment; IV, intravenously; Met, metastatic; mut, mutated; NSCLC, non-small cell lung cancer; P, pemetrexed; Pembro, pembrolizumab; PO, orally; Q3W, every 3 weeks; QD, once daily.

ARC-4 (NCT03846310) Design Features

- The primary objective is to evaluate the safety and tolerability of etrumadenant combination therapy in patients with NSCLC; secondary objectives include determination of clinical activity and PK
- Eligible patients are adults with pathologically-confirmed non-squamous NSCLC that is locally advanced, metastatic, or recurrent with progression, at least 1 measurable lesion per RECIST v1.1, and ECOG performance status of 0 or 1
- For the dose escalation, one of the following criteria must have also be met: 1) no alternative or curative therapy exists; 2) tumor had a genetic mutation/rearrangement for which targeted therapy exists but has no available standard TKI **and** patient has not had prior chemotherapy or anti–PD-1/PD-L1-based therapy; 3) patients were treatment-naive or were considered appropriate study candidates by the principal investigator
- For the dose expansion, patients must have had a sensitizing *EGFR* mutation and have had disease progression or been intolerant to ≥ 1 approved TKI; they must also have had no prior chemotherapy or anti–PD-1/PD-L1-based therapy for locally advanced or metastatic disease
- Study treatment may continue until disease progression, unacceptable toxicity, consent withdrawal, or by the investigator's decision

Statistical Analysis

- Safety analyses included all patients who received ≥ 1 etrumadenant dose; summary statistics are shown for treatment-emergent adverse events (TEAEs), serious TEAEs (TESAEs), TEAE severity and relationship to study drugs
- The efficacy-evaluable population included all patients who received ≥ 1 dose of etrumadenant and had ≥ 1 post-baseline disease assessment; clinical activity was assessed according to RECIST v1.1 criteria

RESULTS

Patient Baseline Characteristics

- As of August 5, 2020, 14 patients have received etrumadenant + chemo/immunotherapy; all but 3 received 150 mg etrumadenant (**Table 1**)
- Four patients in the dose escalation were previously treated with a checkpoint inhibitor (CPI)
- Within study patient medical records, the following genetic data were available:
 - Among the tumors of the 7 patients treated in the dose escalation, 1 had an *EGFR* mutation, 1 had a *BRAF* mutation, and 1 had a *KRAS* mutation
 - All 7 patients treated in the dose expansion had tumors with *EGFR* mutations

RESULTS

Table 1. Baseline Patient Demographics and Characteristics

Parameter	Dose Escalation: 75 mg Etrumadenant QD + C/P + Pembro Q3W n = 3	Dose Escalation: 150 mg Etrumadenant QD + C/P + Pembro Q3W n = 4	Dose Expansion: 150 mg Etrumadenant QD + C/P + Zim Q3W n = 7	Total N = 14
Median age (range), years	62 (50-80)	65.5 (48-74)	68 (54-77)	66 (48-80)
Male, n (%)	2 (67)	1 (25)	3 (43)	6 (43)
Race, n (%)				
White	3 (100)	4 (100)	2 (29)	9 (64)
Asian	0	0	4 (57)	4 (29)
Not reported	0	0	1 (14)	1 (7)
Prior therapies for metastatic disease, n (%)				
0	1 (33)	1 (25)	0	2 (14)
1	1 (33)	0	5 (71)	6 (43)
2	0	1 (25)	2 (29)	3 (21)
3	1 (33)	1 (25)	0	2 (14)
4	0	1 (25)	0	1 (7)
CPI-experienced patients, n (%)	1 (33)	3 (75)	0	4 (29)
EGFR mutation, n (%)	1 (33)	0	7 (100)	8 (57)
BRAF mutation[a], n (%)	0	1 (25)	0	1 (7)
RAS mutations (*KRAS* and *NRAS*)[b], n (%)	0	1 (25)	0	1 (7)

[a] 7 patients did not have available data; [b] 10 patients did not have available data. C, carboplatin; CPI, checkpoint inhibitor; *EGFR*, epidermal growth factor receptor; P, pemetrexed; Pembro, pembrolizumab; Q3W, once every three weeks; QD, once daily; Zim, zimerelimab.

Safety Analyses

- As of August 5, 2020, no dose-limiting toxicities had been reported in any dose cohort
- All patients reported ≥ 1 TEAE and 14 TEAEs were reported by ≥ 4 patients in any arm; the most common TEAEs were anemia (50%), neutrophil count decreased (43%), nausea (43%), and pyrexia (43%; **Table 2**)
- Etrumadenant-related TEAEs were reported by 8/14 (57%) patients and most were Grade 1 or 2
 - Two patients (150 mg etrumadent; 1 each in dose escalation and expansion) experienced Grade 3 events (platelet count decreased, white blood cell decreased, thrombocytopenia) or Grade 4 events (thrombocytopenia) that were also considered related to carboplatin/pemetrexed
- Of all patients, 3/14 (21%) experienced etrumadenant-related TESAEs; all were Grade 1 or 2 events, except for 1 event of Grade 4 thrombocytopenia that was also considered related to carboplatin/pemetrexed
- In total, 5/14 (36%) patients discontinued any study treatment due to TEAEs
- One patient in the 75 mg etrumadent dose escalation group died from respiratory failure 36 weeks after the start of study treatment; the death was attributed to disease progression and was not considered related to any study drug

Table 2. Treatment-Emergent Adverse Events

Patients, n (%)	Dose Escalation: 75 mg Etrumadenant QD + C/P + Pembro Q3W n = 3	Dose Escalation: 150 mg Etrumadenant QD + C/P + Pembro Q3W n = 4	Dose Expansion: 150 mg Etrumadenant QD + C/P + Zim Q3W n = 7	Total N = 14
Any TEAE	3 (100)	4 (100)	7 (100)	14 (100)
Grade ≥ 3	3 (100)	3 (75)	3 (43)	9 (64)
Any TESAE	1 (33)	2 (50)	3 (43)	6 (43)
Grade ≥ 3	1 (33)	2 (50)	1 (14)	4 (29)
Etrumadenant-related TEAEs	2 (67)	1 (25)	5 (71)	8 (57)
Etrumadenant-related TESAEs	0	1 (25)	2 (29)	3 (21)
Any study treatment d/c due to TEAEs	1 (33)	3 (75)	1 (14)	5 (36)
Deaths due to TEAEs	1 (33)	0	0	1 (7)
TEAEs in ≥ 4 patients, n (%)				
Anemia	3 (100)	4 (100)	0	7 (50)
Neutrophil count decreased	3 (100)	1 (25)	2 (29)	6 (43)
Nausea	2 (67)	2 (50)	2 (29)	6 (43)
Pyrexia	1 (33)	2 (50)	3 (43)	6 (43)
Rash maculopapular	2 (67)	1 (25)	2 (29)	5 (36)
Fatigue	1 (33)	2 (50)	2 (29)	5 (36)
Hypokalemia	0	2 (50)	2 (29)	4 (29)
Blood creatinine increased	1 (33)	1 (25)	2 (29)	4 (29)
ALT increased	2 (67)	1 (25)	1 (14)	4 (29)
AST increased	3 (100)	1 (25)	0	4 (29)
Dehydration	1 (33)	2 (50)	1 (14)	4 (29)
Dyspnea	3 (100)	0	1 (14)	4 (29)
Constipation	3 (100)	0	1 (14)	4 (29)
Dizziness	1 (33)	1 (25)	2 (29)	4 (29)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; C, carboplatin; d/c, discontinuation; P, pemetrexed; Pembro, pembrolizumab; Q3W, once every 3 weeks; QD, once daily; TEAE, treatment-emergent adverse event; TESAE, treatment-emergent serious adverse event; Zim, zimberelimab.

Clinical Activity

- As of August 5, 2020, there were 6/14 (43%) patients who remained on active treatment; the best overall responses for 13 evaluable patients are shown in **Figures 3 and 4**
- In the dose escalation:
 - Etrumadenant 75 mg – 3 partial responses (PRs)
 - Etrumadenant 150 mg – 2 stable disease (SD), 1 disease progression (PD; mixed response), 1 clinical progression
 - Notable responders included a TKI-experienced patient with an *EGFR*-mutated tumor that failed 3 prior lines of therapy and a CPI-experienced patient who had PD while on ipilimumab/nivolumab
- In the dose expansion:
 - Etrumadenant 150 mg – 1 PR (ongoing), 4 SD (all ongoing), 1 PD

RESULTS

Figure 3. Time on Treatment and RECIST v1.1 Response

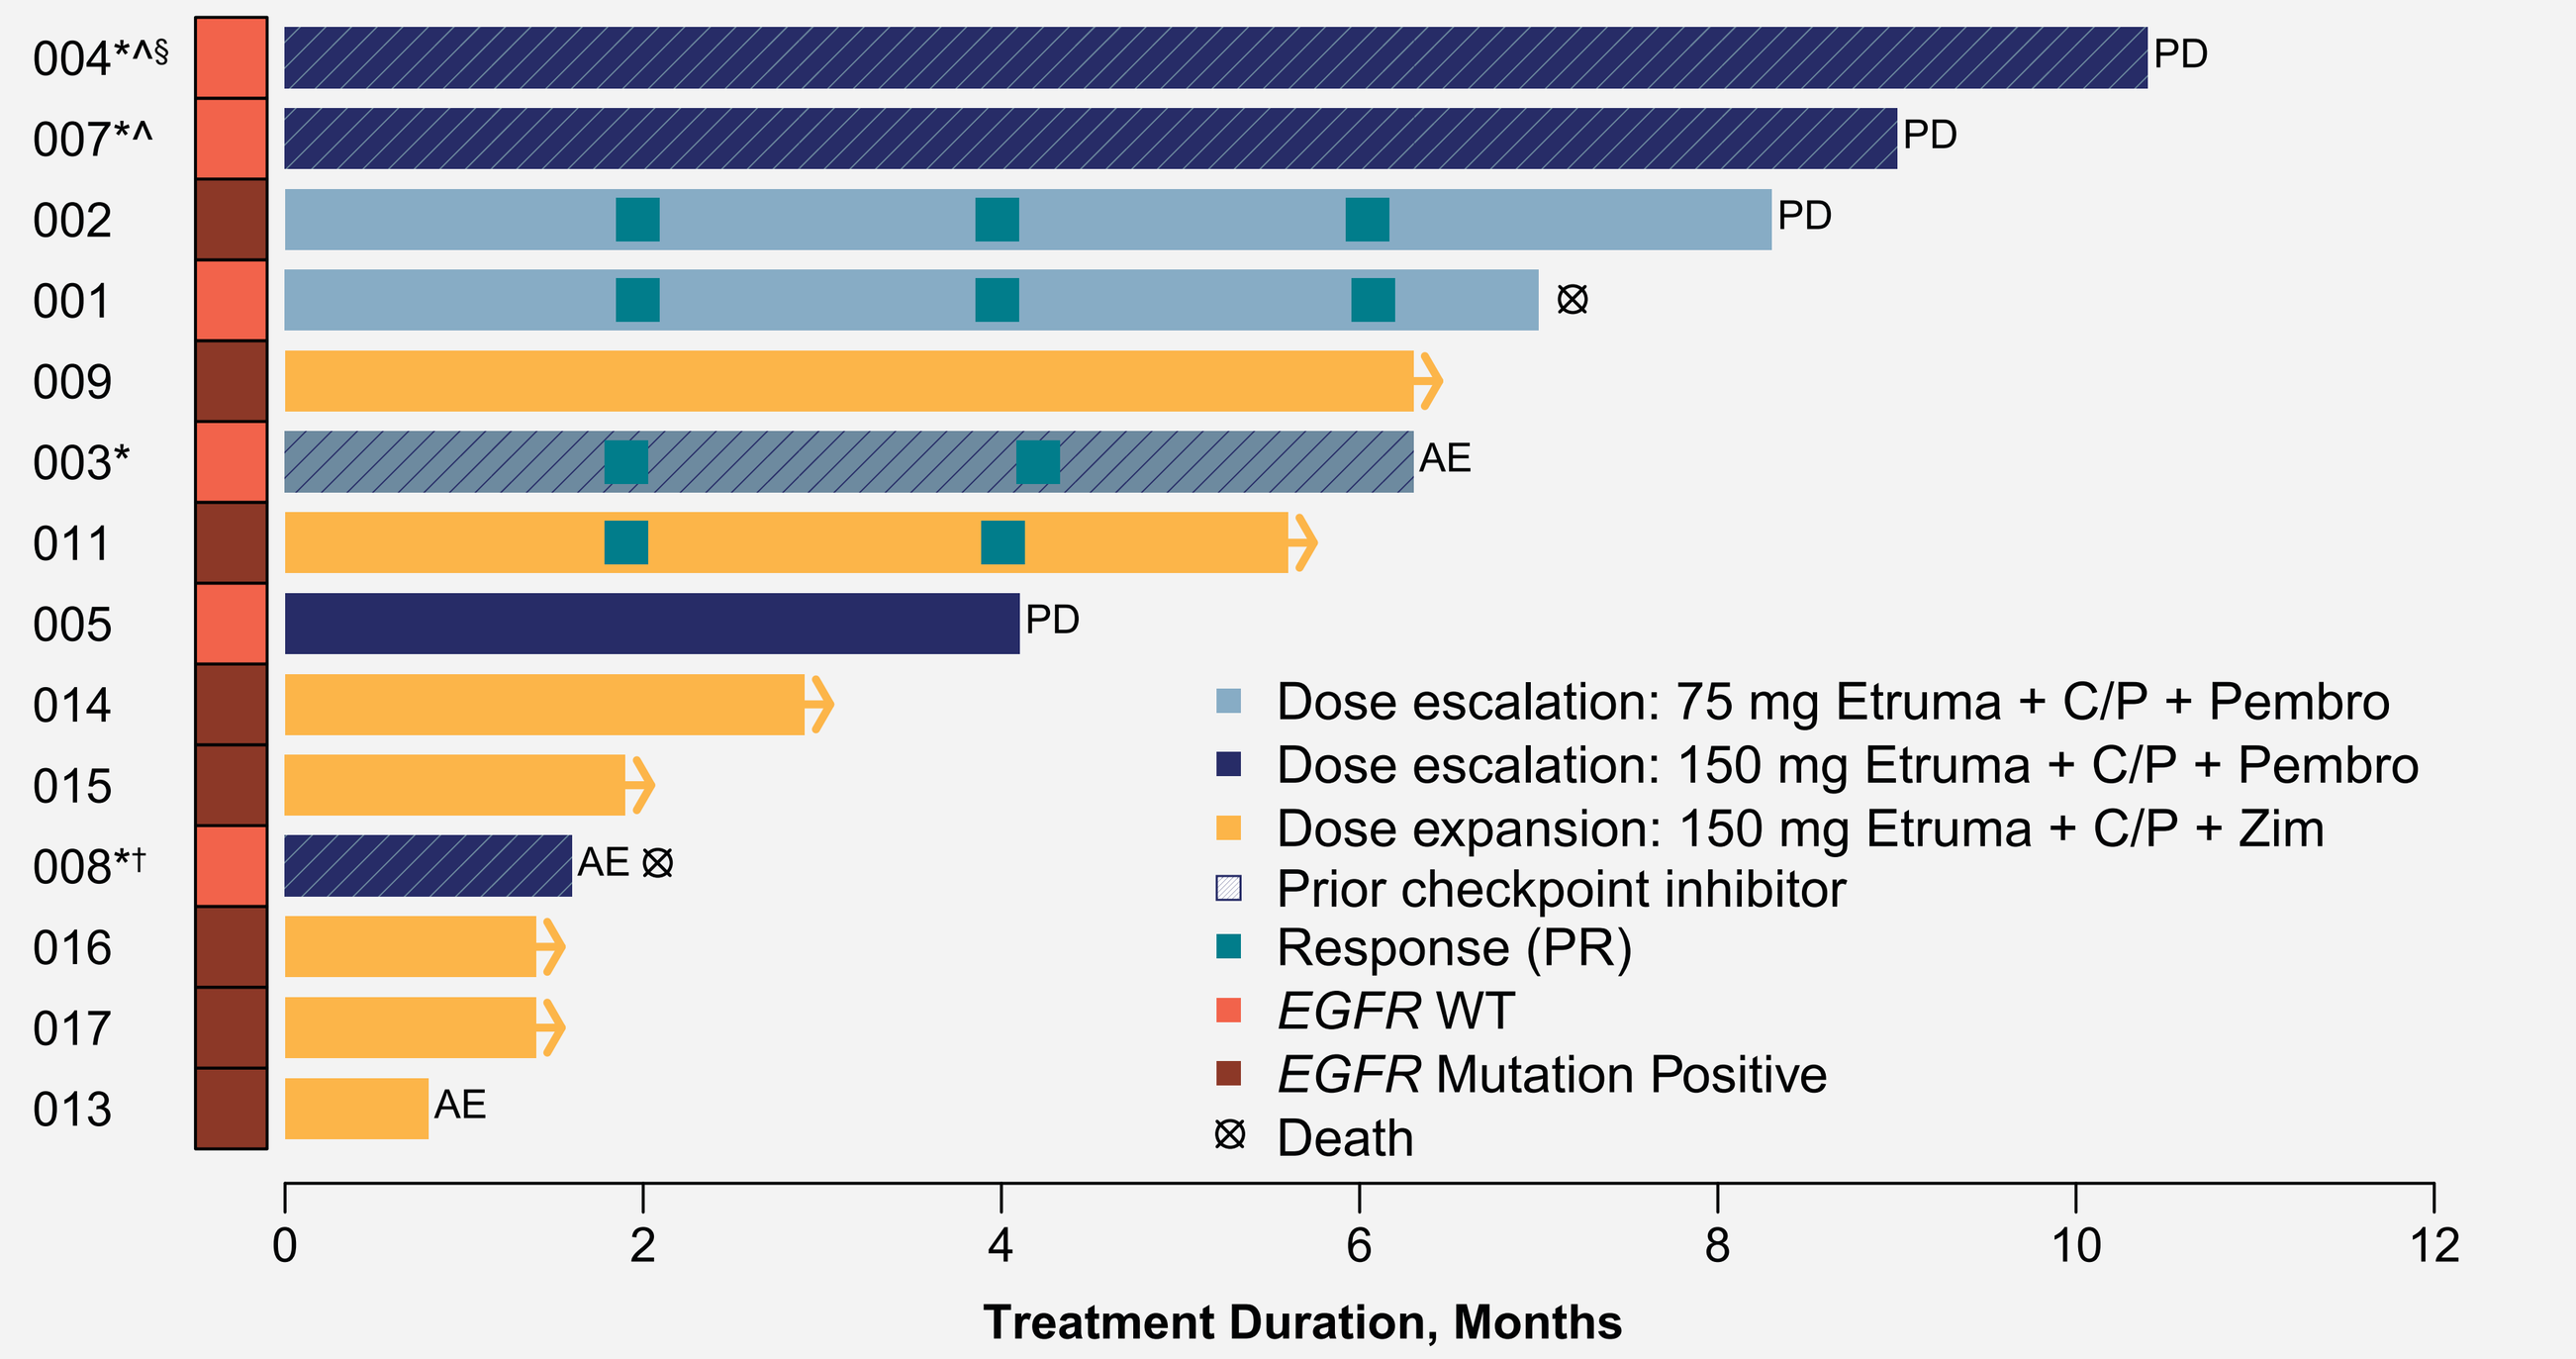
004*^§
007*^
002
001
009
003*
011
005
014
015
008*†
016
017
013
PD
PD
PD
AE
PD
AE
AE
Dose escalation: 75 mg Etruma + C/P + Pembro
Dose escalation: 150 mg Etruma + C/P + Pembro
Dose expansion: 150 mg Etruma + C/P + Zim
Prior checkpoint inhibitor
Response (PR)
EGFR WT
EGFR Mutation Positive
Death
0
2
4
6
8
10
12
Treatment Duration, Months

* Patients who were CPI-experienced; ^ Patient 004 had a *KRAS*-mutated tumor; patient 007 had a *BRAF*-mutated tumor; § Patient had a new non-target lesion at the first disease assessment; † Patient discontinued the study prior to the first disease assessment and was considered non-evaluable. AE, discontinuation due to adverse event; C, carboplatin; CPI, checkpoint inhibitor; EGFR, epidermal growth factor receptor; Etruma, etrumadenant; P, pemetrexed; PD, progressive disease; Pembro, pembrolizumab; PR, partial response; WT, wild type; Zim, zimberelimab.

Best Overall Response

Figure 4. Waterfall Plot of Best Percent Change from Baseline in Sum of Target Lesions

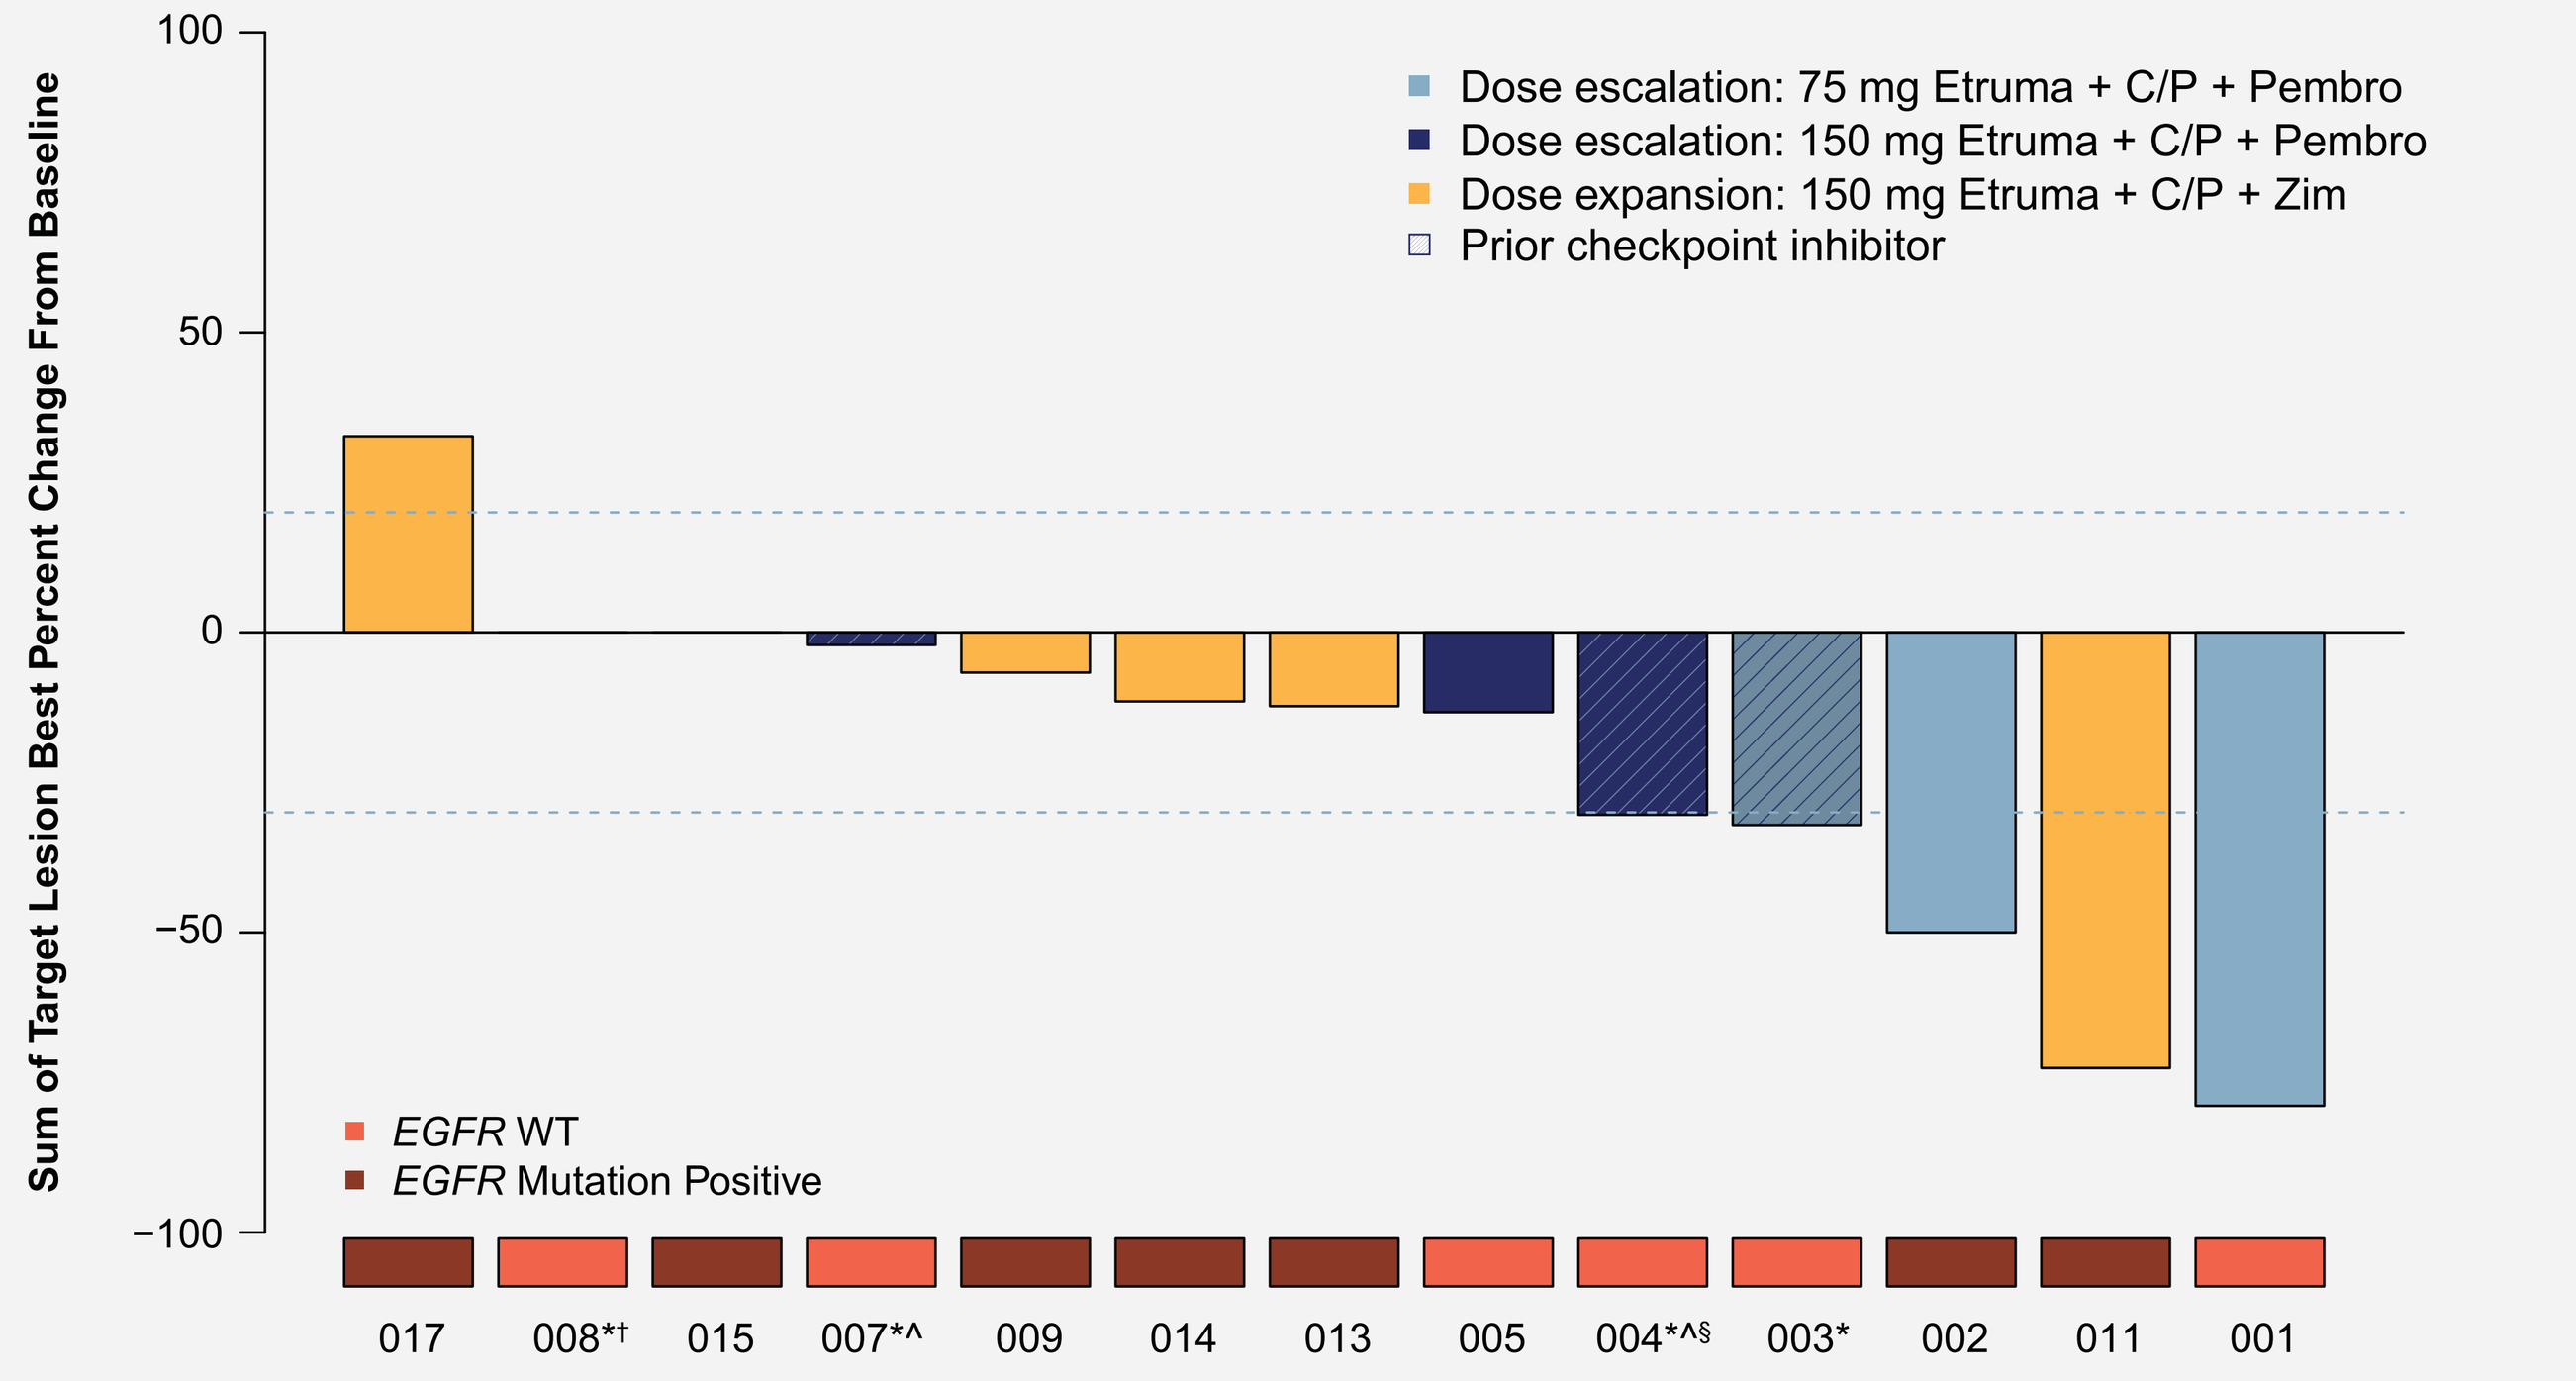
Sum of Target Lesion Best Percent Change From Baseline
100
50
0
–50
–100
Dose escalation: 75 mg Etruma + C/P + Pembro
Dose escalation: 150 mg Etruma + C/P + Pembro
Dose expansion: 150 mg Etruma + C/P + Zim
Prior checkpoint inhibitor
EGFR WT
EGFR Mutation Positive
017
008*†
015
007*^
009
014
013
005
004*^§
003*
002
011
001

* Patients who were CPI-experienced; ^ Patient 004 had a *KRAS*-mutated tumor; patient 007 had a *BRAF*-mutated tumor; § Patient had a new non-target lesion at the first disease assessment; † Patient discontinued the study prior to the first disease assessment and was considered non-evaluable. C, carboplatin; CPI, checkpoint inhibitor; EGFR, epidermal growth factor receptor; Etruma, etrumadenant; P, pemetrexed; Pembro, pembrolizumab; WT, wild type; Zim, zimberelimab.

CONCLUSIONS

- The combination of etrumadenant + chemotherapy + anti–PD-1 antibody had a manageable safety profile in patients with locally advanced, metastatic, or recurrent NSCLC; etrumadenant 150 mg PO QD was identified as the recommended dose for expansion
- Clinical activity with combination treatment was seen across multiple treatment cohorts, including responses in patients with *EGFR*-mutated tumors with recurrent disease after prior TKI or immunotherapy
- Dose expansion is ongoing in patients with *EGFR*-mutated NSCLC who have progressed after one or more TKIs with randomization triggered to begin post-futility assessment

ACKNOWLEDGMENTS AND DISCLOSURES

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation in this clinical trial. They would also like to thank the ARC-4 Principal Investigators and study staff for their efforts in carrying out the study. A Spira has received grants from EMD Serono and Turning Point Therapeutics; and has received grants and personal fees from Amgen, AstraZeneca, Bristol Myers Squibb, CytomX Therapeutics, Merck, and Novartis. Medical writing support was provided by Amanda Martin, PhD, of Medical Expressions (Chicago, IL) and was funded by Arcus Biosciences, Inc. This study was funded by Arcus Biosciences, Inc.

REFERENCES

1. Martins I, et al. *Cell Cycle*. 2009;8(22):3723-3728.
2. Vijayan D, et al. *Nat Rev Cancer*. 2017;17(12):709-724.
3. Cekic C, et al. *Nat Rev Immunol*. 2016;16(3):177-192.
4. Gao ZG, et al. *Int J Mol Sci*. 2019;20(20).
5. Powderly J, et al. *ESMO 2019*. Abstract 4854.
6. Zhang C, et al. *J Hematol Oncol*. 2019;12(1):45.
7. NCCN guidelines for NSCLC. Available at: https://www.nccn.org/professionals/physician_gls/pdf/nscl.pdf.
8. Yuan M, et al. *Signal Transduct Target Ther*. 2019;4:61.
9. Reck M, et al. *N Engl J Med*. 2016;375(19):1823-1833.
10. Reck M, et al. *J Clin Oncol*. 2019;37(7):537-546.
11. DiRenzo D, et al. *SITC 2019*. Poster P557.
12. Anderson AE, et al. *SITC 2019*. Poster P260.
13. Fong P, et al. *ASCO 2019*. Abstract 5010.
14. Giannakis M. *ASCO 2017*. Abstract 3004.
15. Inoue Y, et al. *Oncotarget*. 2017;8(5):8738-8751.

Corresponding Author: Dawn Colburn (dcolburn@arcus.com)

ESMO Virtual Congress 2020