ARC-8: Phase 1/1b Study to Evaluate Safety and Tolerability of AB680 + Chemotherapy + Zimberelimab (AB122) in Patients with Treatment-Naive Metastatic Pancreatic Adenocarcinoma

Abstract #404

GA Manji[1], ZA Wainberg[2], K Krishnan[3], N Giafis[3], A Udyavar[3], CS Quah[3], S Liu[3], W Berry[3], D DiRenzo[3], K Gerrick[3], L Jin[3], JC Bendell[4]

[1]Columbia University Herbert Irving Comprehensive Cancer Center, New York, NY; [2]University of California Los Angeles David Geffen School of Medicine, Los Angeles, CA; [3]Arcus Biosciences, Inc., Hayward, CA; [4]Sarah Cannon Research Institute, Nashville, TN

INTRODUCTION

The Adenosine Axis in Cancer

- Standard chemotherapy regimens may contribute to immunosuppression by elevating intratumoral levels of adenosine triphosphate (ATP) in the tumor microenvironment (TME) where the enzymes CD39 and CD73 successively convert ATP to adenosine[1,2] (**Figure 1**)
- By binding adenosine receptors 2a and 2b ($A_{2a}R$ and $A_{2b}R$) expressed on immune cells, adenosine promotes immunosuppression by inhibiting critical components of the antitumor immune response, ultimately enabling tumors to evade destruction[2]
- Additionally, $A_{2a}R$ signaling impairs the activation, proliferation, and cytotoxic activity of effector T cells[3]
- Initial research focused on $A_{2a}R$ as the most relevant adenosine receptor in cancer physiology; however, $A_{2b}R$ signaling through MAP kinase pathway activation mediates unique functions, such as cancer cell intrinsic survival and dendritic cell activation and function[4]
- Thus, adenosine pathway blockade may be necessary to overcome adenosine-dependent immunosuppression leading to enhanced therapeutic efficacy of some chemotherapeutic agents[2]

Figure 1. Critical Role of Adenosine Pathway in the Immunosuppressive TME

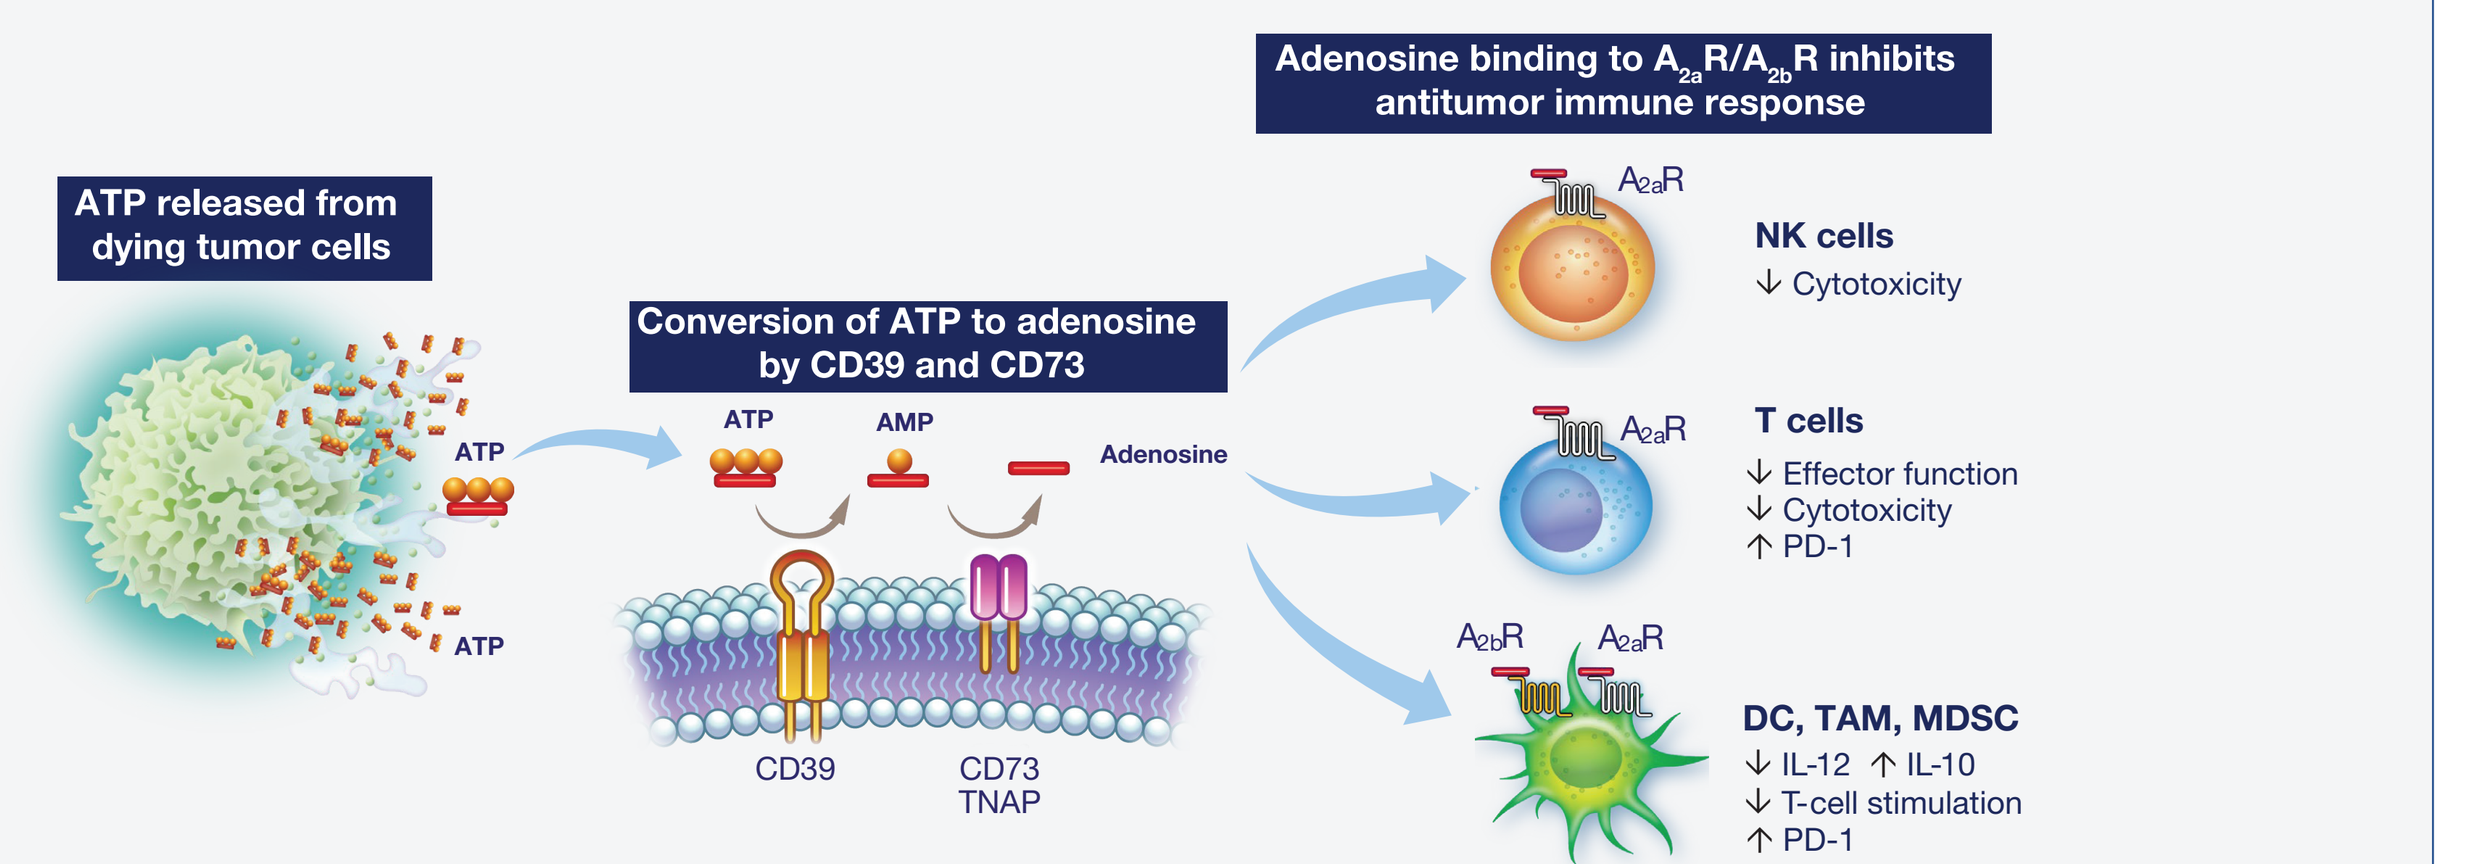
Adenosine binding to A2aR/A2bR inhibits antitumor immune response
ATP released from dying tumor cells
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
TNAP
A2aR
NK cells
↓ Cytotoxicity
T cells
↓ Effector function
↓ Cytotoxicity
↑ PD-1
A2bR
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation
↑ PD-1

AMP, adenosine monophosphate; ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; IL, interleukin; MDSC, myeloid-derived suppressor cell; NK, natural killer; PD-1, programmed cell death protein-1; TAM, tumor-associated macrophage; TME, tumor microenvironment; TNAP, tissue nonspecific alkaline phosphatase.

- AB680 is a potent, selective small-molecule inhibitor of soluble and membrane-bound CD73 developed with the aim of eliminating adenosine-mediated immunosuppression in the TME; it is the first clinical-stage small-molecule CD73 inhibitor
- Preliminary data indicated that systemic exposure (C_{max} and AUC) of 25-75 mg AB680 administered intravenously (IV) once every 2 weeks (Q2W) increased in a dose-proportional manner; AB680 pharmacokinetics (PK) profiles were similar in healthy volunteers and patients with cancer[5]
- AB680 administration to healthy volunteers at doses as low as 16 mg results in ~100% inhibition of circulating soluble CD73 for an extended period of time (>7 days), thus, the starting AB680 dose level in ARC-8 (25 mg Q2W) is known to be pharmacologically active and to provide close to maximal inhibition of CD73 in circulation[6]

ARC-8 Study Rationale

- Standard treatment for metastatic pancreatic ductal adenocarcinoma (mPDAC) includes combination chemotherapy regimens such as nab-paclitaxel and gemcitabine (NP/Gem); however, the overall and complete response rates for patients with mPDAC are 23-29% and 1%, respectively, with a 9% 5-year survival rate[7,8]
- Additionally, combinations of NP/Gem + programmed cell death protein-1 (PD-1) inhibitors have demonstrated an 18-27% overall response rate (ORR)[9,10], further highlighting an urgent unmet need for new targeted treatment options
- Human pancreatic tumors express high levels of CD73, which are associated with *KRAS* mutation[5] (**Figure 2**)
- Over 90% of invasive PDACs have mutated *KRAS*; compared with patients who have PDAC and wild-type *KRAS*, those with mutated *KRAS* have worse clinical outcomes[6,11]
- Higher CD73 expression is strongly associated with worse progression-free survival in patients with PDAC, regardless of *KRAS* status[12]
- In a murine B16F10 melanoma model, tumor growth was suppressed with AB680 monotherapy, an effect that was further enhanced with the addition of anti–PD-1 antibodies[5]
 - AB680 + anti–PD-1 treatment increased the number of intratumoral effector CD4+ and CD8+ T cells and decreased the numbers of Tregs and myeloid-derived suppressor cells[5]
 - Further, AMP-mediated suppression of human CD4+ and CD8+ T cell activation was reversed with AB680 treatment[5]
- Based on these observations, novel combinations of AB680 and PD-1 pathway inhibition may potentially offer a complementary approach to increase antitumor immune activation and improved anticancer therapy

Figure 2. KRAS Mutation Correlates With Elevated CD73 Expression

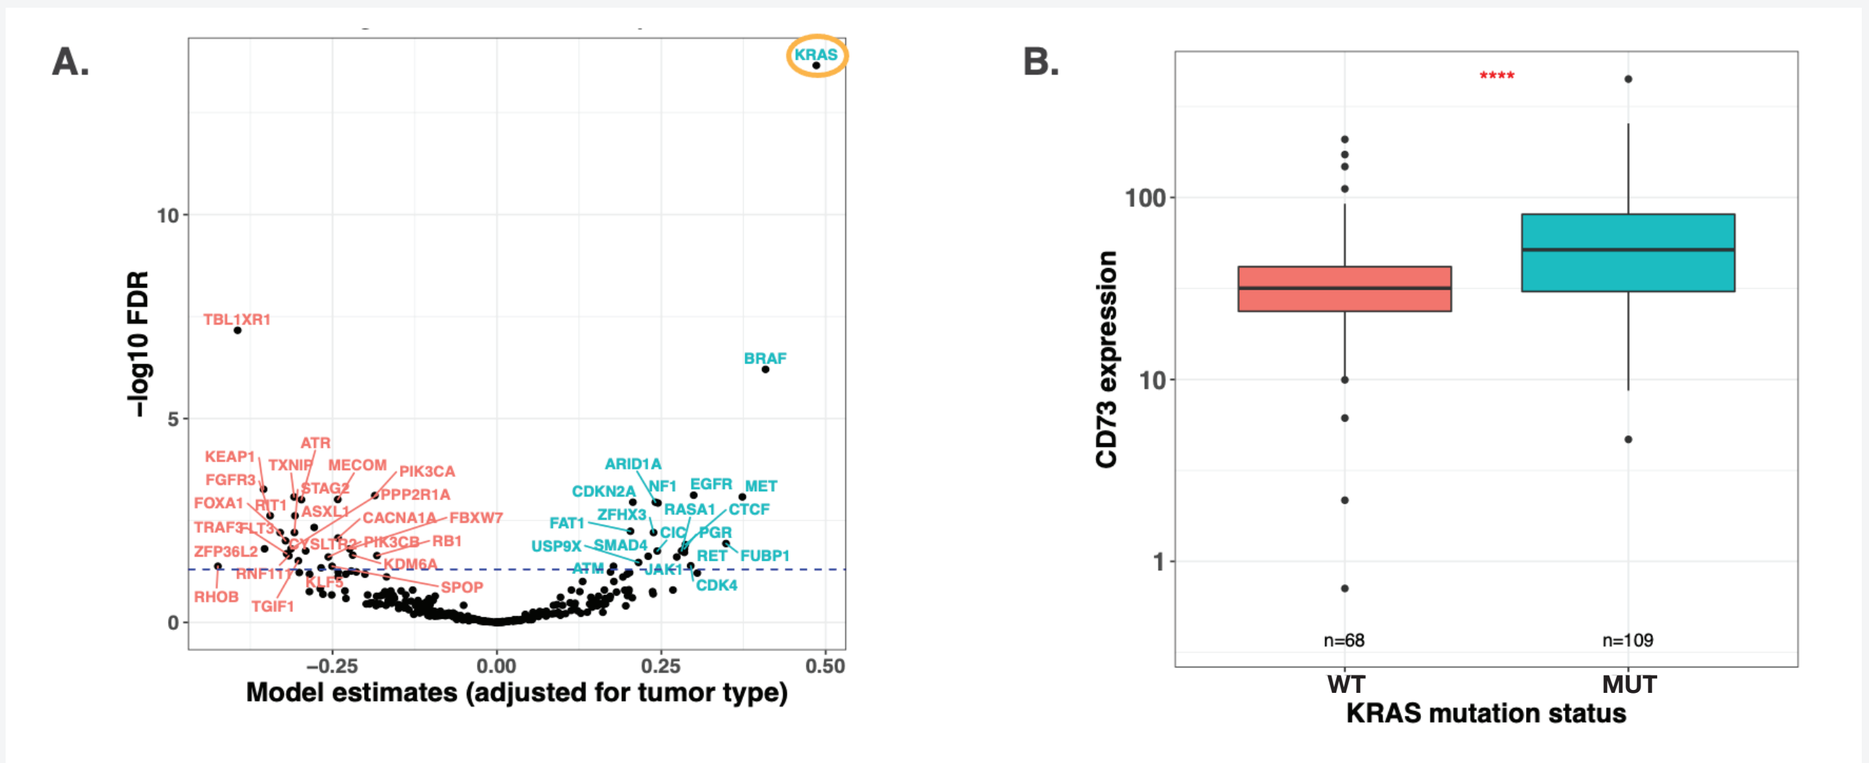
A.
B.
KRAS
BRAF
TBL1XR1
–log10 FDR
Model estimates (adjusted for tumor type)
CD73 expression
n=68
n=109
WT
MUT
KRAS mutation status

(A) Linear model estimates adjusted for tumor type of alterations in cancer driver genes that predict CD73 expression (B) Pancreatic adenocarcinoma samples from the TCGA demonstrate that tumors harboring *KRAS* mutations have significantly elevated CD73 expression levels (****p<0.001). FDR, false discovery rate; MUT, mutated, WT, wild type.

METHODS

Study Design

- ARC-8 (NCT04104672) is an ongoing, Phase 1/1b, open-label, dose-escalation, and dose-expansion study to evaluate the safety, tolerability, PK and clinical activity of AB680 in combination with NP/Gem and zimberelimab (Zim; AB122; anti–PD-1) as a first-line treatment for patients with mPDAC (**Figure 3**)
- The dose-escalation stage consists of a standard 3+3 design in which AB680 (25, 50, 75, or 100 mg) is administered IV Q2W with standard doses of NP/Gem + Zim (240 mg) to determine the recommended dose for expansion (RDE)
- In the dose-expansion, AB680 will be administered at the RDE with NP/Gem + Zim

Figure 3. ARC-8 Study Design

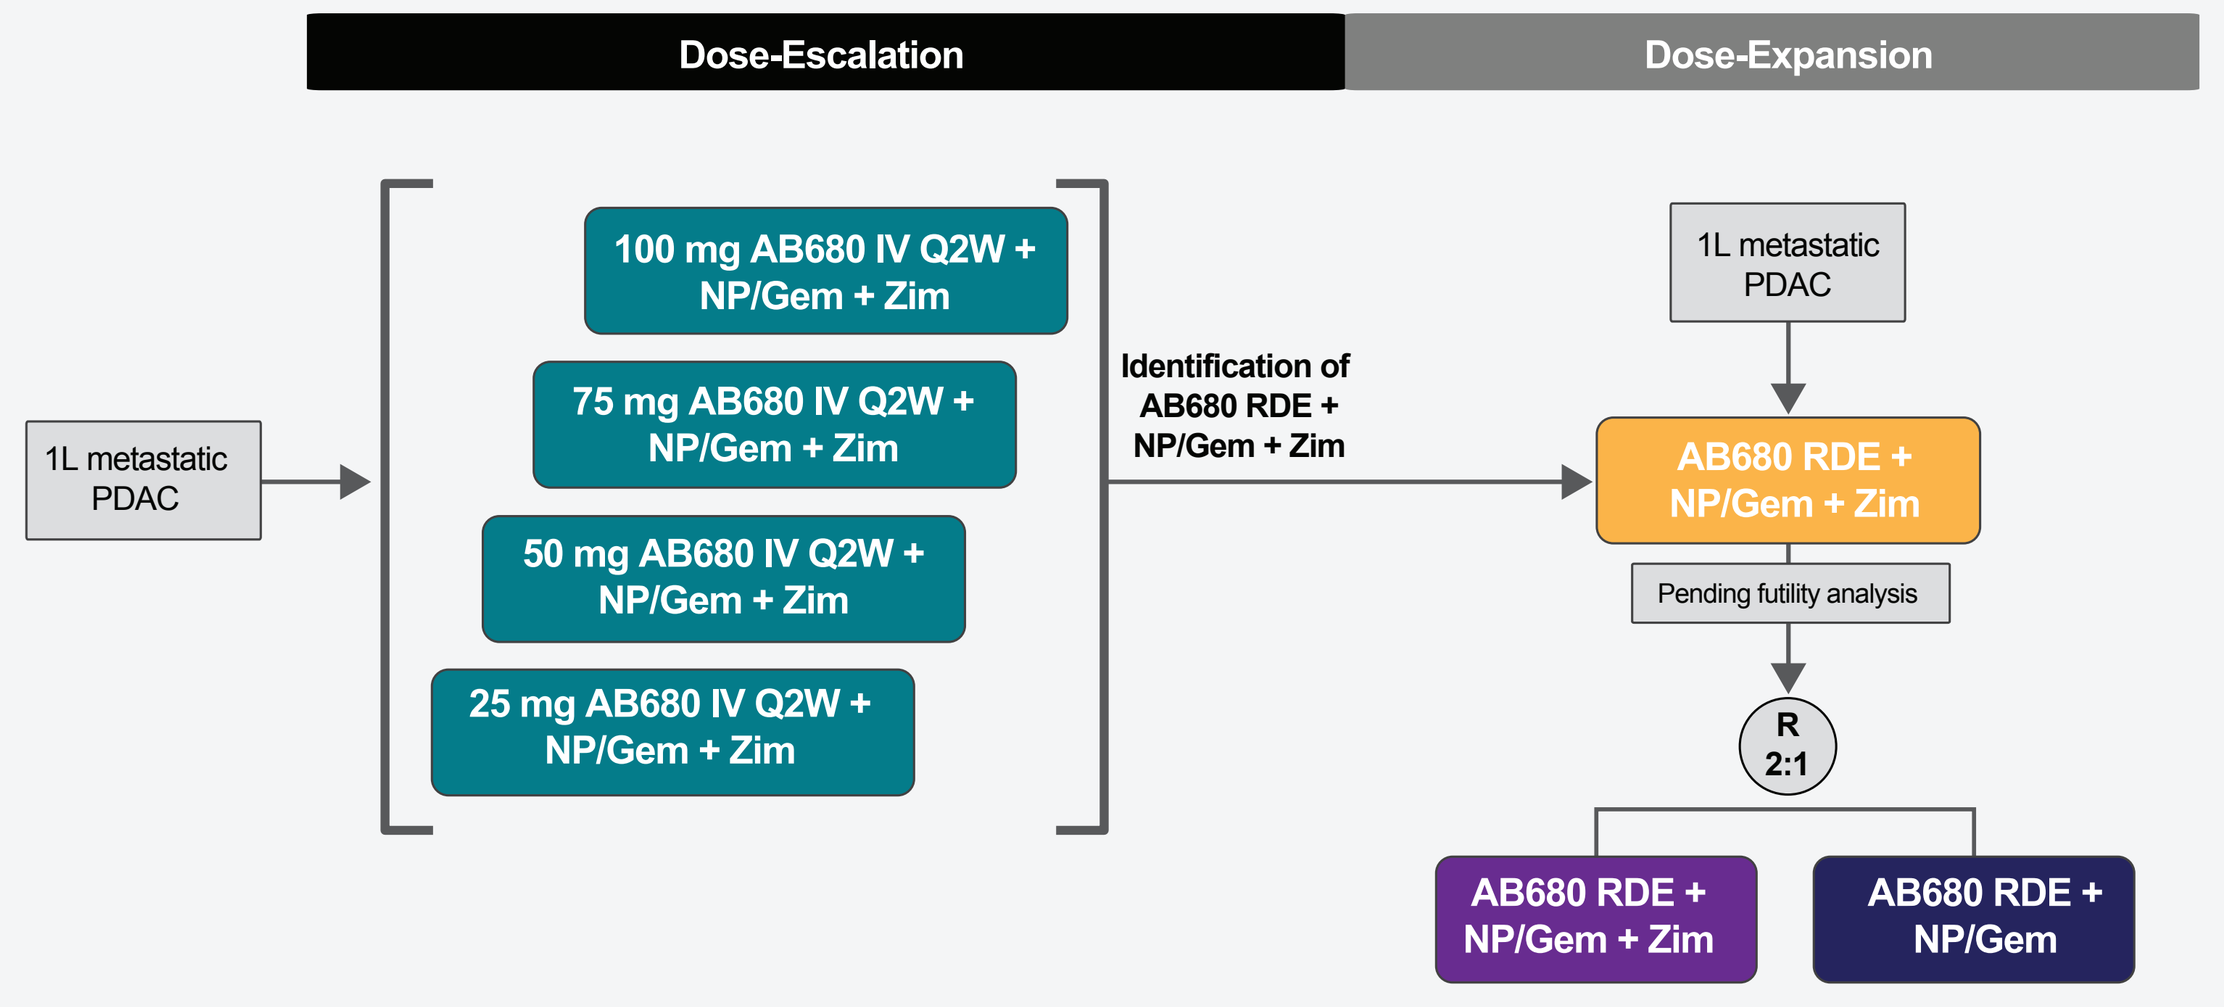
Dose-Escalation
Dose-Expansion
1L metastatic PDAC
100 mg AB680 IV Q2W + NP/Gem + Zim
75 mg AB680 IV Q2W + NP/Gem + Zim
50 mg AB680 IV Q2W + NP/Gem + Zim
25 mg AB680 IV Q2W + NP/Gem + Zim
Identification of AB680 RDE + NP/Gem + Zim
1L metastatic PDAC
AB680 RDE + NP/Gem + Zim
Pending futility analysis
R 2:1
AB680 RDE + NP/Gem + Zim
AB680 RDE + NP/Gem

Safety monitoring throughout treatment period; radiographic disease evaluation every 8 weeks

1L, first-line; IV, intravenously; Gem, gemcitabine; NP, nab-paclitaxel; PDAC, pancreatic ductal adenocarcinoma; Q2W, every 2 weeks; R, randomization; RDE, recommended dose for expansion; Zim, zimberelimab.

ARC-8 Design Features

- The primary objective of the study is to assess safety and tolerability of AB680 combination therapy in patients with mPDAC with secondary objectives that include AB680 PK and clinical activity
- Eligible patients are adults who have histologically- or cytologically-confirmed mPDAC and have not previously received treatment for metastatic disease; ≥1 measurable lesion per RECIST v1.1; and an ECOG performance status of 0-1
 - Prior (neo)adjuvant treatment for PDAC (chemotherapy [including NP or Gem] and/or radiotherapy) is allowed if it was completed ≥6 months prior to study enrollment
 - Patients initially diagnosed with locally advanced PDAC who have undergone chemotherapy then resection and had no evidence of disease are eligible for the study if relapse of metastatic disease has occurred and the last dose of chemotherapy was received ≥6 months prior to study enrollment
- Baseline archival tumor samples (≤12 months old) or on-treatment biopsies (if medically feasible) are collected from all patients
- Study treatment may continue until disease progression, unacceptable toxicity, consent withdrawal, or by the investigator's decision

Statistical Analysis

- Safety analyses included all patients who received ≥1 dose of any study drug; summary statistics are shown for treatment-emergent adverse events (TEAEs), serious adverse events (TESAEs), event severity, and relationship to study drugs
- Efficacy analyses included all patients who had a baseline and ≥1 post-baseline assessment or discontinued study treatment due to progressive disease or death; patients were considered efficacy-evaluable if they had ≥1 disease assessment

RESULTS

Patient Baseline Characteristics

- As of November 11, 2020, 19 patients have received AB680 + NP/Gem + Zim: 25 mg AB680 (n=4), 50 mg AB680 (n=6), 75 mg AB680 (n=3), and 100 mg AB680 (n=6; **Table 1**)
 - Based on the totality of the data including PK, PK/PD correlation, and available safety data, 100 mg AB680 has been tentatively selected as the RDE
- The majority of study patients were men; the mean age of all study patients was 64 years

Table 1. Patient Demographics and Characteristics

Parameter	25 mg AB680 + NP/Gem + Zim (n=4)	50 mg AB680 + NP/Gem + Zim (n=6)	75 mg AB680 + NP/Gem + Zim (n=3)	100 mg AB680 + NP/Gem + Zim (n=6)	All Patients (N=19)
Age, mean (SD), years	67.3 (2.2)	61.8 (12.7)	55.3 (2.1)	68.5 (8.7)	64.1 (9.5)
Male, n (%)	2 (50)	4 (67)	2 (67)	3 (50)	11 (58)
Race, n (%)					
White	4 (100)	4 (67)	2 (67)	4 (67)	14 (74)
Asian	0	1 (17)	1 (33)	1 (17)	3 (16)
Not reported	0	1 (17)	0	1 (17)	2 (11)
ECOG PS, n (%)					
0	4 (100)	4 (67)	3 (100)	3 (50)	14 (74)
1	0	2 (33)	0	3 (50)	5 (26)

ECOG PS, Eastern Cooperative Oncology Group performance status; NP/Gem, nab-paclitaxel/gemcitabine; SD, standard deviation; Zim, zimberelimab.

Safety Analyses

- As of November 11, 2020, 1 dose-limiting toxicity (Grade 2 autoimmune hepatitis) occurred in the 50 mg AB680 cohort; the event resolved completely with steroid treatment and the patient resumed study treatment
- Of all safety-evaluable patients, 18/19 (95%) experienced ≥1 TEAE; the most common TEAEs were fatigue (n=13; 68%), anemia (n=10; 53%), alopecia (n=8; 42%), diarrhea (n=8; 42%), and neutrophil count decreased (n=8; 42%; **Table 2**)
- Study drug-related TEAEs were reported by 8/19 (42%) patients; 4 patients had Grade 1 or 2 study-drug related TEAEs and 4 patients experienced Grade 3 study drug-related TEAEs (n=2 in 50 mg AB680 cohort; n=1 each in 75 mg and 100 mg AB680 cohorts)
 - Grade 3 study drug-related TEAEs: anemia (n=2), neutrophil count decreased (n=2), lymphocyte count decreased (n=1), platelet count decreased (n=1), and C. difficile colitis (n=1)
- TESAEs were reported by 7/19 (37%) patients; for 2 patients, these TESAEs experienced were study drug-related (Grade 2 blood bilirubin increased [n=1 in 50 mg AB680 cohort] and Grade 3 *C. difficile* colitis [n=1 in 100 mg AB680 cohort])
- There were no patients who discontinued study treatment due to TEAEs
- In limited patient numbers across each dose-escalation cohort, no significant additive toxicity has been observed over that expected with NP/Gem alone

Table 2. Treatment Emergent Adverse Events

Parameter, n (%)	25 mg AB680 + NP/Gem + Zim (n=4)	50 mg AB680 + NP/Gem + Zim (n=6)	75 mg AB680 + NP/Gem + Zim (n=3)	100 mg AB680 + NP/Gem + Zim (n=6)	All Patients (N=19)
Any TEAE	4 (100)	6 (100)	3 (100)	5 (83)	18 (95)
Grade ≥3	3 (75)	5 (83)	1 (33)	3 (50)	12 (63)
Any TESAE	1 (25)	3 (50)	0	3 (50)	7 (37)
Grade ≥3	1 (25)	2 (33)	0	3 (50)	6 (32)
Study drug-related TEAEs	1 (25)	3 (50)	2 (67)	2 (33)	8 (42)
Grade ≥3	0	2 (33)	1 (33)	1 (17)	4 (21)
Study drug-related TESAEs	0	1 (17)	0	1 (17)	2 (11)
Grade ≥3	0	0	0	1 (17)	1 (5)
TEAEs in ≥4 patients					
Fatigue	4 (100)	6 (100)	1 (33)	2 (33)	13 (68)
Anemia	1 (25)	5 (83)	2 (67)	2 (33)	10 (53)
Alopecia	2 (50)	2 (33)	2 (67)	2 (33)	8 (42)
Diarrhea	2 (50)	4 (67)	1 (33)	1 (17)	8 (42)
Neutrophil count decreased	2 (50)	4 (67)	1 (33)	1 (17)	8 (42)
Nausea	1 (25)	3 (50)	2 (67)	1 (17)	7 (37)
Platelet count decreased	2 (50)	2 (33)	1 (33)	2 (33)	7 (37)
Pyrexia	3 (75)	3 (50)	1 (33)	0	7 (37)
AST increased	1 (25)	2 (33)	2 (67)	1 (17)	6 (32)
Vomiting	2 (50)	3 (50)	1 (33)	0	6 (32)
ALT increased	0	2 (33)	2 (67)	1 (17)	5 (26)
Blood ALP increased	0	1 (17)	2 (67)	2 (33)	5 (26)
Constipation	1 (25)	3 (50)	1 (33)	0	5 (26)
Lymphocyte count decreased	0	1 (17)	1 (33)	3 (50)	5 (26)
Chills	1 (25)	1 (17)	1 (33)	1 (17)	4 (21)
Decreased appetite	0	2 (33)	0	2 (33)	4 (21)
Hypoalbuminemia	0	1 (17)	1 (33)	2 (33)	4 (21)

ALP, alkaline phosphatase; ALT, alanine aminotransferase; AST, aspartate aminotransferase; NP/Gem, nab-paclitaxel/gemcitabine; TEAE, treatment-emergent adverse event; TESAE, treatment-emergent serious adverse event; Zim, zimberelimab.

Clinical Activity

- As of December 9, 2020, there were 17 efficacy-evaluable patients; partial responses (PR) were observed in 7 patients (41%), including 3 confirmed responses (**Figure 4**); of the 4 unconfirmed responders, 3 responded at the first tumor assessment, and all 4 remain on study treatment
- As of December 9, 2020, 16 patients remained on active treatment; with limited follow-up, 6/6 patients receiving 100 mg AB680 (cohort 4) continue on therapy

Figure 4. Time on Treatment and RECIST v1.1 Response

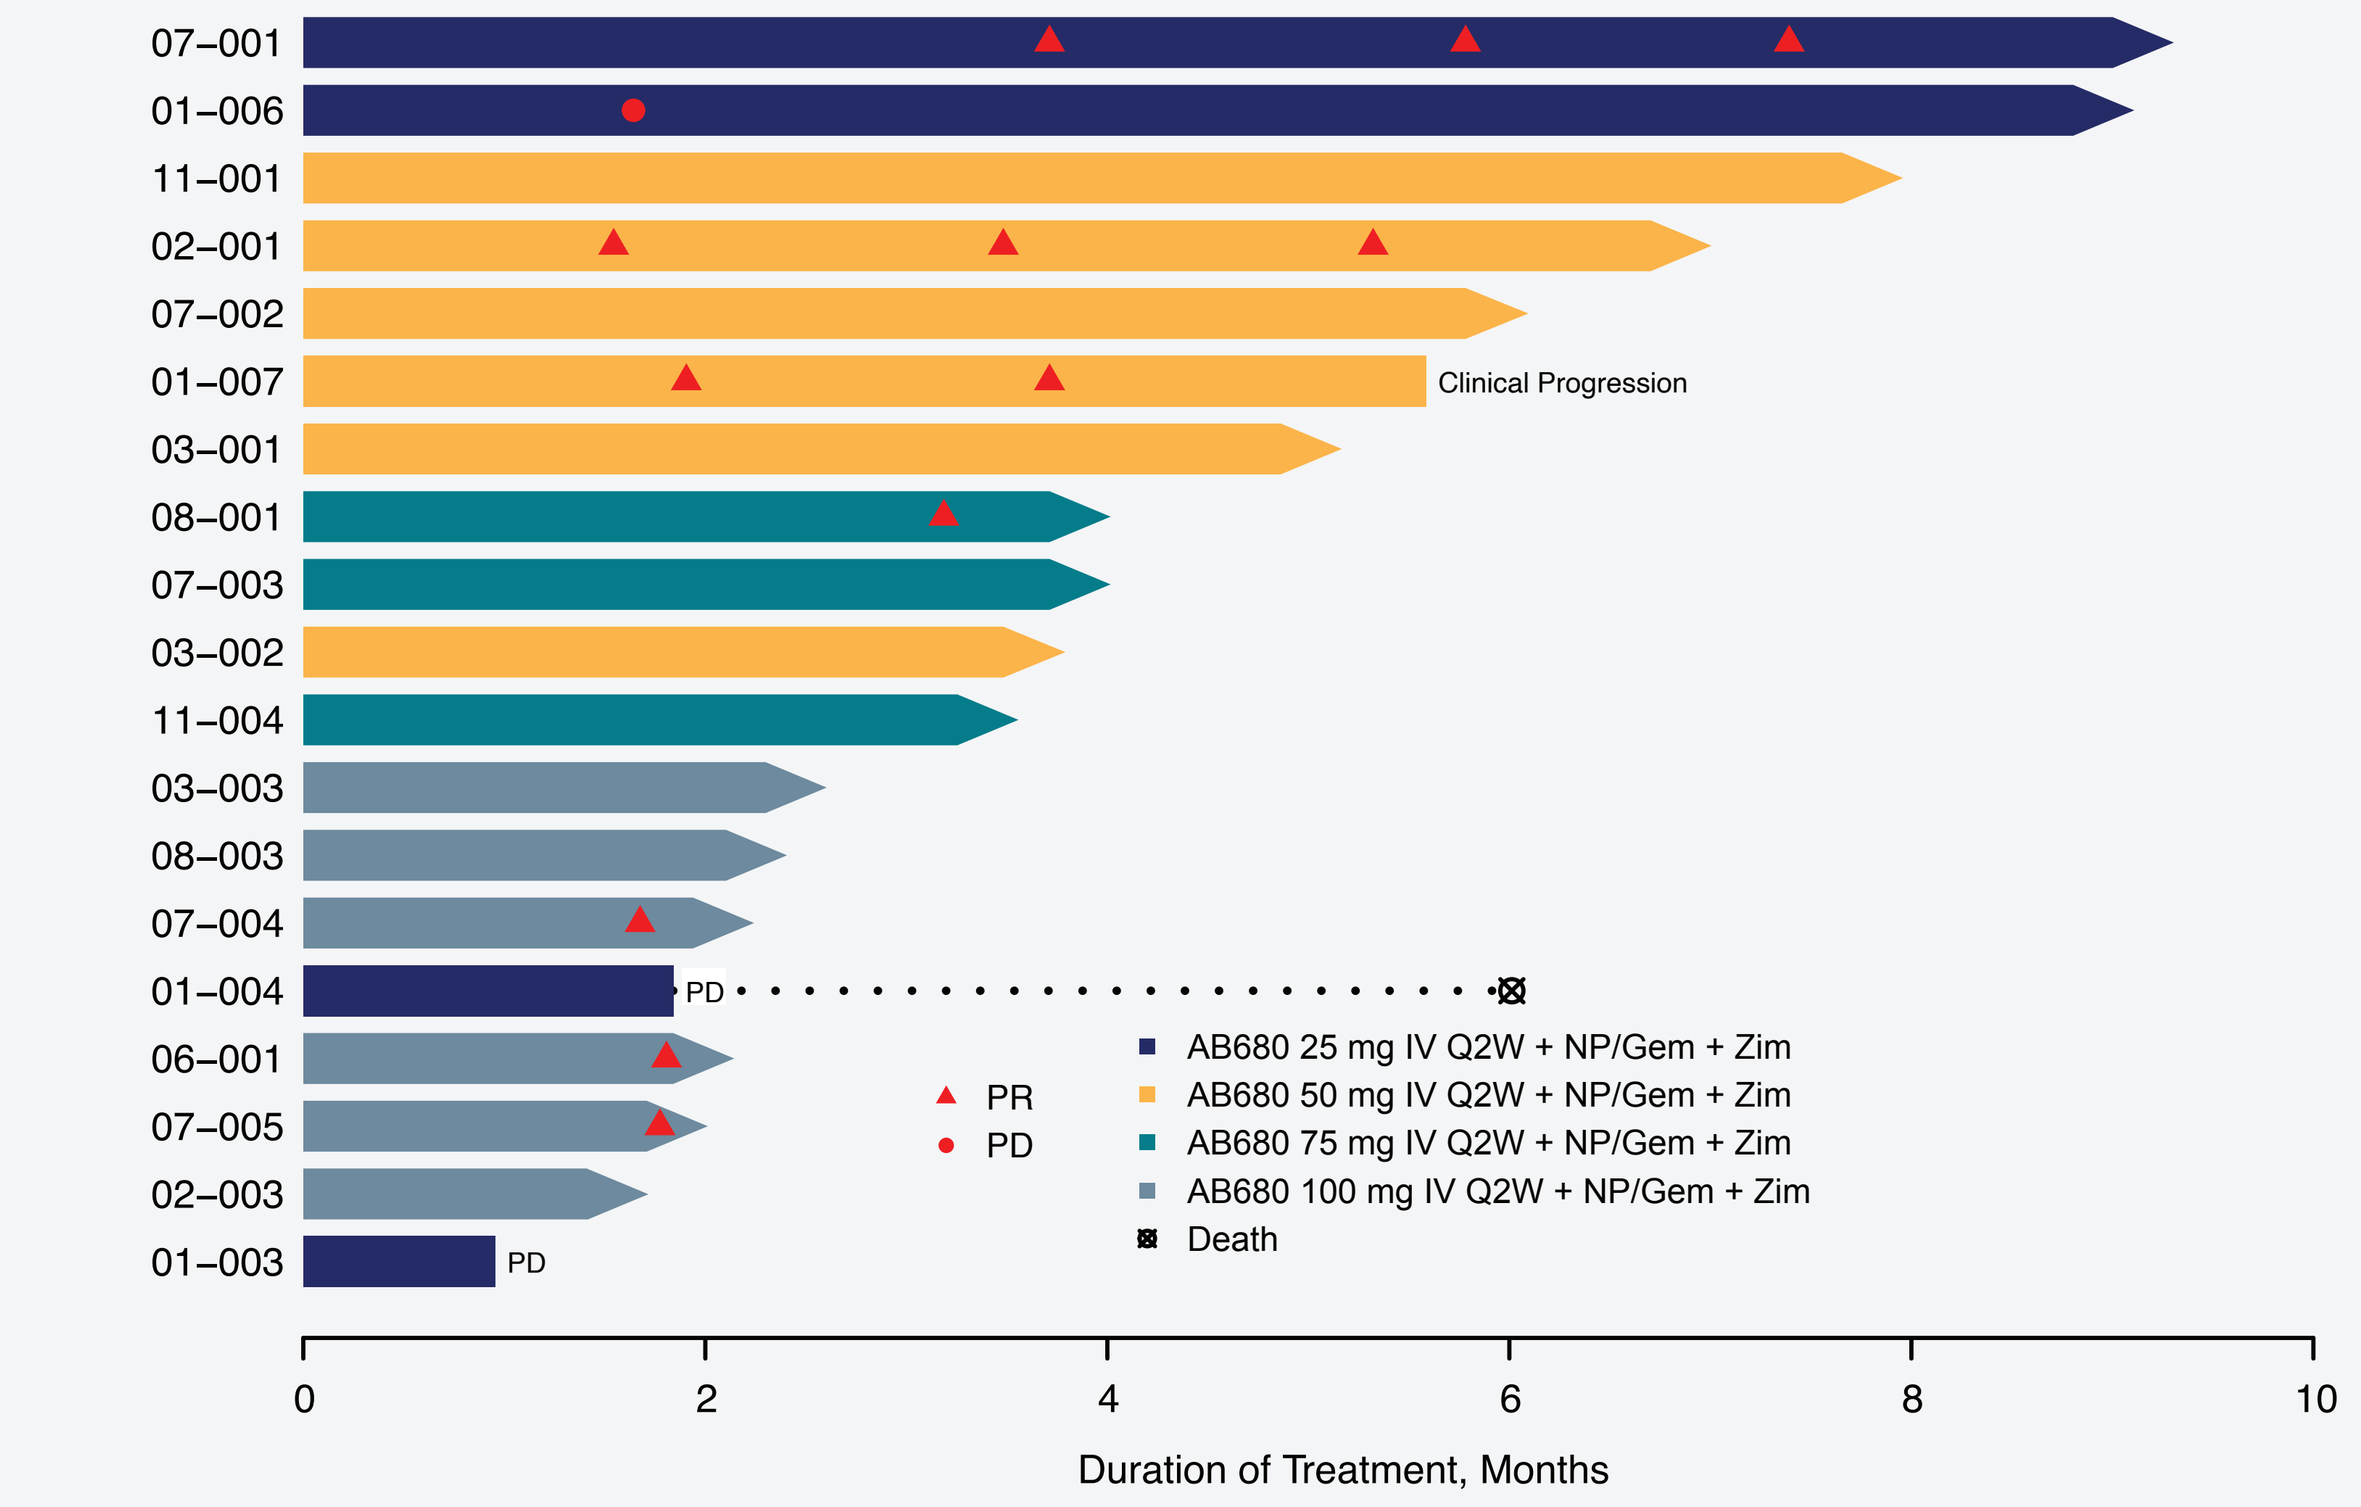
07–001
01–006
11–001
02–001
07–002
01–007
03–001
08–001
07–003
03–002
11–004
03–003
08–003
07–004
01–004
06–001
07–005
02–003
01–003
Clinical Progression
PD
PD
PR
PD
AB680 25 mg IV Q2W + NP/Gem + Zim
AB680 50 mg IV Q2W + NP/Gem + Zim
AB680 75 mg IV Q2W + NP/Gem + Zim
AB680 100 mg IV Q2W + NP/Gem + Zim
Death
0
2
4
6
8
10
Duration of Treatment, Months

IV, intravenously; NP/Gem, nab-paclitaxel/gemcitabine; PD, progressive disease; PR, partial response; Q2W, every 2 weeks; Zim, zimberelimab.

- Changes in target lesions over time are shown in **Figures 5 and 6**

Figure 5. Waterfall Plot of Best Percent Change from Baseline in Sum of Target Lesions

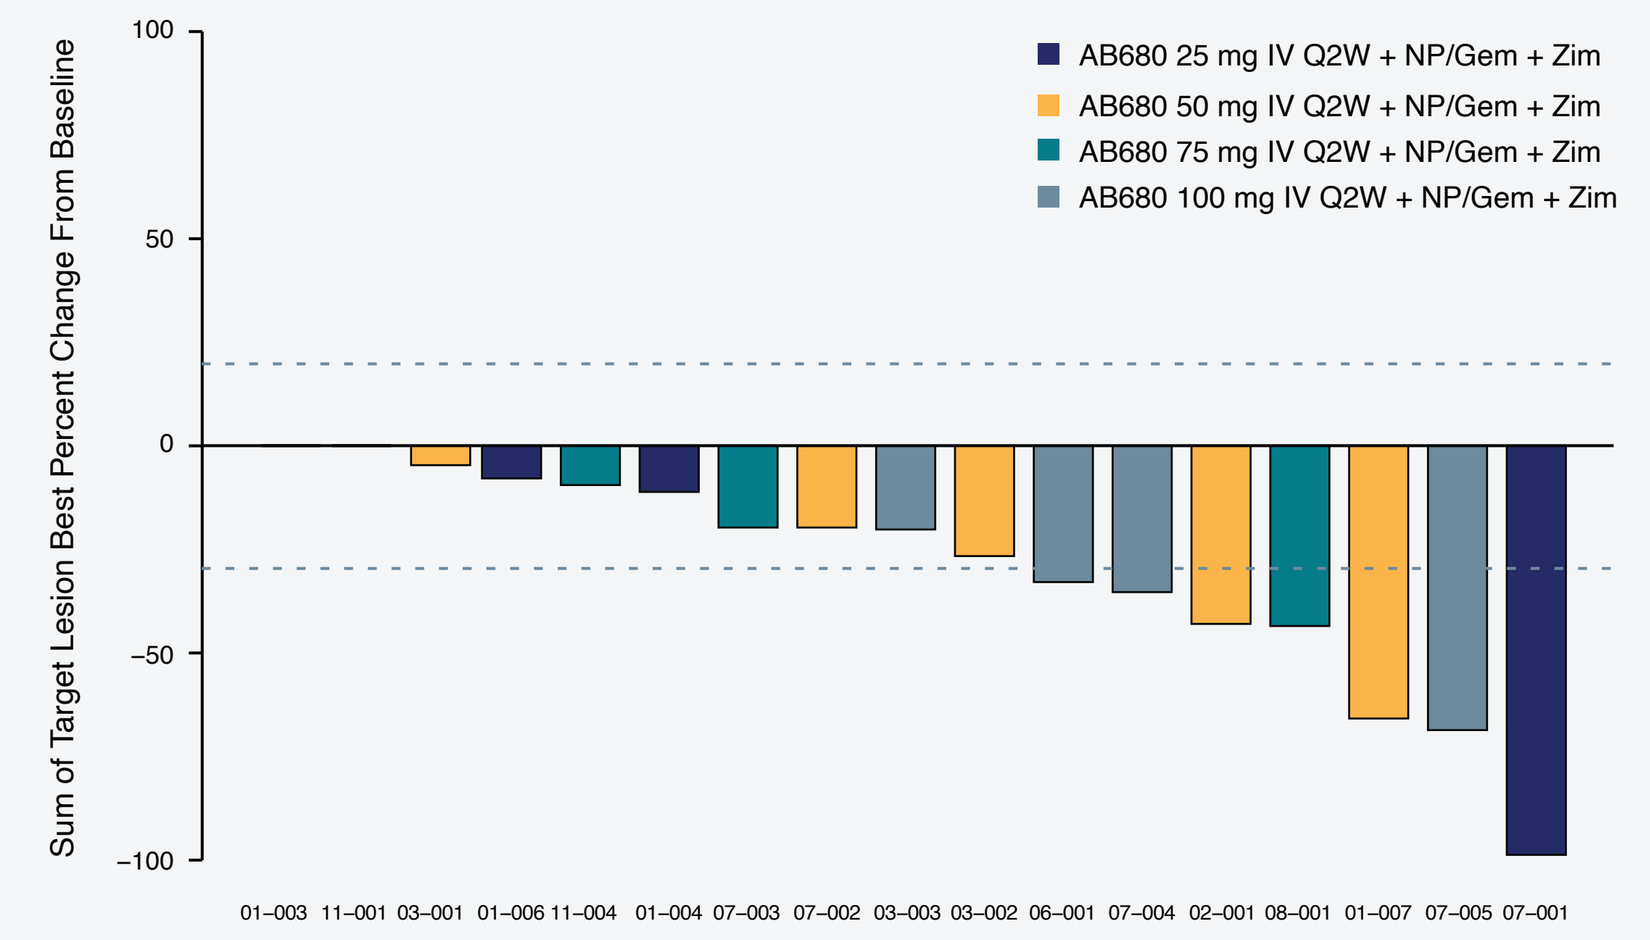
AB680 25 mg IV Q2W + NP/Gem + Zim
AB680 50 mg IV Q2W + NP/Gem + Zim
AB680 75 mg IV Q2W + NP/Gem + Zim
AB680 100 mg IV Q2W + NP/Gem + Zim
Sum of Target Lesion Best Percent Change From Baseline
01–003 11–001 03–001 01–006 11–004 01–004 07–003 07–002 03–003 03–002 06–001 07–004 02–001 08–001 01–007 07–005 07–001

IV, intravenously; NP/Gem, nab-paclitaxel/gemcitabine; Q2W, every 2 weeks; Zim, zimberelimab.

Figure 6. Spider Plot of Percent Change from Baseline in Sum of Target Lesions

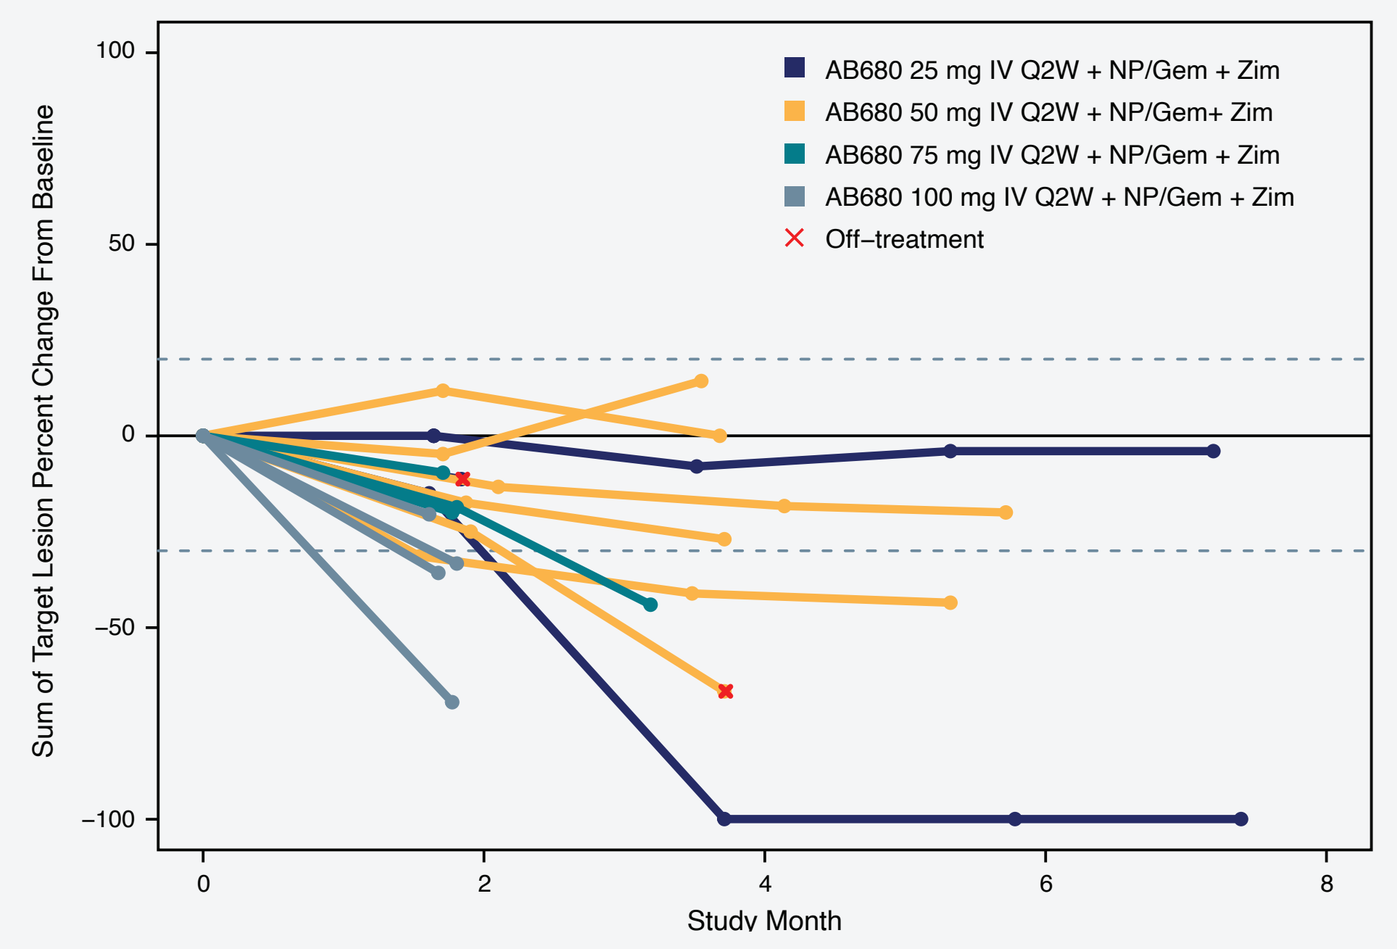
AB680 25 mg IV Q2W + NP/Gem + Zim
AB680 50 mg IV Q2W + NP/Gem+ Zim
AB680 75 mg IV Q2W + NP/Gem + Zim
AB680 100 mg IV Q2W + NP/Gem + Zim
Off–treatment
Sum of Target Lesion Percent Change From Baseline
Study Month

IV, intravenously; NP/Gem, nab-paclitaxel/gemcitabine; Q2W, every 2 weeks; Zim, zimberelimab.

- **Figure 7** shows radiologic images and CA19-9 levels over time for a 60 year old male patient who received his first AB680 dose (50 mg IV Q2W) on May 11, 2020. After Cycle 1, immunotherapy was interrupted due to Grade 2 autoimmune hepatitis, which resolved after pulse steroid therapy. Treatment with all 4 drugs resumed at the start of Cycle 3. Tumor shrinkage was observed across the patient's 3 target lesions and CA19-9 levels declined precipitously after Cycle 2.

Figure 7. Patient 02-001: Radiological Disease Evaluation and CA19-9 Levels

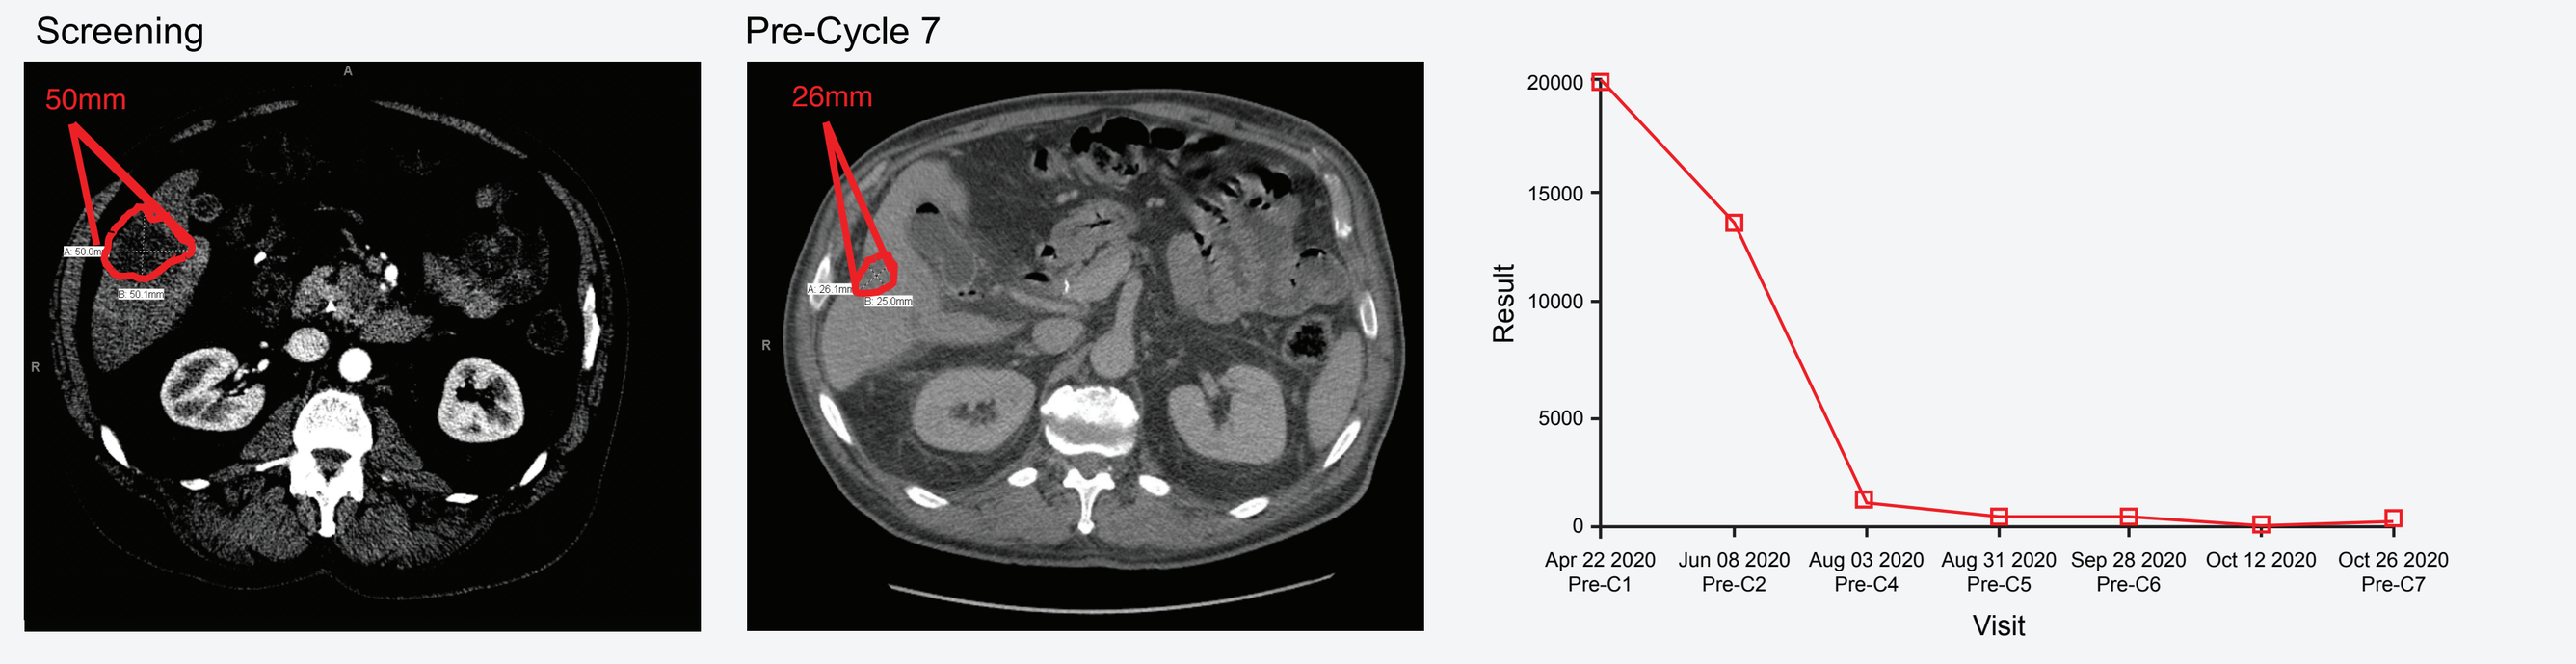
Screening
Pre-Cycle 7
50mm
26mm
Result
Apr 22 2020 Pre-C1
Jun 08 2020 Pre-C2
Aug 03 2020 Pre-C4
Aug 31 2020 Pre-C5
Sep 28 2020 Pre-C6
Oct 12 2020
Oct 26 2020 Pre-C7
Visit

C, cycle.

CONCLUSIONS

- Preliminary results from ARC-8 indicate that AB680, the first clinical-stage small-molecule CD73 inhibitor, in combination with standard-of-care chemotherapy + Zim has a manageable safety profile consistent with that expected for each agent alone and demonstrates early signals of clinical activity
- Combination treatment in 17 efficacy-evaluable patients resulted in a 41% overall response rate (7/17) which compares favorably with current standard-of-care chemotherapy (23-29% ORR for NP/Gem)
- Enrollment into the 100 mg AB680 (Cohort 4) is ongoing to inform selection of the RDE; initiation of the dose-expansion stage is expected in December 2020, followed by a 2:1 randomization evaluating AB680 + NP/Gem + Zim vs AB680 + NP/Gem pending futility analysis

ACKNOWLEDGMENTS AND DISCLOSURES

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation in this clinical trial. They would also like to thank the ARC-8 Principal Investigators and study staff for carrying out the study. GA Manji has been a consultant/advisory board member for Ardelyx, BioLineRx, and Roche and has received research funding from BioLineRx, Merck, Regeneron, and Roche/Genentech. ZA Wainberg has been a consultant/advisory board member for Array BioPharma, AstraZeneca/MedImmune, Bayer, Bristol Myers Squibb, Eli Lilly, Five Prime Therapeutics, Ipsen, Merck & Co., Merck KGaA, Macrogenics, and Novartis; and has received research funding from Five Prime Therapeutics, Merck & Co, Novartis, Pfizer, and Plexxikon. K Krishnan, N Giafis, A Udyavar, CS Quah, S Liu, W Berry, D DiRenzo, K Gerrick, and L Jin are employees of Arcus Biosciences, Inc. and may own company stock. J Bendell has been a consultant for Amgen, Apexigen, Arch Oncology, ARMO BioSciences, Array BioPharma, AstraZeneca, Bayer, Beigene, Bicycle Therapeutics, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Cerulean Pharma, Continuum Clinical, Cyteir, Daiichi Sankyo, EMD Serono, Evelo Therapeutics, Five Prime Therapeutics, FORMA Therapeutics, Genentech/Roche, Gilead Sciences, GlaxoSmithKline, Incyte, Innate Pharma, Ipsen, Janssen, Kyn Therapeutics, Leap Therapeutics, Lilly, Macrogenics, MedImmune, Merck, Merrimack, Moderna Therapeutics, Molecular Partners, Novartis, Oncogenex, OncoMed, Phoenix Biotech, Prelude Therapeutics, Relay Therapeutics, Sanofi, Seattle Genetics, Taiho Pharmaceutical, Tanabe Research, TD2, TG Therapeutics, Tizona Therapeutics, Inc., Tolero Pharmaceuticals, and Torque; and has received research funding from Abbott/AbbVie, Acerta Pharma, ADC Therapeutics, Agios, Amgen, Apexigen, Arch Oncology, Arcus Biosciences, ARMO BioSciences, Array BioPharma, Arrys Therapeutics, AstraZeneca/MedImmune, AtlasMedx, Bayer, BeiGene, Bellicum Pharmaceuticals, Bicycle Therapeutics, Blueprint Medicines, Boehringer Ingelheim, Boston Biomedical, Bristol Myers Squibb, Calithera Biosciences, Celgene, Celldex, Cyteir, CytomX Therapeutics, Daiichi Sankyo, eFFECTOR Therapeutics, Eisai, EMD Serono, Evelo Therapeutics, Ipsen, Five Prime Therapeutics, FORMA Therapeutics, Forty Seven, Genentech/Roche, Gilead Sciences, GlaxoSmithKline, Gossamer Bio, Gritstone Oncology, Harpoon Therapeutics, Hutchison MediPharma, IGM, Incyte, Innate Pharma, ImClone Systems, Jacobio, Janssen, Kolltan Pharmaceuticals, Leap Therapeutics, Lilly, MabSpace Biosciences, Macrogenics, Marshall Edwards, Merck, Merrimack, Mersana, Merus, Millenium Pharmaceuticals, Molecular Partners, Nektar, NGM Biopharmaceuticals, Novartis, Novocure, Numab, Oncogenex, OncoMed, Onyx, Pfizer, Phoenix Biotech, Pieris Pharmaceuticals, Prelude Therapeutics, Relay Therapeutics, Repare Therapeutics, Revolution Medicines, Rgenix, Sanofi, Scholar Rock, Seattle Genetics, Shattuck Labs, Sierra Oncology, Sorrento Therapeutics, Stem CentRx, SynDevRx, Synthorx, Taiho Pharmaceutical, Takeda, Tarveda Therapeutics, Tempest Therapeutics, TG Therapeutics, Tizona Therapeutics, Inc., Torque, TRACON Pharma, Treadwell Therapeutics, Tyrogenex, Unum Therapeutic, Vyriad, and Zymeworks. Medical writing support was provided by Amanda Martin, PhD, of Medical Expressions (Chicago, IL) and was funded by Arcus Biosciences, Inc.

REFERENCES

1. Martins I, et al. *Cell Cycle*. 2009;8(22):3723-3728.
2. Vijayan D, et al. *Nat Rev Cancer.* 2017;17(12):709-724.
3. Cekic C, et al. *Nat Rev Immunol.* 2016;16(3):177-192.
4. Gao ZG and Jacobson KA. *Int J Mol Sci.* 2019;20(20):5139.
5. Bendell J, et al. *ASCO GI 2020.* Abstract TPS788.
6. Ashok D, et al. *SITC 2019*. Abstract P379.
7. Von Hoff DD, et al. *N Eng J Med*. 2013;369(18):1691-1703.
8. Orth M, et al. *Radiat Oncol.* 2019;14(1):141.
9. Singh RR and O'Reilly EM. *Drugs*. 2020;80(7):647-669.
10. Wainberg ZA, et al. *Clin Cancer Res*. 2020;26(18):4814-4822.
11. Qian ZR, et al. *JAMA Oncol*. 2018;4(3):e173420.
12. Udyavar A, et al., unpublished data.

Corresponding Author: Cheng Seok Quah (cquah@arcusbio.com)

ASCO GI Virtual Congress 2021