ARC-7: A Phase 2 Study to Evaluate the Safety and Efficacy of Zimberelimab Monotherapy and Domvanalimab (AB154) + Zimberelimab ± Etrumadenant (AB928) in Front-Line, PD-L1-High Expressing, Non-Small Cell Lung Cancer

Poster # 1419TiP

A Chaudhry[1], M Johnson[2], D Colburn[3], HN Gilbert[3], J Coffie[3], A Garofalo[3], M Paoloni[3], K Krishnan[3]

[1]Medical Oncology Associates, Spokane, WA; [2]Sarah Cannon Research Institute, Nashville, TN; [3]Arcus Biosciences, Inc., Hayward, CA

CHECKPOINT INHIBITION AND THE TIGIT PATHWAY

- The programmed cell death protein-1 (PD-1) inhibitory pathway is a key immune checkpoint that can be exploited by tumors to enable their survival[1]
 - Zimberelimab (AB122), a monoclonal antibody (mAb) in early clinical development, potently blocks PD-1 and has an anticipated safety profile similar to other approved anti–PD-1 mAbs
- The T cell immunoreceptor with Ig and ITIM domain (TIGIT) inhibitory pathway has been previously identified as a novel immune checkpoint that influences the antitumor immune response[2] (**Figure 1**)
 - TIGIT, expressed on T cells and NK cells, binds its tumor cell-expressed ligand CD155 and as a result, immune cell effector function is abrogated[3]
 - CD155 has a greater affinity for TIGIT, but can also bind the receptor DNAX accessory molecule-1 (DNAM-1); if this occurs, the resulting signaling leads to proinflammatory cytokine production and immune cell cytotoxicity[3]
 - Thus, inhibition of TIGIT-CD155 binding promotes activation of the immune response as opposed to suppression in the tumor microenvironment (TME)

Figure 1. TIGIT-CD155 Binding Promotes Immunosuppression

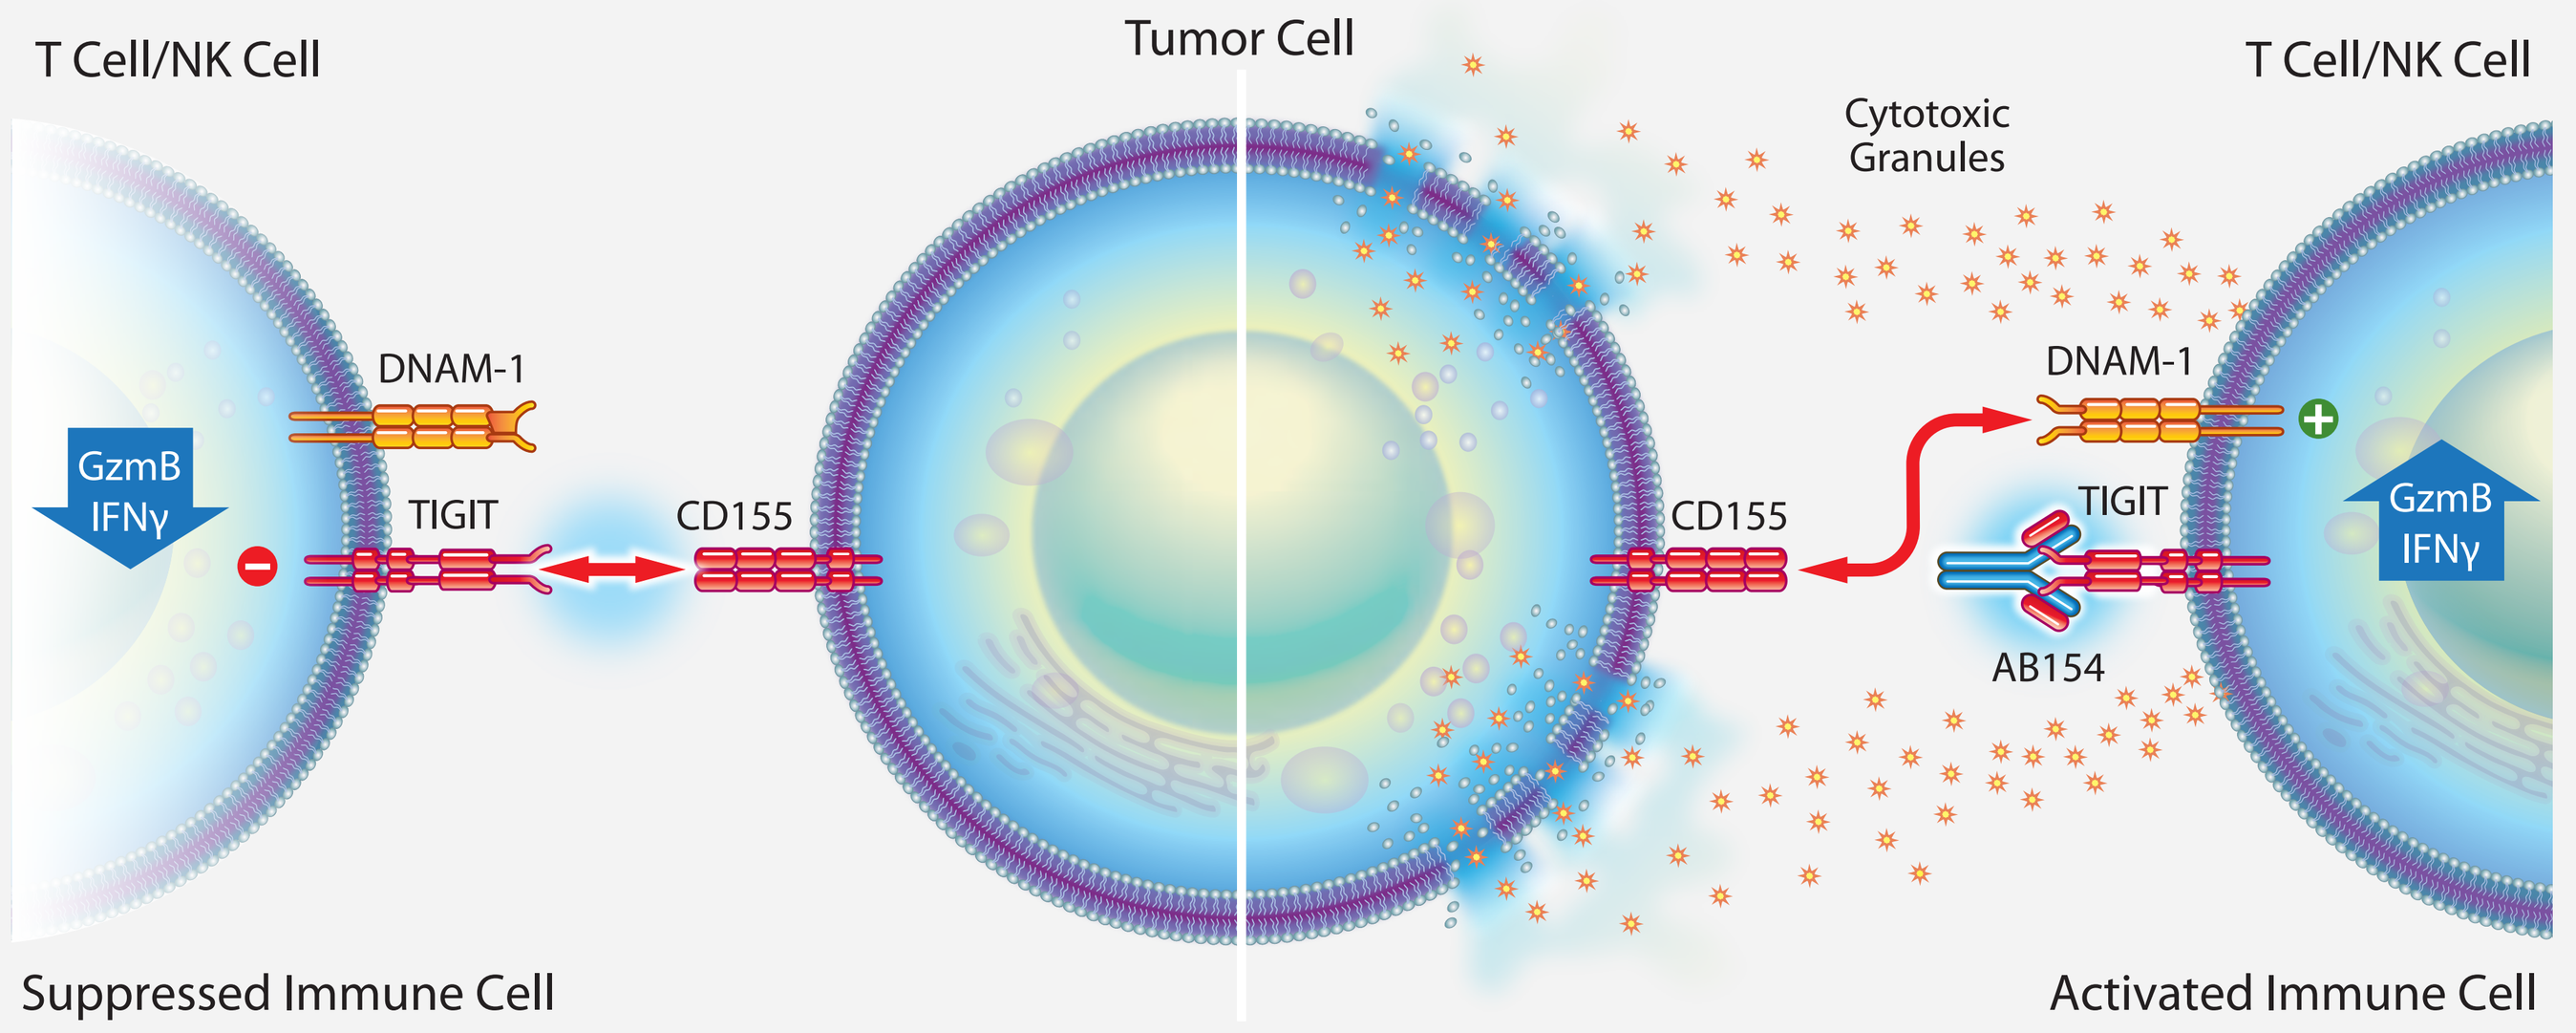
T Cell/NK Cell
Tumor Cell
T Cell/NK Cell
Cytotoxic Granules
DNAM-1
TIGIT
CD155
GzmB IFNγ
CD155
DNAM-1
TIGIT
AB154
GzmB IFNγ
Suppressed Immune Cell
Activated Immune Cell

AB154, domvanalimab; DNAM-1, DNAX accessory molecule-1; GzmB, granzyme B; IFNγ, interferon gamma; NK, natural killer; TIGIT, T cell immunoreceptor with Ig and ITIM domain.

- Domvanalimab (AB154) is a humanized mAb that blocks TIGIT and is engineered to lack FcγR binding function to minimize the risk of depleting intratumoral $CD8^+$ effector T cells[4]
- Treatment with domvanalimab, particularly in combination with other checkpoint inhibitors such as zimberelimab, has the potential to promote sustained immune activation and tumor clearance

TIGIT AND PD-1 BIOLOGY IN NSCLC

- In non-small cell lung cancer (NSCLC), CD155 protein is expressed within the cytoplasm and tumor cell membranes, and on adjacent blood vessels and tertiary lymphoid structures in the TME (**Figure 2**)

Figure 2. CD155 Protein is Found Throughout the Tumor Microenvironment[a]

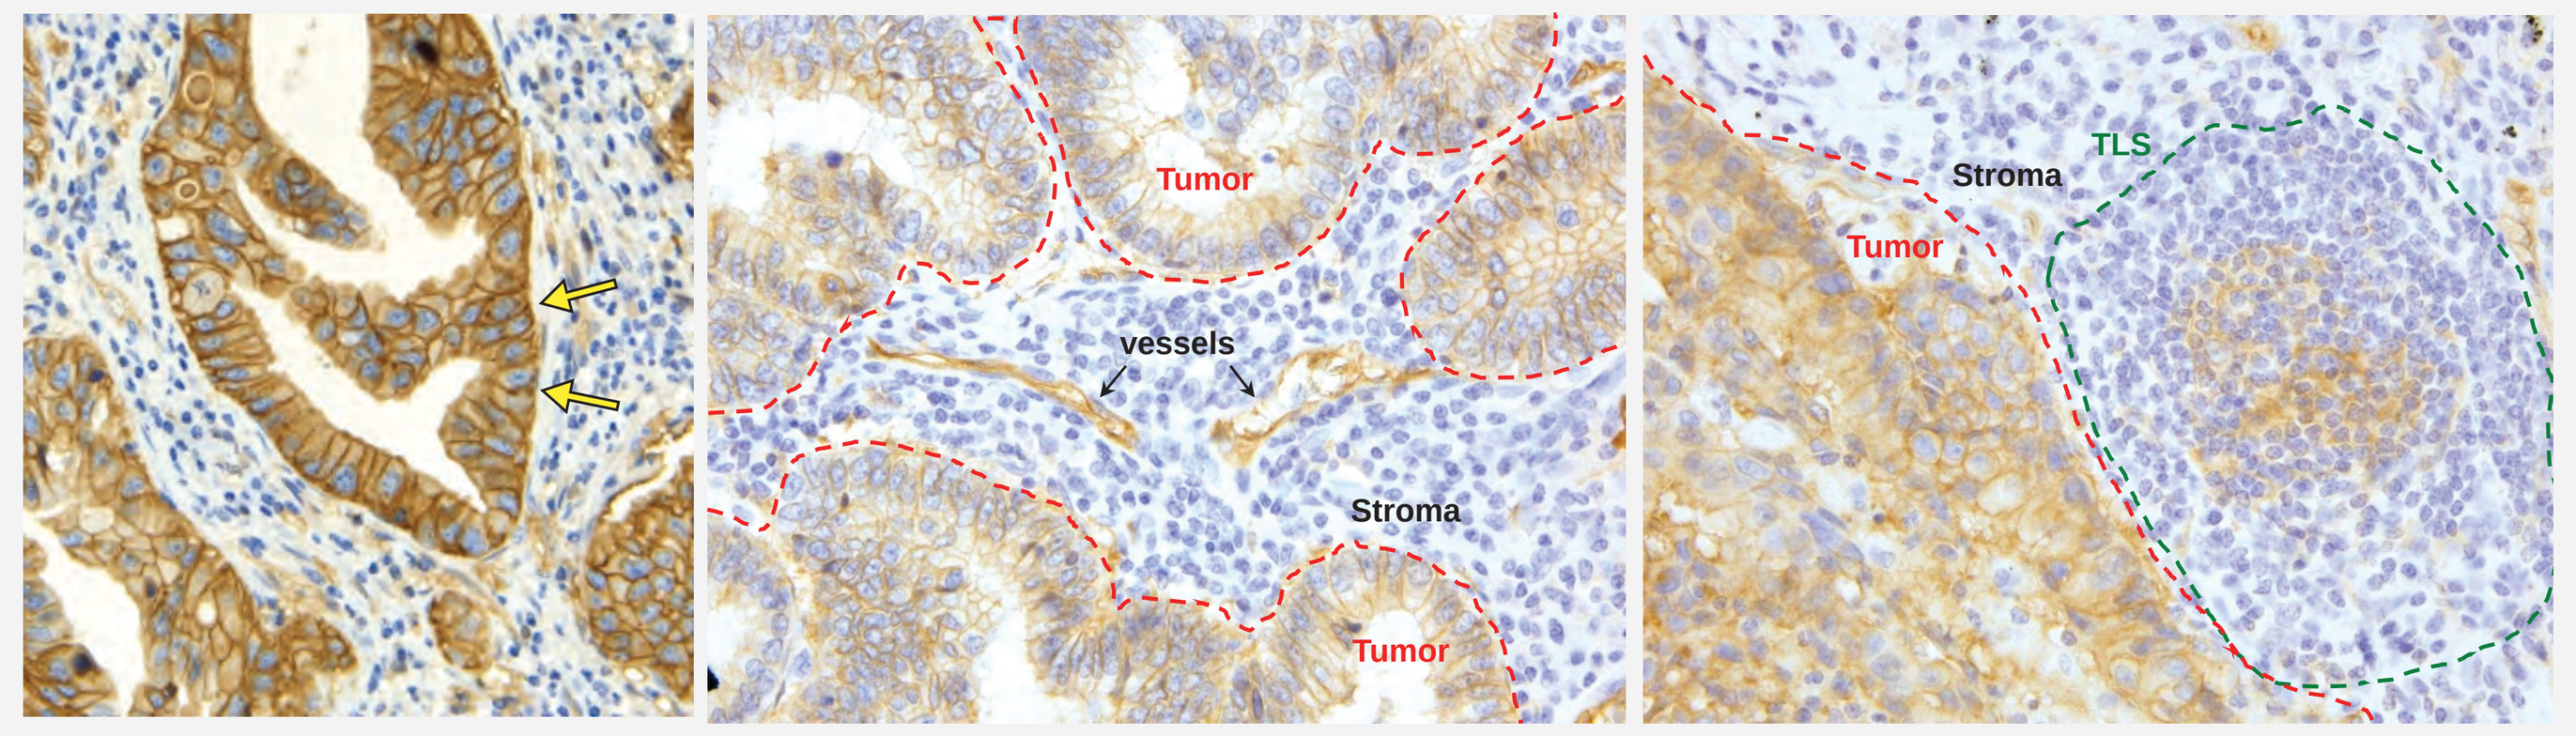
Tumor
vessels
Stroma
Tumor
Stroma
TLS
Tumor

[a] Yellow arrows indicate positive CD155 staining on IHC of human NSCLC tumors. IHC, immunohistochemistry; NSCLC, non-small cell lung cancer; TLS, tertiary lymphoid structure.

- Tumors from patients with NSCLC have high expression of TIGIT, DNAM-1, and PD-1[4], suggesting that the TIGIT and PD-1 pathways may be particularly important for the growth and persistence of this tumor type (**Figure 3**)

Figure 3. TIGIT, DNAM-1, and PD-1 are Coexpressed on Human Tumors[a]

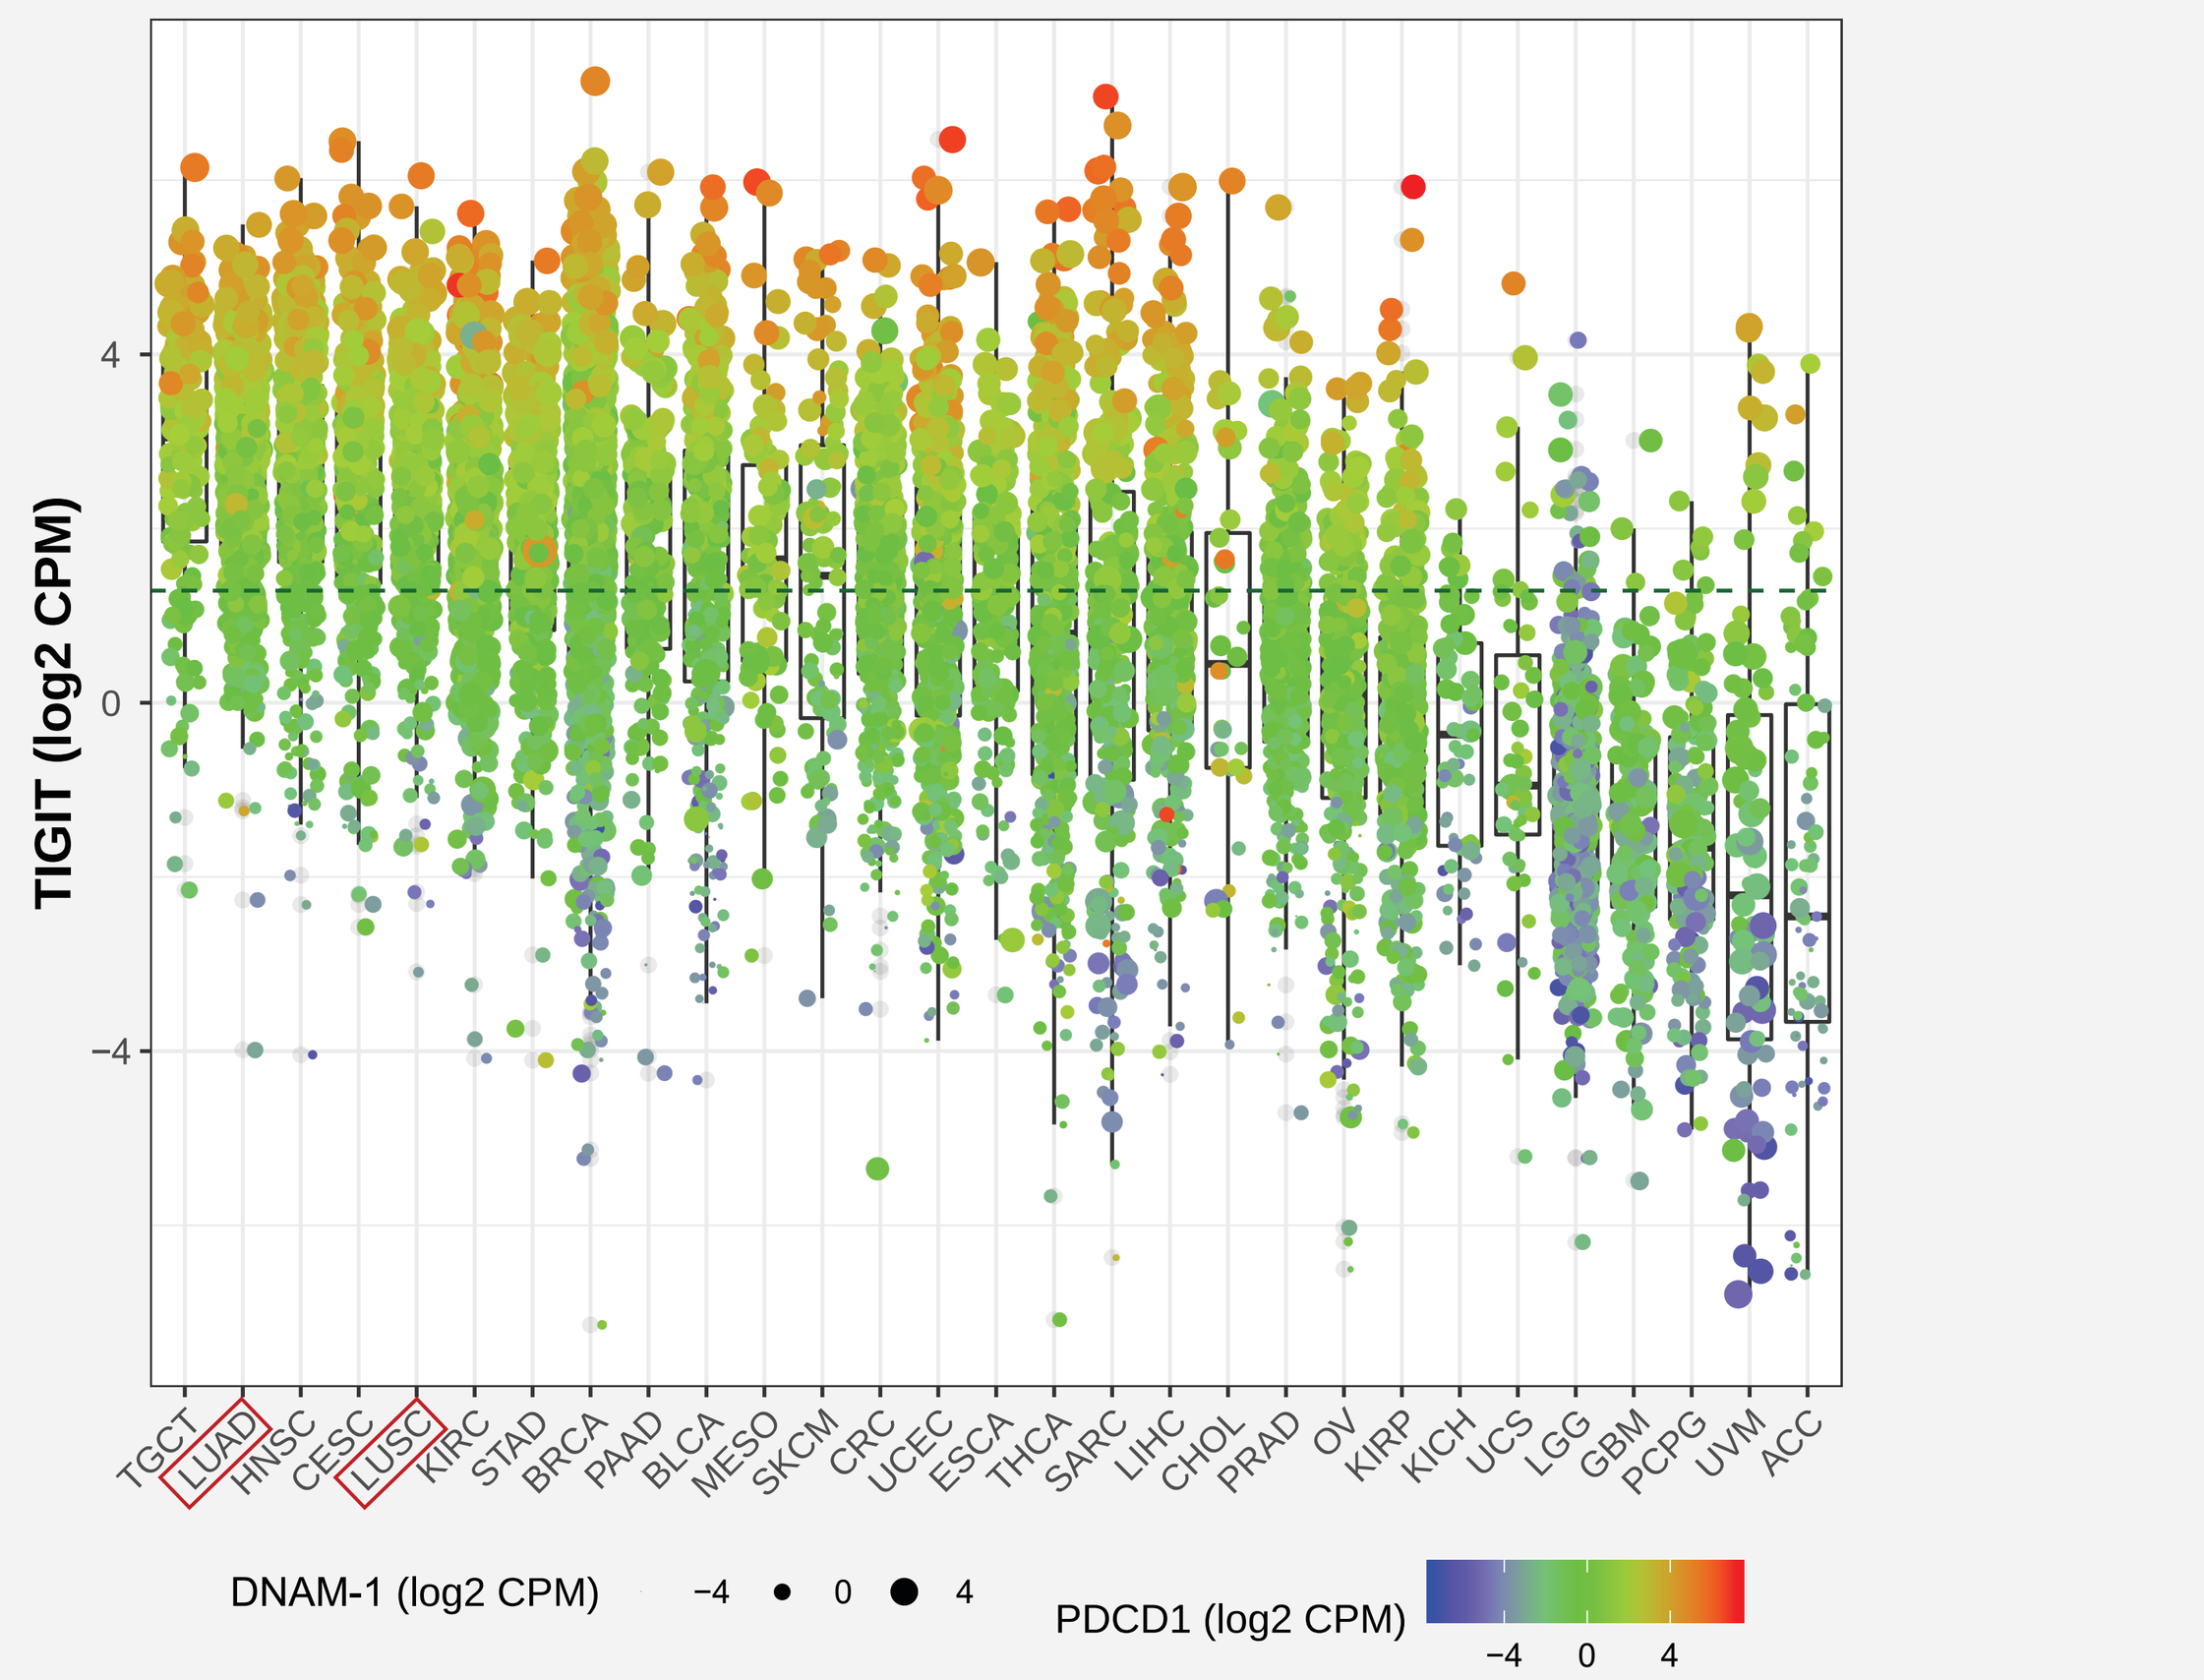
TIGIT (log2 CPM)
4
0
−4
TGCT LUAD HNSC CESC LUSC KIRC STAD BRCA PAAD BLCA MESO SKCM CRC UCEC ESCA THCA SARC LIHC CHOL PRAD OV KIRP KICH UCS LGG GBM PCPG UVM ACC
DNAM-1 (log2 CPM) −4 0 4
PDCD1 (log2 CPM) −4 0 4

[a] RNAseq data from The Cancer Genome Atlas. CPM, counts per million; DNAM-1, DNAX accessory molecule-1; LUAD, lung adenocarcinoma; LUSC, lung squamous cell carcinoma; TIGIT; T cell immunoreceptor with Ig and ITIM domain.

THE ADENOSINE AXIS IN CANCER

- Dying tumor cells release high levels of adenosine triphosphate (ATP) into the TME where CD39 and CD73 convert it to adenosine[5,6] (**Figure 4**)
- By binding adenosine receptors 2a and 2b ($A_{2a}R$ and $A_{2b}R$) expressed on immune cells, adenosine promotes immunosuppression by inhibiting critical components of the antitumor immune response and ultimately enables tumor cells to evade destruction[6]

Figure 4. Critical Role of Adenosine Pathway in Immunosuppressive Tumor Microenvironment

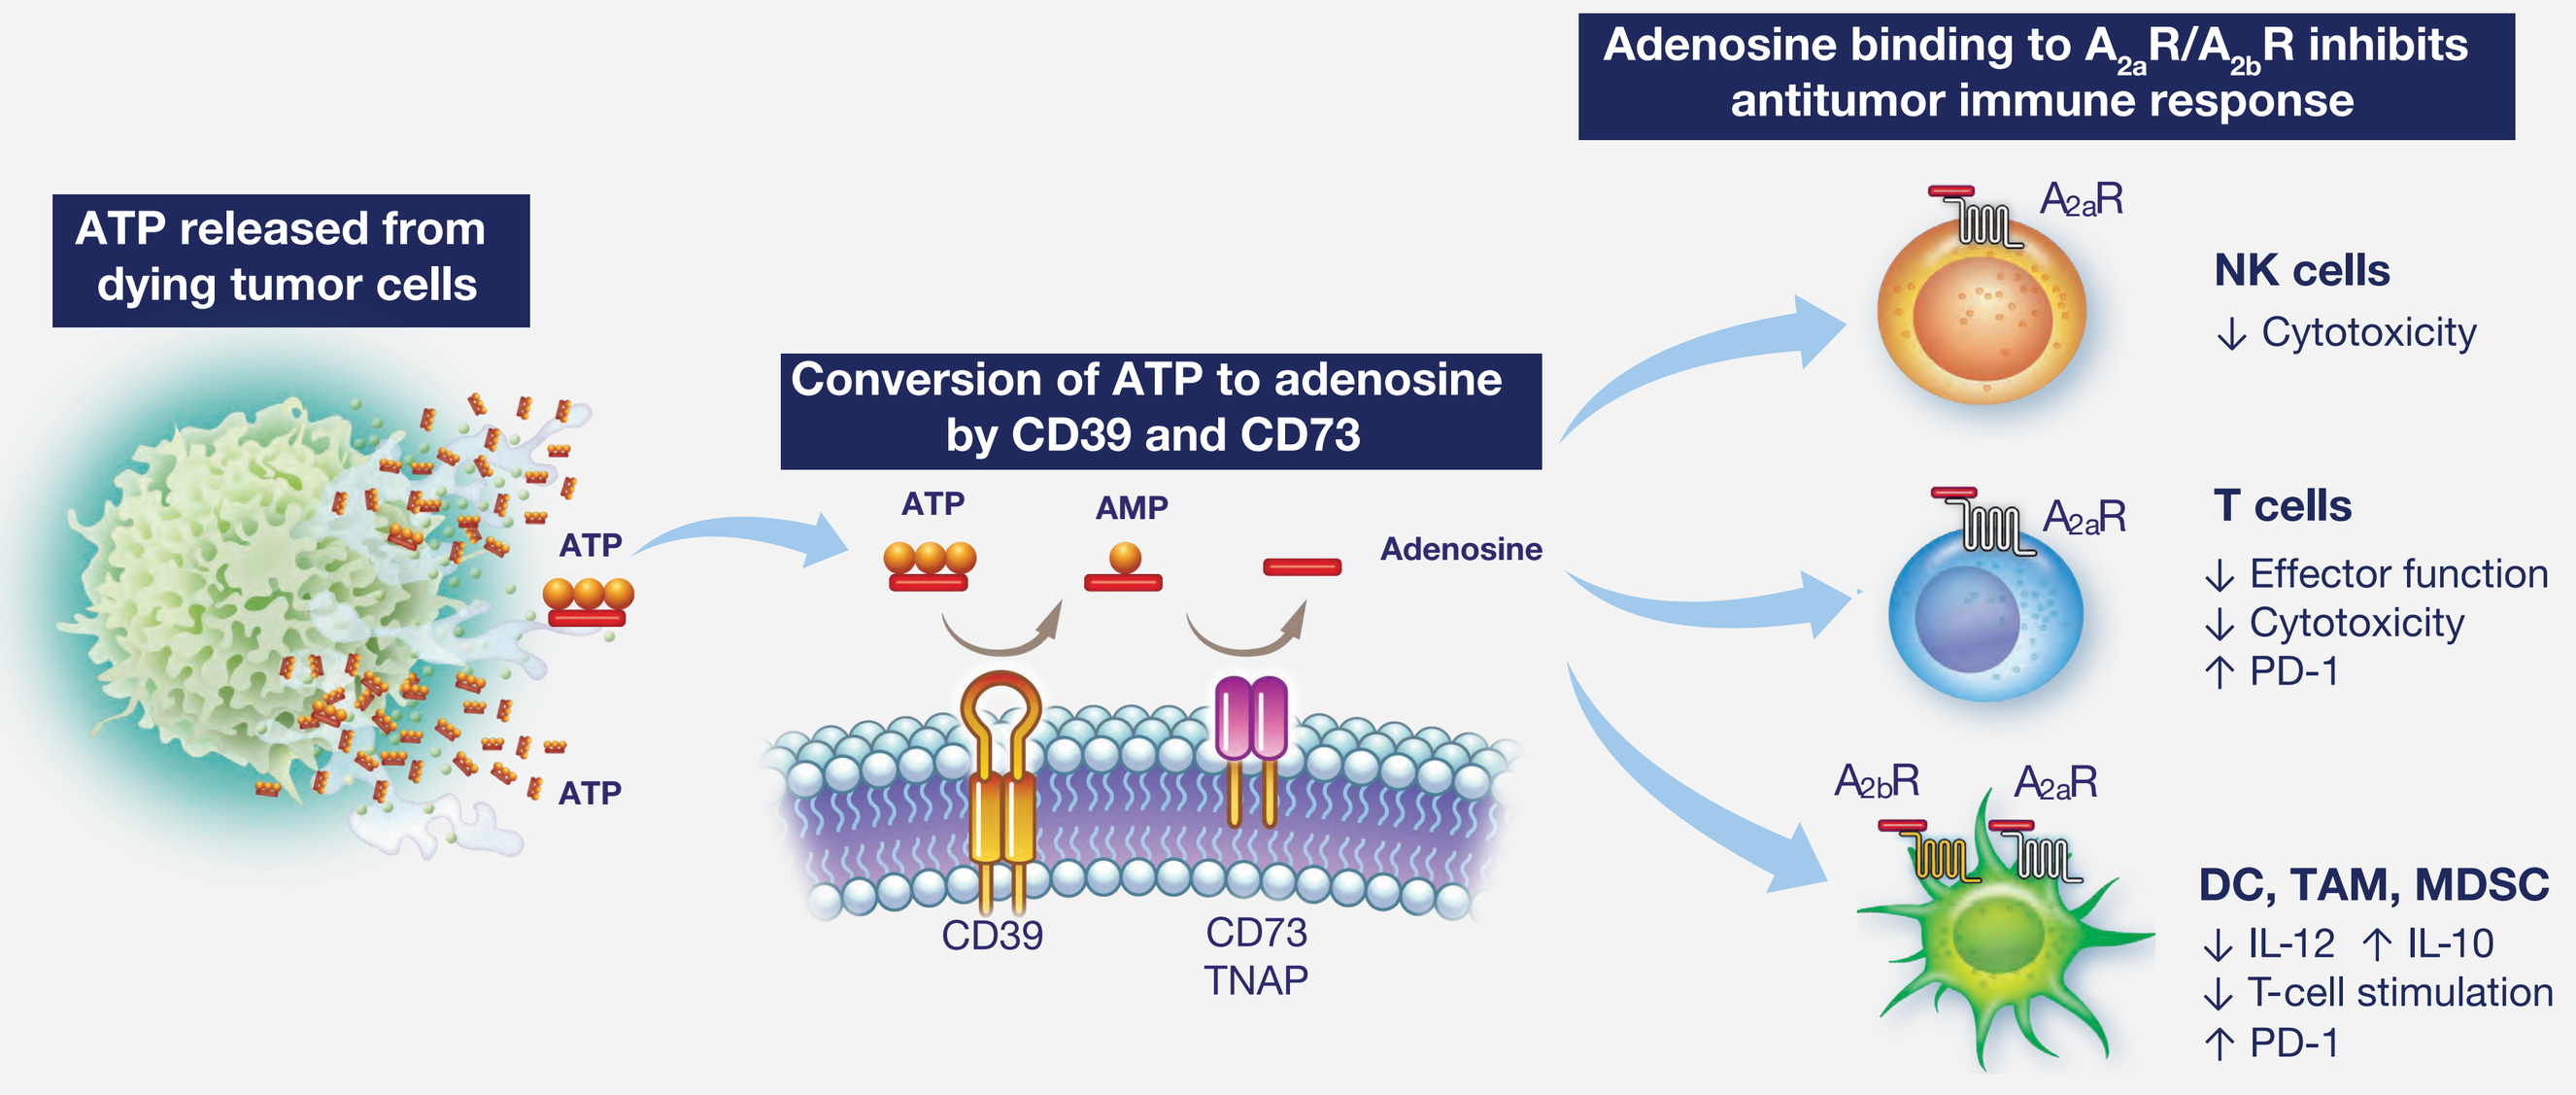
Adenosine binding to A2aR/A2bR inhibits antitumor immune response
ATP released from dying tumor cells
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
TNAP
A2aR
NK cells
↓ Cytotoxicity
T cells
↓ Effector function
↓ Cytotoxicity
↑ PD-1
A2bR
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation
↑ PD-1

AMP, adenosine monophosphate; ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; IL, interleukin; MDSC, myeloid-derived suppressor cells; NK, natural killer; TAM, tumor-associated macrophage; TNAP, tissue nonspecific alkaline phosphatase.

- Etrumadenant (AB928) is an orally bioavailable, small-molecule, selective dual antagonist of $A_{2a}R$ and $A_{2b}R$ that was specifically designed to block the immunosuppressive effects associated with high adenosine concentration within the TME; it is the only adenosine receptor antagonist in active clinical trials that potently blocks $A_{2b}R$

RATIONALE FOR TIGIT COMBINATIONS IN NSCLC

- For locally advanced or metastatic NSCLC, first-line treatment has historically included platinum-containing chemotherapy.[8] Median overall survival and 5-year survival rates associated with these regimens are low.[9]
- Immunotherapy agents targeting the PD-1 axis have improved outcomes for patients with NSCLC[10]; however, many patients neither initially respond to checkpoint inhibitors nor have durable responses[11], leaving an unmet need for new therapeutic approaches
- Combination therapy that includes inhibitors of the adenosine, TIGIT, and/or PD-1 pathways may hold promise for increasing efficacy without introducing significant new toxicity
 - In preclinical studies, blocking TIGIT in combination with PD-1 treatment yields greater antitumor activity and survival relative to either agent alone[11]
 - In patients with NSCLC, the adenosine pathway is a potential mechanism of resistance to anti–PD-1 therapy[12,13] and high tumor expression of CD73 is associated with poor prognosis[14], which may indicate a therapeutic advantage to combination $A_{2a}R/A_{2b}R$ and PD-1 blockade

ARC-7 Study Overview

- ARC-7 (NCT04262856) is a phase 2, multicenter, randomized, open-label, proof-of-concept study to evaluate the safety and efficacy of zimberelimab monotherapy and zimberelimab + domvanalimab ± etrumadenant in patients with PD-L1-high expressing NSCLC (**Figure 5**)

Figure 5. ARC-7 Study Design

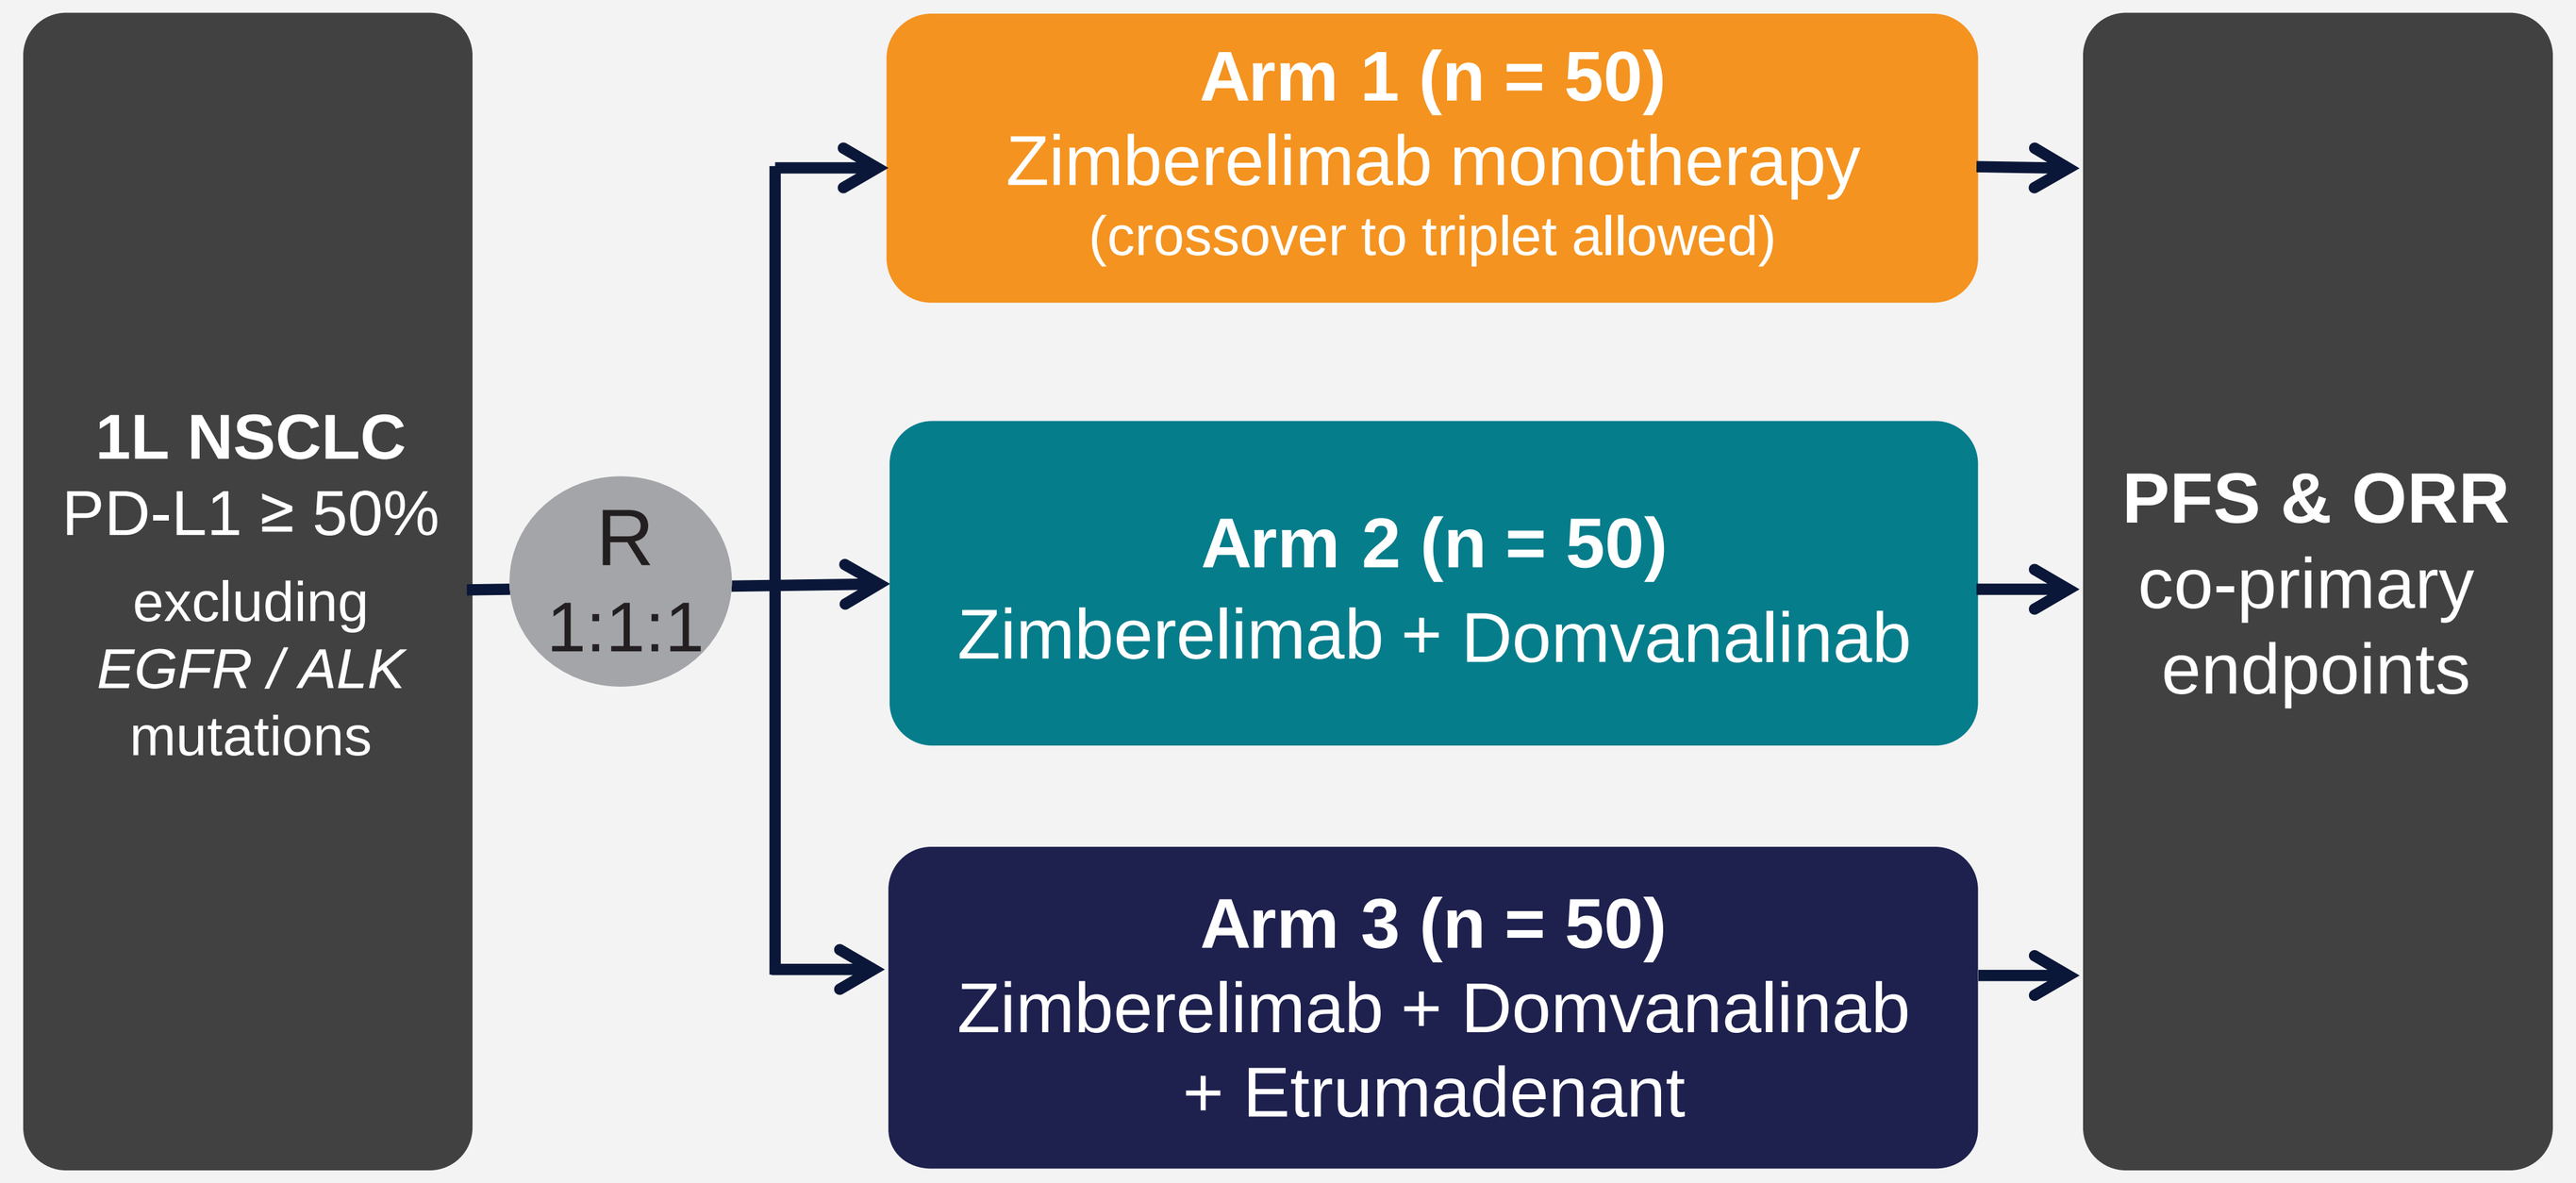
1L NSCLC
PD-L1 ≥ 50%
excluding
EGFR / ALK
mutations
R
1:1:1
Arm 1 (n = 50)
Zimberelimab monotherapy
(crossover to triplet allowed)
Arm 2 (n = 50)
Zimberelimab + Domvanalinab
Arm 3 (n = 50)
Zimberelimab + Domvanalinab
+ Etrumadenant
PFS & ORR
co-primary
endpoints

1L, first-line; *ALK*, anaplastic lymphoma kinase; *EGFR*, epidermal growth factor receptor; NSCLC, non-small cell lung cancer; ORR, objective response rate; PD-L1, programmed death-ligand 1; PFS, progression free survival; R, randomization.

- Key inclusion and exclusion criteria are shown in **Table 1**

Table 1. Key Eligibility Criteria for ARC-7

Key inclusion criteria	• Histologically-confirmed, treatment-naive, metastatic NSCLC • High PD-L1 expression (TPS ≥ 50%) • No *EGFR* or *ALK* genomic tumor aberrations • ≥ 1 measurable lesion per RECIST v1.1 • ECOG PS of 0 or 1
Key exclusion criteria	• Concurrent requirement for immunosuppressive medication or supraphysiologic doses of corticosteroids • Positive for hepatitis B, hepatitis C, or HIV antibody at screening

ALK, anaplastic lymphoma kinase; ECOG PS, Eastern Cooperative Oncology Group performance status; *EGFR*, epithelial growth factor receptor; HIV, human immunodeficiency virus; NSCLC, non-small cell lung cancer; PD-L1, programmed death-ligand 1; RECIST, Response Evaluation Criteria in Solid Tumors; TPS, tumor proportion score.

- Eligible patients (~150) will be stratified by ECOG PS (0 vs 1) and sex
- Patients randomized to Arm 1 (zimberelimab) will have the option to crossover to Arm 3 (zimberelimab + domvanalimab + etrumadenant) at the time of confirmed progressive disease (cPD); those who crossover will remain on treatment until developing cPD in Arm 3
- Primary endpoints are objective response rate and progression-free survival (RECIST v1.1); secondary endpoints include duration of response, disease control rate, overall survival, adverse events, PK, and immunogenicity
- Tumor biopsy will be performed during screening if an archival tumor sample obtained ≤ 24 months prior to screening is unavailable; an optional on-study biopsy will be performed at Day 29 if feasible
- Tumor assessments will be performed until disease progression, study withdrawal, or initiation of another anticancer treatment
- Safety and survival follow-up will continue until death, withdrawal of consent, or the end of the study, whichever occurs first
- ARC-7 is actively recruiting patients in the US, Australia, and Asia

CONCLUSIONS

- Blockade of the PD-1/PD-L1 immune checkpoint pathway has previously demonstrated clinical activity in patients with NSCLC; however, targeting additional mechanisms of immunosuppression, such as the TIGIT pathway or the adenosine axis, may be required to further improve clinical outcomes
- ARC-7 is a clinical study for patients with PD-L1–high expressing NSCLC aimed at investigating the potential clinical benefit of combination therapies targeting the PD-1, TIGIT, and adenosine pathways
- This will be the first study to investigate the triple combination of anti–PD-1, anti-TIGIT, and adenosine receptor antagonism including in patients who have progressed on PD-1 monotherapy for whom there is a high unmet need

ACKNOWLEDGMENTS AND DISCLOSURES

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation in this clinical trial. They also thank the ARC-7 Principal Investigators and study staff for their efforts in carrying out the study. A Chaudhry has been a speaker for Bayer, has participated in advisory boards for Astellas Pharma and Exelixis, and owns company stock from Novartis. His institution has received financial support from AbbVie, Alkermes, Amgen, Arcus Biosciences, Inc., AstraZeneca, Basilea Pharmaceutica, Bayer, BeiGene, BerGenBio, Blueprint Medicines Corp, Bristol Myers Squibb, Eli Lilly, Exelixis, Genentech, Gilead Sciences, InnoCare Pharma, Ipsen, Janssen Research, Merck, Millennium Pharmaceuticals, Novartis, and Pfizer. Medical writing support was provided by Amanda Martin, PhD, and Kendall Foote, PhD, of Medical Expressions (Chicago, IL) and was funded by Arcus Biosciences, Inc. This study is sponsored by Arcus Biosciences, Inc.

REFERENCES

1. Sharpe AH and Pauken KE. *Nat Rev Immunol*. 2018;18(3):153–167.
2. Dougall WC, et al. *Immunol Rev*. 2017;276(1):112–120.
3. Blake SJ, et al. *Clin Cancer Res*. 2016;22(21):5183–5138.
4. Anderson AE, et al. *SITC 2019*. Poster P260.
5. Martins I, et al. *Cell Cycle*. 2009;8(22):3723–3728.
6. Vijayan D, et al. *Nat Rev Cancer*. 2017;17(12)709–724.
7. Powderly J, et al. *ESMO 2019*. Abstract 4854.
8. Zhang C, et al. *J Hematol Oncol*. 2019;12(1):45.
9. SEER Cancer Stat Facts: Lung and Bronchus Cancer. National Cancer Institute. Bethesda, MD, https://seer.cancer.gov/statfacts/html/lungb.html
10. Reck M, et al. *N Engl J Med*. 2016;375(19):1823–1833.
11. Dixon KO, et al. *J Immunol*. 2018;200(8):3000–3007.
12. Fong L, et al, *ASCO 2017*. Abstract 3004.
13. Giannakis M, et al. *ASCO 2017*. Abstract 3036.
14. Inoue Y, et al. *Oncotarget*. 2017;8(5):8738–8751.

Corresponding Author: Dawn Colburn (dcolburn@arcusbio.com)

ESMO Virtual Congress 2020