EAACI Congress 2025
June 13-16th , Glasgow, UK
Thematic Poster Session 7 1
Poster Board # D3.411

Targeting MRGPRX2 for the Treatment of Mast Cell-Driven Diseases

Nina K. Serwas, Diane Nathaniel, Clement Opoku-Temeng, Jas Singh, Venkatesh Kattamuri, Joice Thomas, Ruchira Mirji, Ehesan U. Sharif, Manmohan R. Leleti, Matthew J. Walters, Kelsey E. Sivick, Rustom Falahati

Arcus Biosciences, Inc., Hayward, CA USA

Background

- ❖ Mas-related G protein-coupled receptor X2 (**MRGPRX2**) is a mast cell-specific G protein-coupled receptor (GPCR) that triggers robust mast cell activation through an IgE-independent mechanism[1,2].
- ❖ MRGPRX2 sits at the neuro-immune interface and can be activated by numerous endogenous agonists including neuropeptides such as Substance P[3].
- ❖ MRGPRX2 has an emerging role in driving inflammatory dermal diseases, in particular Atopic Dermatitis and Chronic Spontaneous Urticaria, prompting development of potent and selective MRGPRX2 antagonists[4-7].

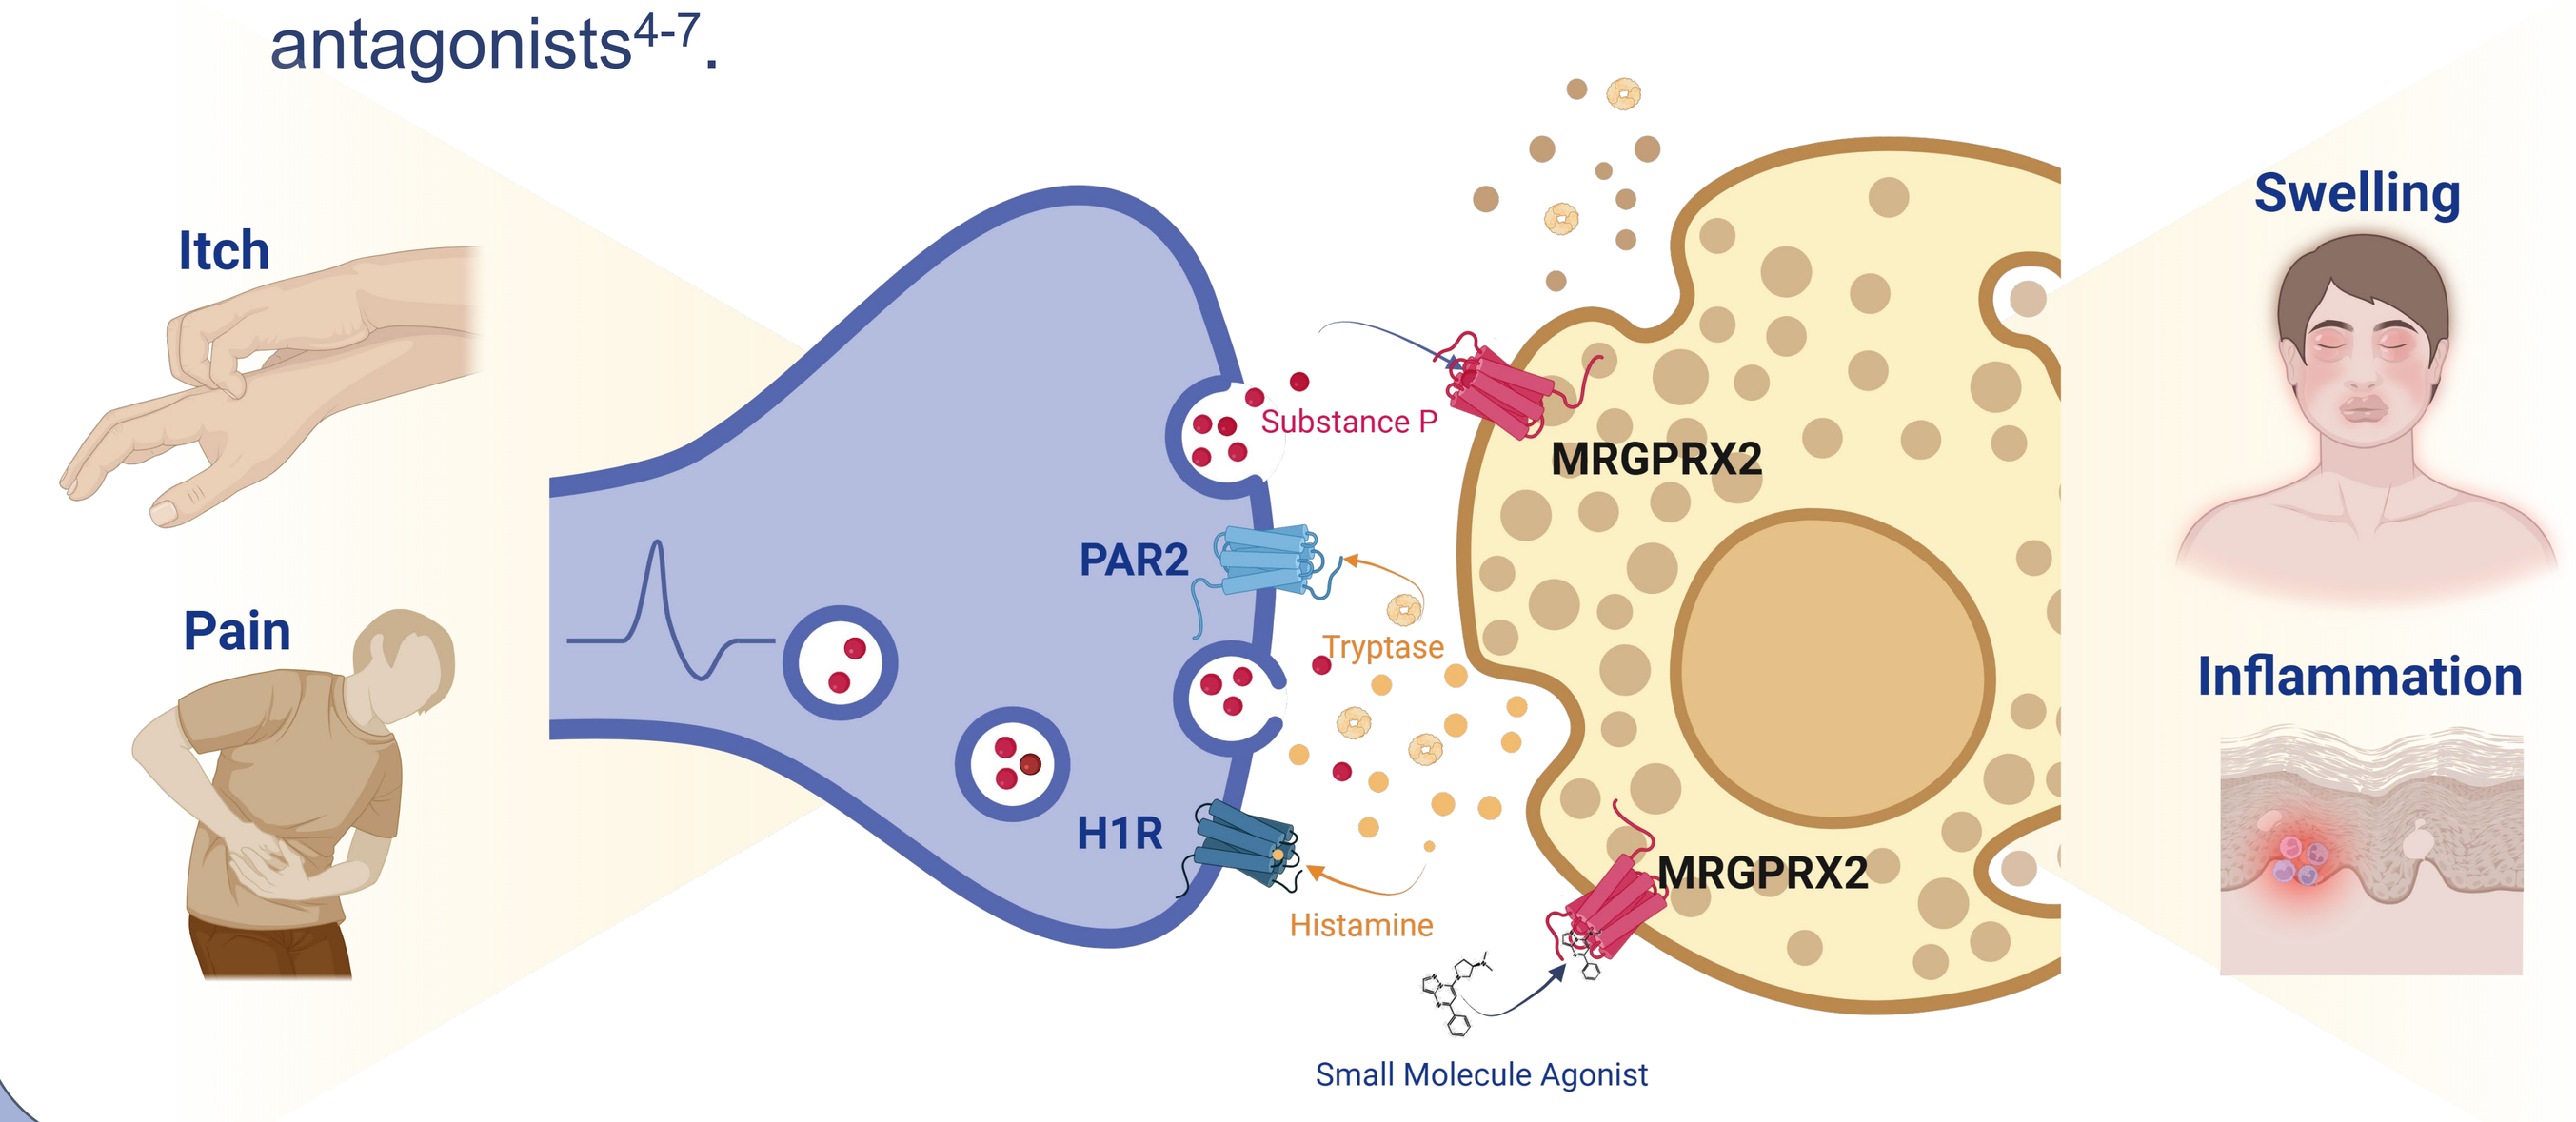
Itch
Pain
PAR2
H1R
Substance P
Tryptase
Histamine
MRGPRX2
MRGPRX2
Small Molecule Agonist
Swelling
Inflammation

Results

MRGPRX2 Surface Levels on Primary Human Skin Mast Cells and Human Mast Cell Line LAD2 are Similar

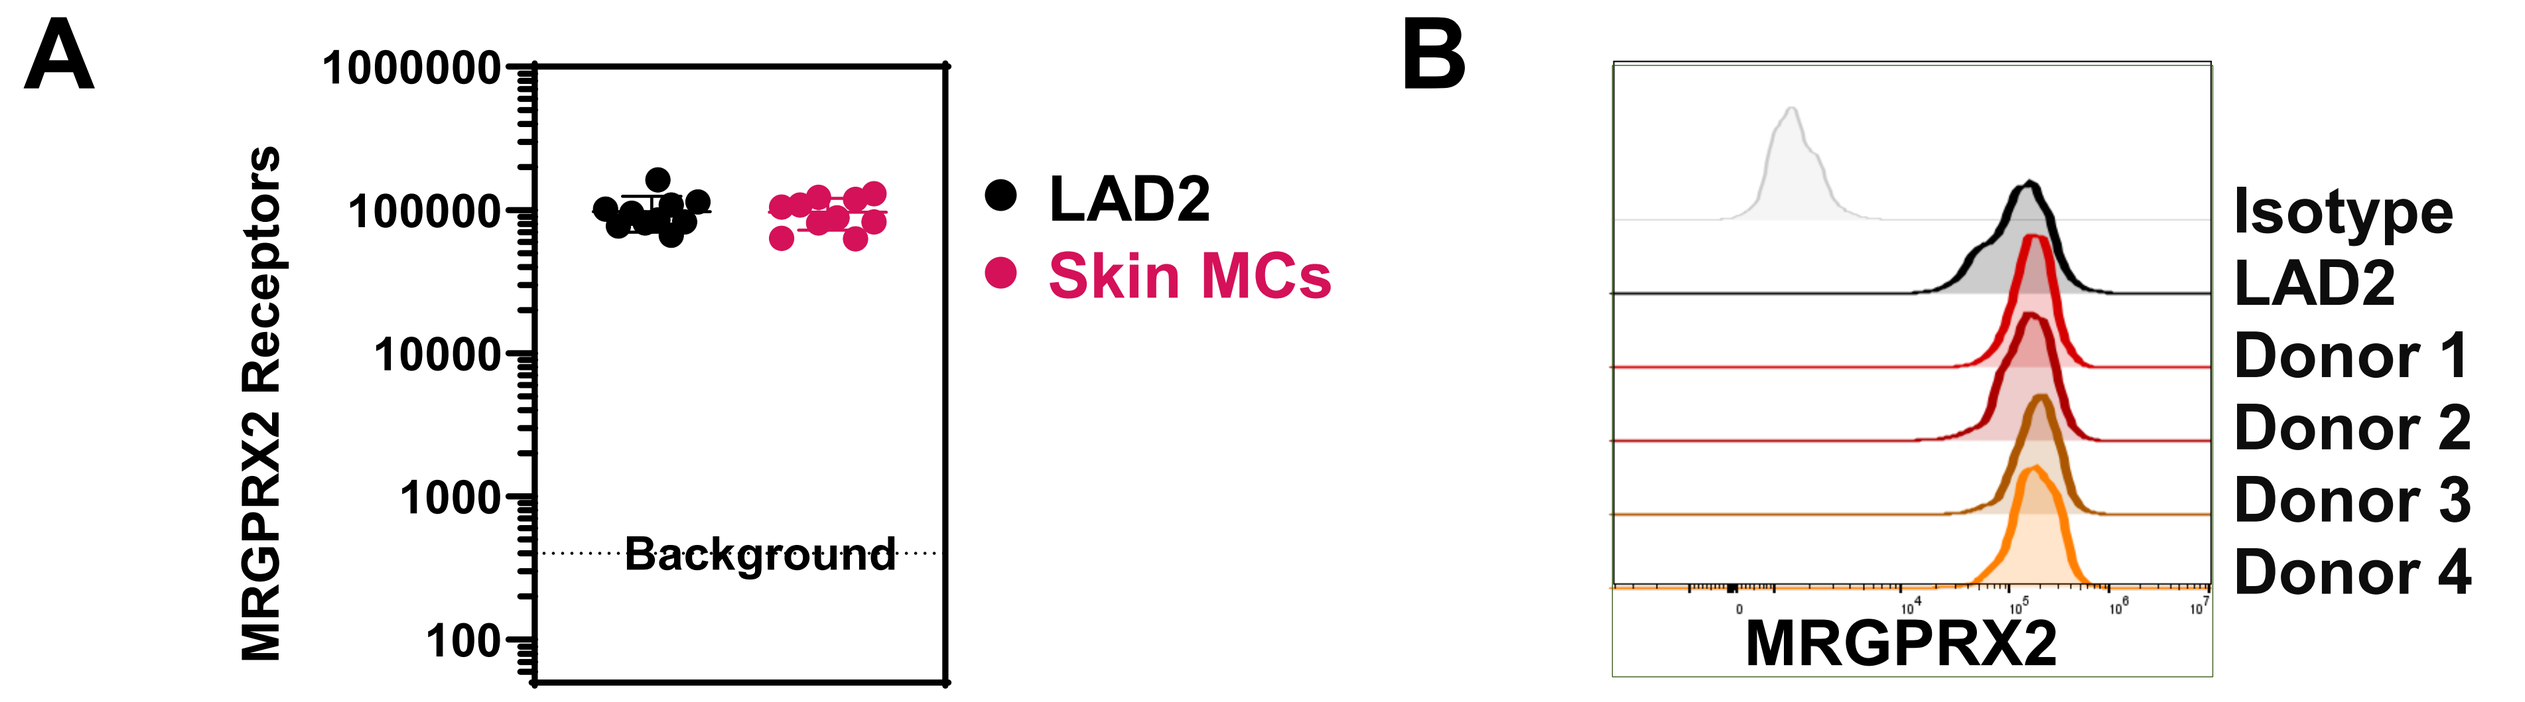
A
MRGPRX2 Receptors
1000000
100000
10000
1000
100
Background
LAD2
Skin MCs
B
Isotype
LAD2
Donor 1
Donor 2
Donor 3
Donor 4
MRGPRX2

Figure 1 – Primary mast cells uniformly express MRGPRX2 on their cell surface. (A) MRGPRX2 receptor density was determined by defining molecules of equivalent soluble fluorochrome (MESF) values corrected for labeling efficiency of MRGPRX2 antibody. LAD2 is a mast cell line, skin mast cells were isolated from abdominoplasties. (B) Representative histogram of skin mast cell populations from 4 different human donors and LAD2 cells.

MRGPRX2 Activation Drives Rapid and Robust Degranulation of Preformed Inflammatory Mediators From Human Mast Cells

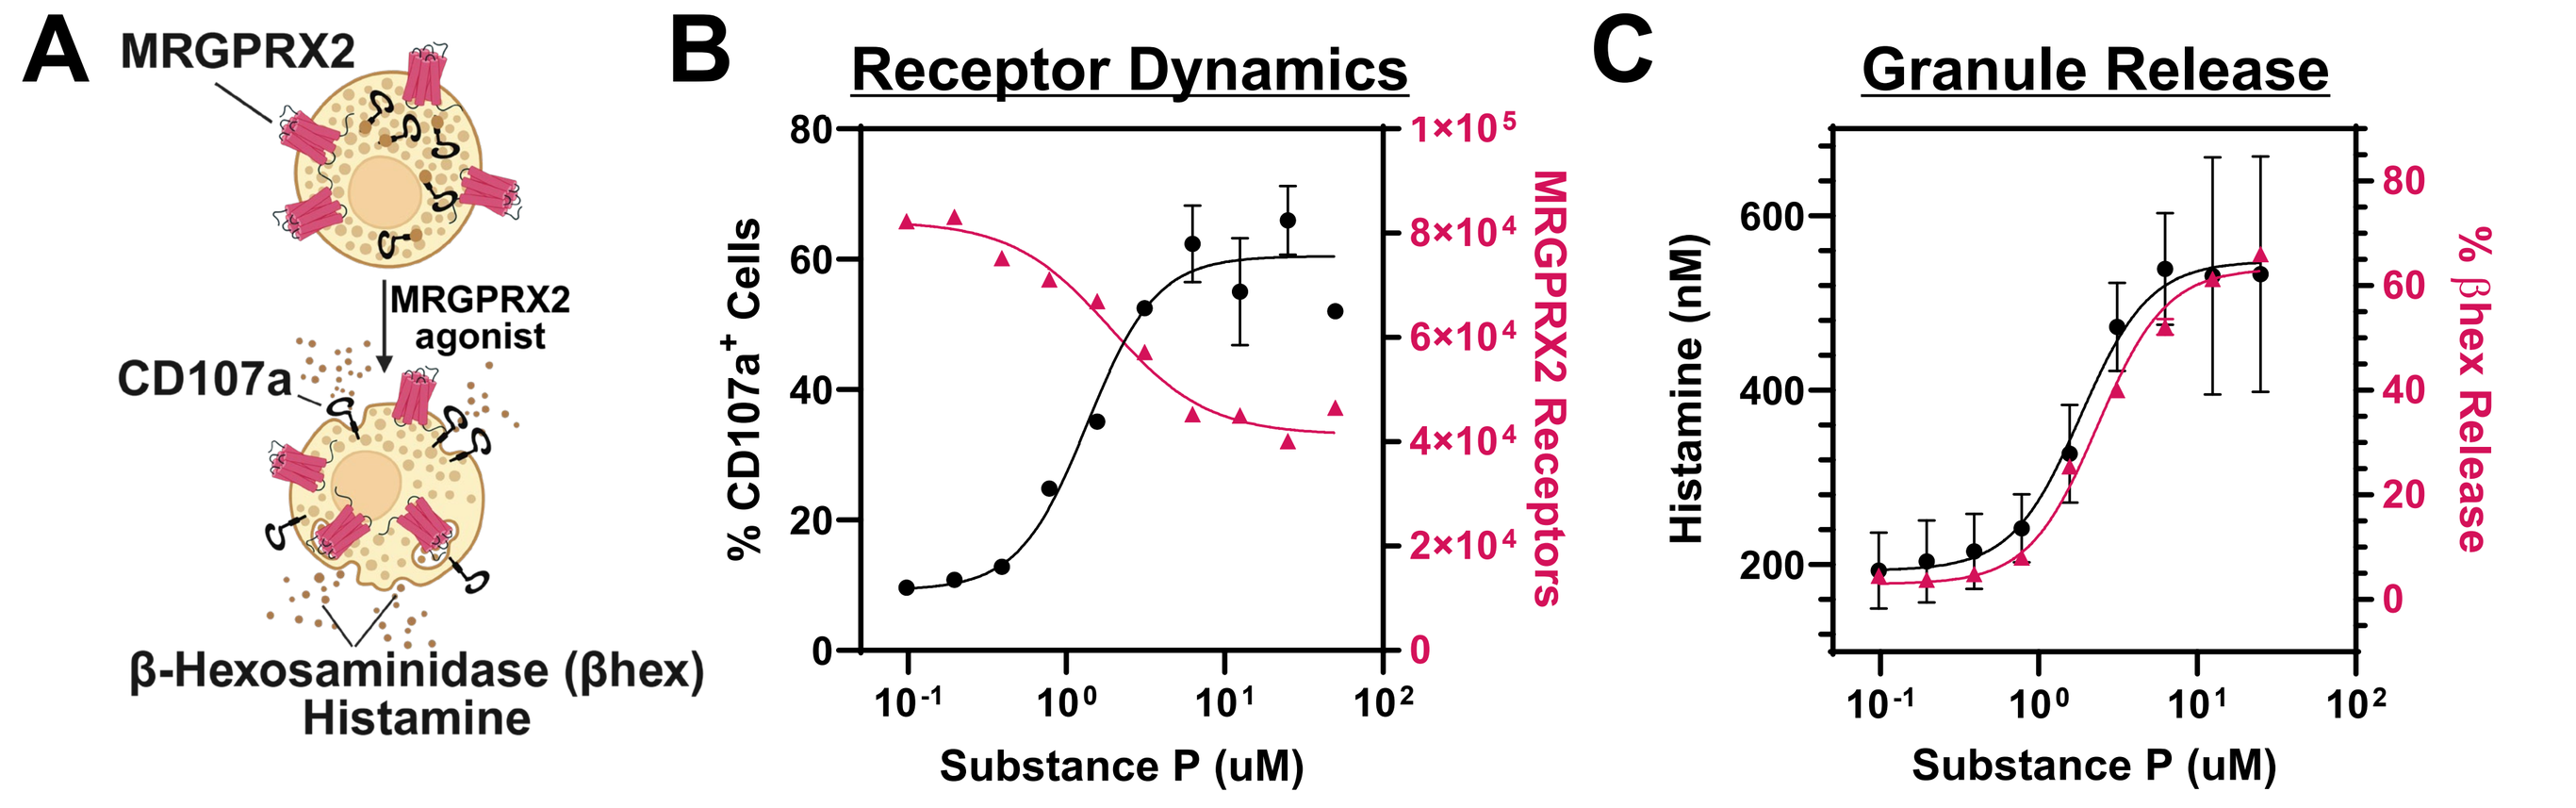
A
MRGPRX2
MRGPRX2 agonist
CD107a
β-Hexosaminidase (βhex)
Histamine
B
Receptor Dynamics
% CD107a+ Cells
MRGPRX2 Receptors
Substance P (uM)
C
Granule Release
Histamine (nM)
% βhex Release
Substance P (uM)

Figure 2 – MRGPRX2 agonist Substance P activates LAD2 cells to degranulate and release preformed inflammatory mediators from granules (histamine, βhex) with externalization of activation marker CD107a and partial internalization of MRGPRX2. (A) Schematic of readouts assessed after MRGPRX2 activation. (B) CD107a is transported to the cell surface whereas MRGPRX2 is partially internalized as detected by flow cytometry. (C) Quantification of histamine by enzyme immunoassay and βhex by a plate-based colorimetric readout in cell supernatants.

Results

MRGPRX2 Biology is Potentiated by IgE Receptor Activation in Atopic Settings

Table 1 – MRGPRX2 is activated by an array of endogenous and exogenous agonists at high concentrations.

Agonist	Classification	Source	LAD2 Degranulation EC_{50} ± SD (μM) n= # of experiments
PAMP-12	Neuropeptide	Endogenous	0.25 ± 0.10 $^{n=7}$
Substance P	Neuropeptide	Endogenous	1.58 ± 0.61 $^{n=33}$
Somatostatin 14	Neuropeptide	Endogenous	2.96 ± 0.96 $^{n=5}$
CXCL14	Chemokine	Endogenous	1.29 ± 0.76 $^{n=2}$
(R)-ZINC-3573	Synthetic	Exogenous	3.11 ± 0.80 $^{n=7}$
Ciprofloxacin	Antibiotic	Exogenous	465 ± 159 $^{n=4}$

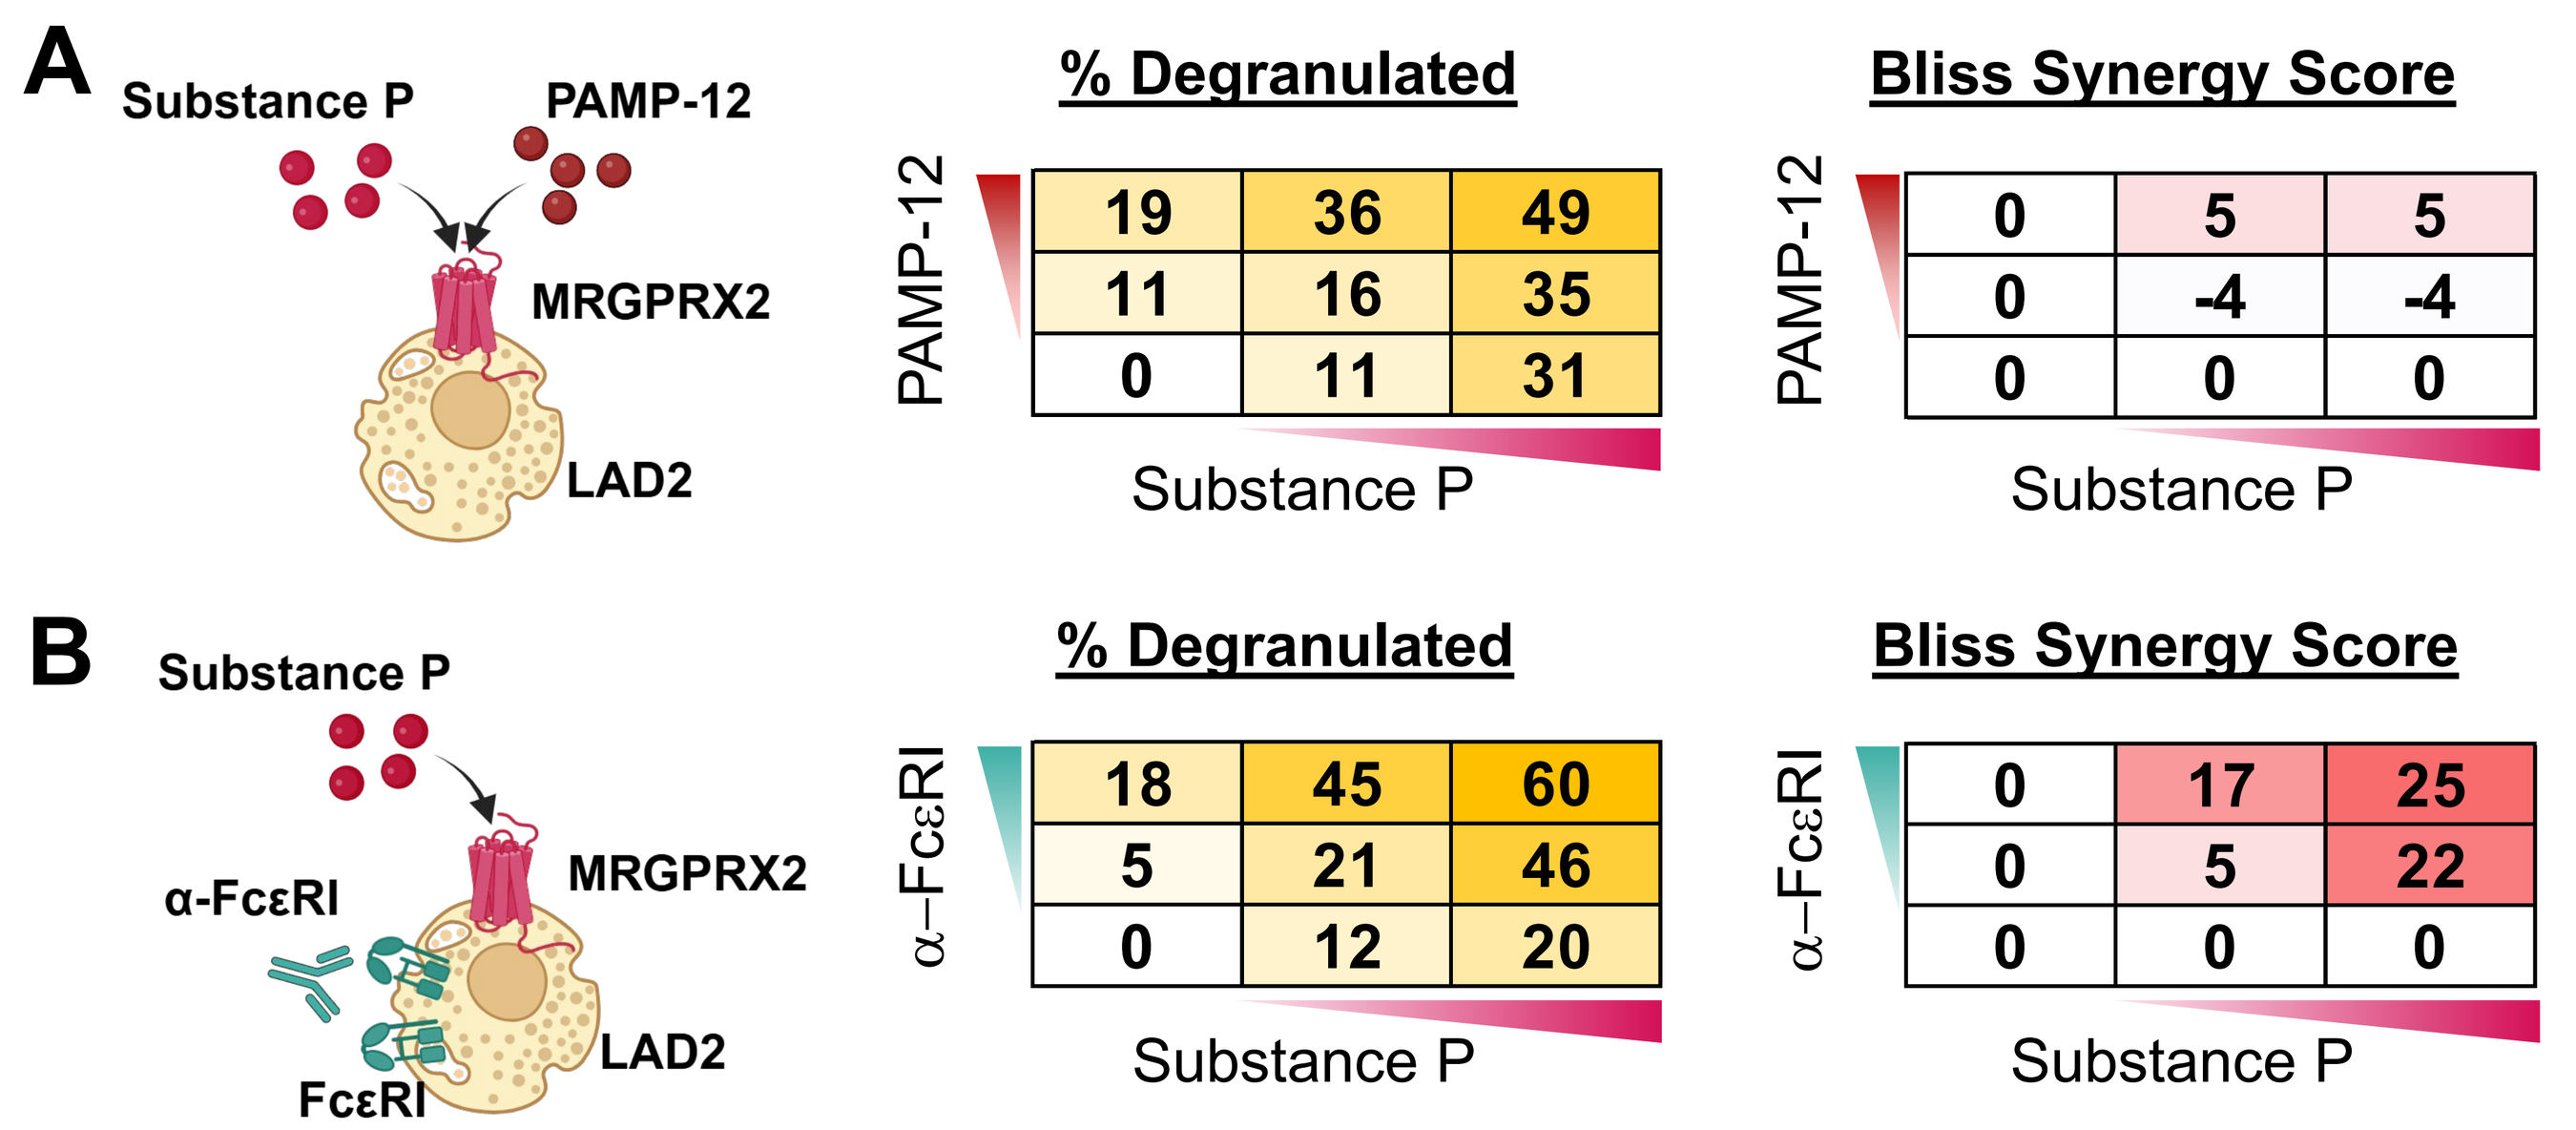
A
Substance P
PAMP-12
MRGPRX2
LAD2
% Degranulated
PAMP-12
19 36 49
11 16 35
0 11 31
Substance P
Bliss Synergy Score
PAMP-12
0 5 5
0 -4 -4
0 0 0
Substance P
B
Substance P
α-FcεRI
FcεRI
MRGPRX2
LAD2
% Degranulated
α–FcεRI
18 45 60
5 21 46
0 12 20
Substance P
Bliss Synergy Score
α–FcεRI
0 17 25
0 5 22
0 0 0
Substance P

Figure 3 – MRGPRX2 agonists are additive whereas combination of MRGPRX2 and IgE receptor activation is synergistic. (A) LAD2 cells were stimulated with increasing concentrations of Substance P and/or PAMP-12 and analyzed for βhex release (data below EC_{50} shown). (B) High-affinity IgE receptors (FcεRI) were crosslinked with α-FcεRIα antibody in the presence or absence of MRGPRX2 stimulation with Substance P. Bliss Synergy Scores show synergy (score >10) for combined MRGPRX2 and IgE receptor activation.

MRGPRX2 Mediates a Distinct Activation Gene Signature in Human Mast Cells

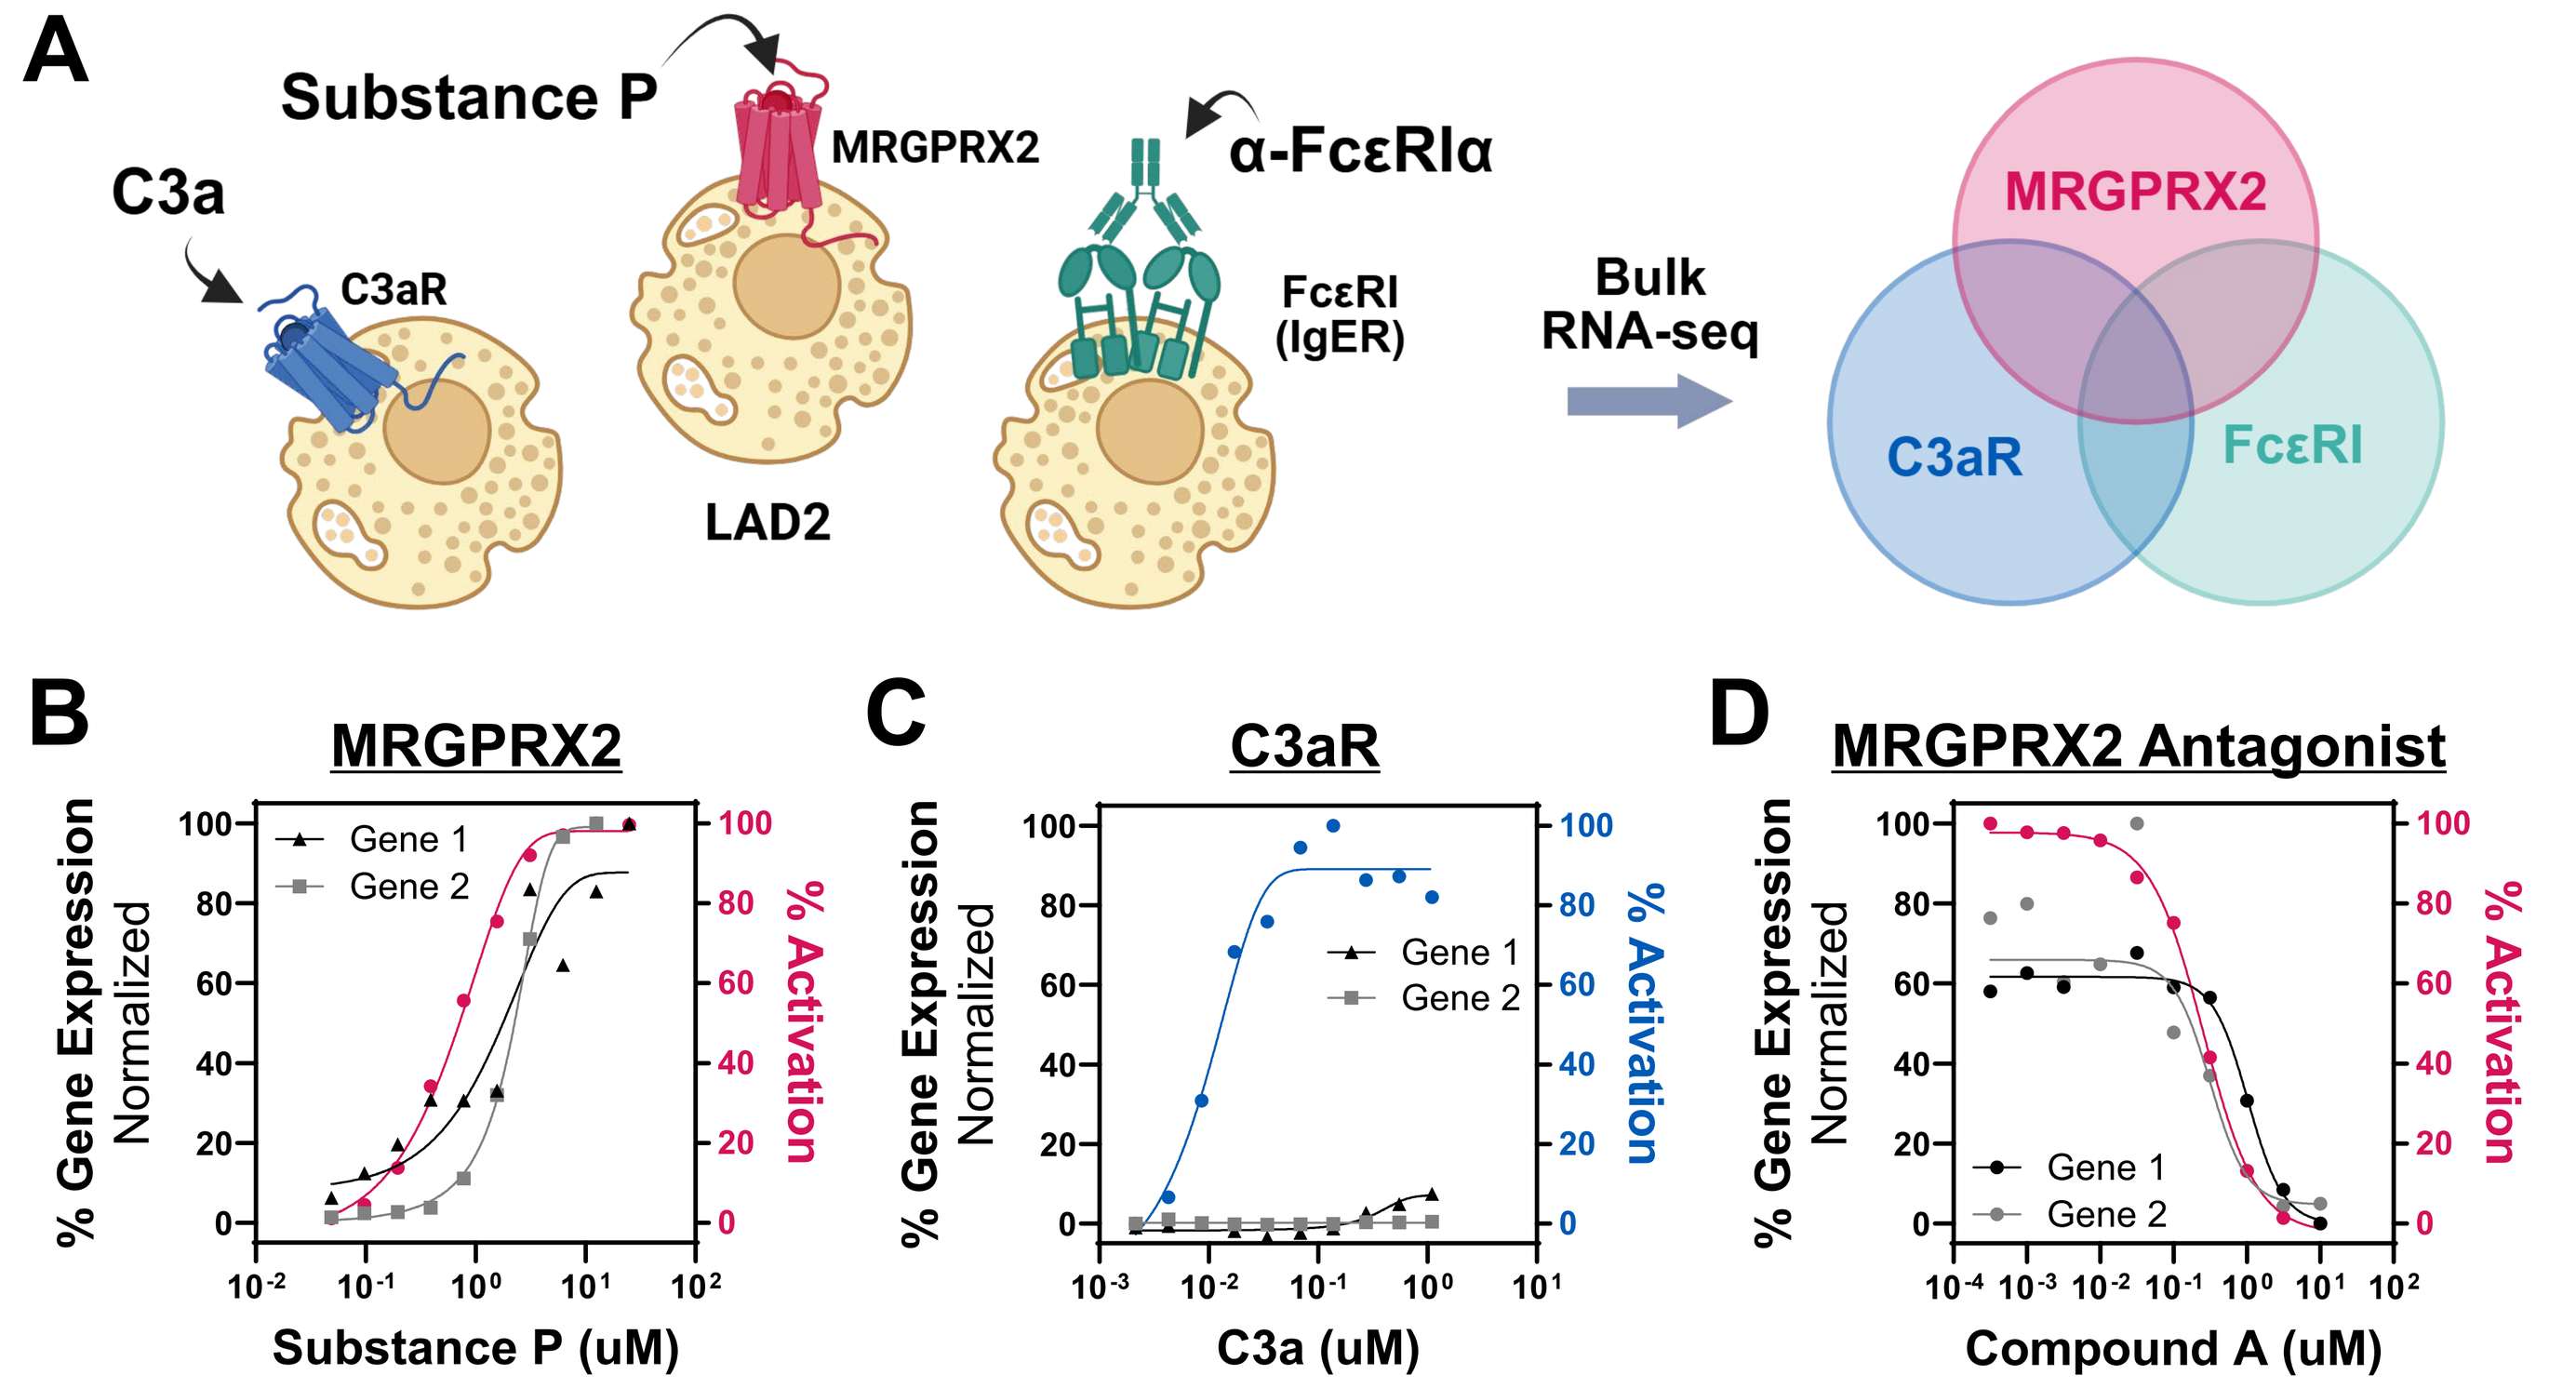
A
Substance P
C3a
C3aR
MRGPRX2
α-FcεRIα
FcεRI (IgER)
LAD2
Bulk RNA-seq
MRGPRX2
C3aR
FcεRI
B
MRGPRX2
% Gene Expression Normalized
% Activation
Gene 1
Gene 2
Substance P (uM)
C
C3aR
C3a (uM)
D
MRGPRX2 Antagonist
Compound A (uM)

Figure 4 – MRGPRX2 activation causes transcription of a unique gene signature. (A) Schematic of experimental setup to identify MRGPRX2-specific transcriptional changes. In brief, LAD2 cells were stimulated through MRGPRX2, C3a receptor (C3aR), or high-affinity IgE receptor (FcεRI) using EC_{90} of each respective ligand/agonist for up to 72 hours. Transcriptional changes were analyzed by bulk RNA sequencing. MRGPRX2-specific genes were determined as genes with at least 2-fold upregulation after Substance P stimulation and no upregulation after C3aR or FcεRI activation. (B) Select genes were validated (two genes are shown) after stimulation with Substance P and follow similar dynamics as degranulation measured by surface CD107a. (C) The same genes are not upregulated after stimulation with indicated C3a concentrations. (D) MRGPRX2-specific upregulation of Gene 1 and Gene 2 following incubation with Substance P can be inhibited by pre-incubation of cells with MRGPRX2 antagonist Compound A.

Results

MRGPRX2 Antagonist Selectively Inhibits MRGPRX2-Mediated Mast Cell Degranulation

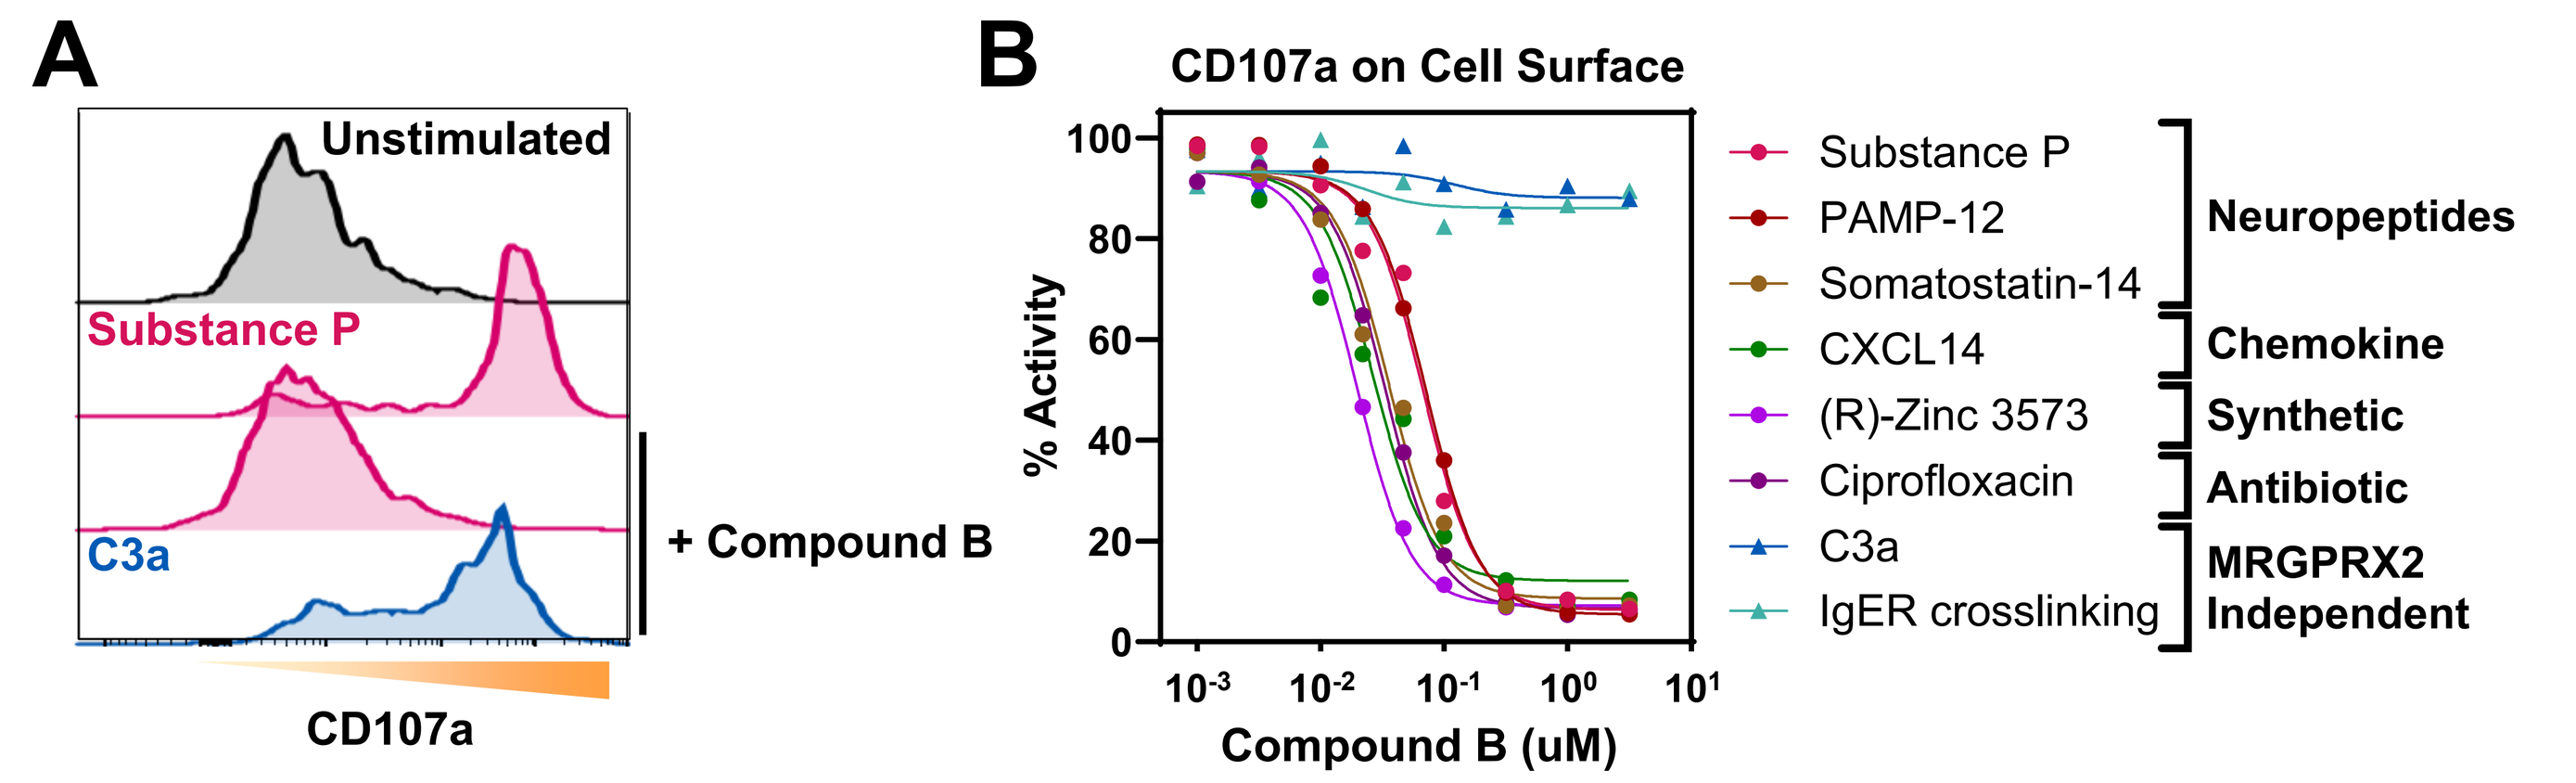
A
Unstimulated
Substance P
C3a
+ Compound B
CD107a
B
CD107a on Cell Surface
% Activity
Compound B (uM)
Substance P
PAMP-12
Somatostatin-14
CXCL14
(R)-Zinc 3573
Ciprofloxacin
C3a
IgER crosslinking
Neuropeptides
Chemokine
Synthetic
Antibiotic
MRGPRX2 Independent

Figure 5 – Preincubation of LAD2 cells with MRGPRX2 Antagonist Compound B prevents degranulation of mast cells triggered through activation by EC_{90} concentrations of all tested MRGPRX2 agonists. (A) Representative histograms and (B) normalized activity are shown. Similar results were obtained for β-hexosaminidase and histamine release.

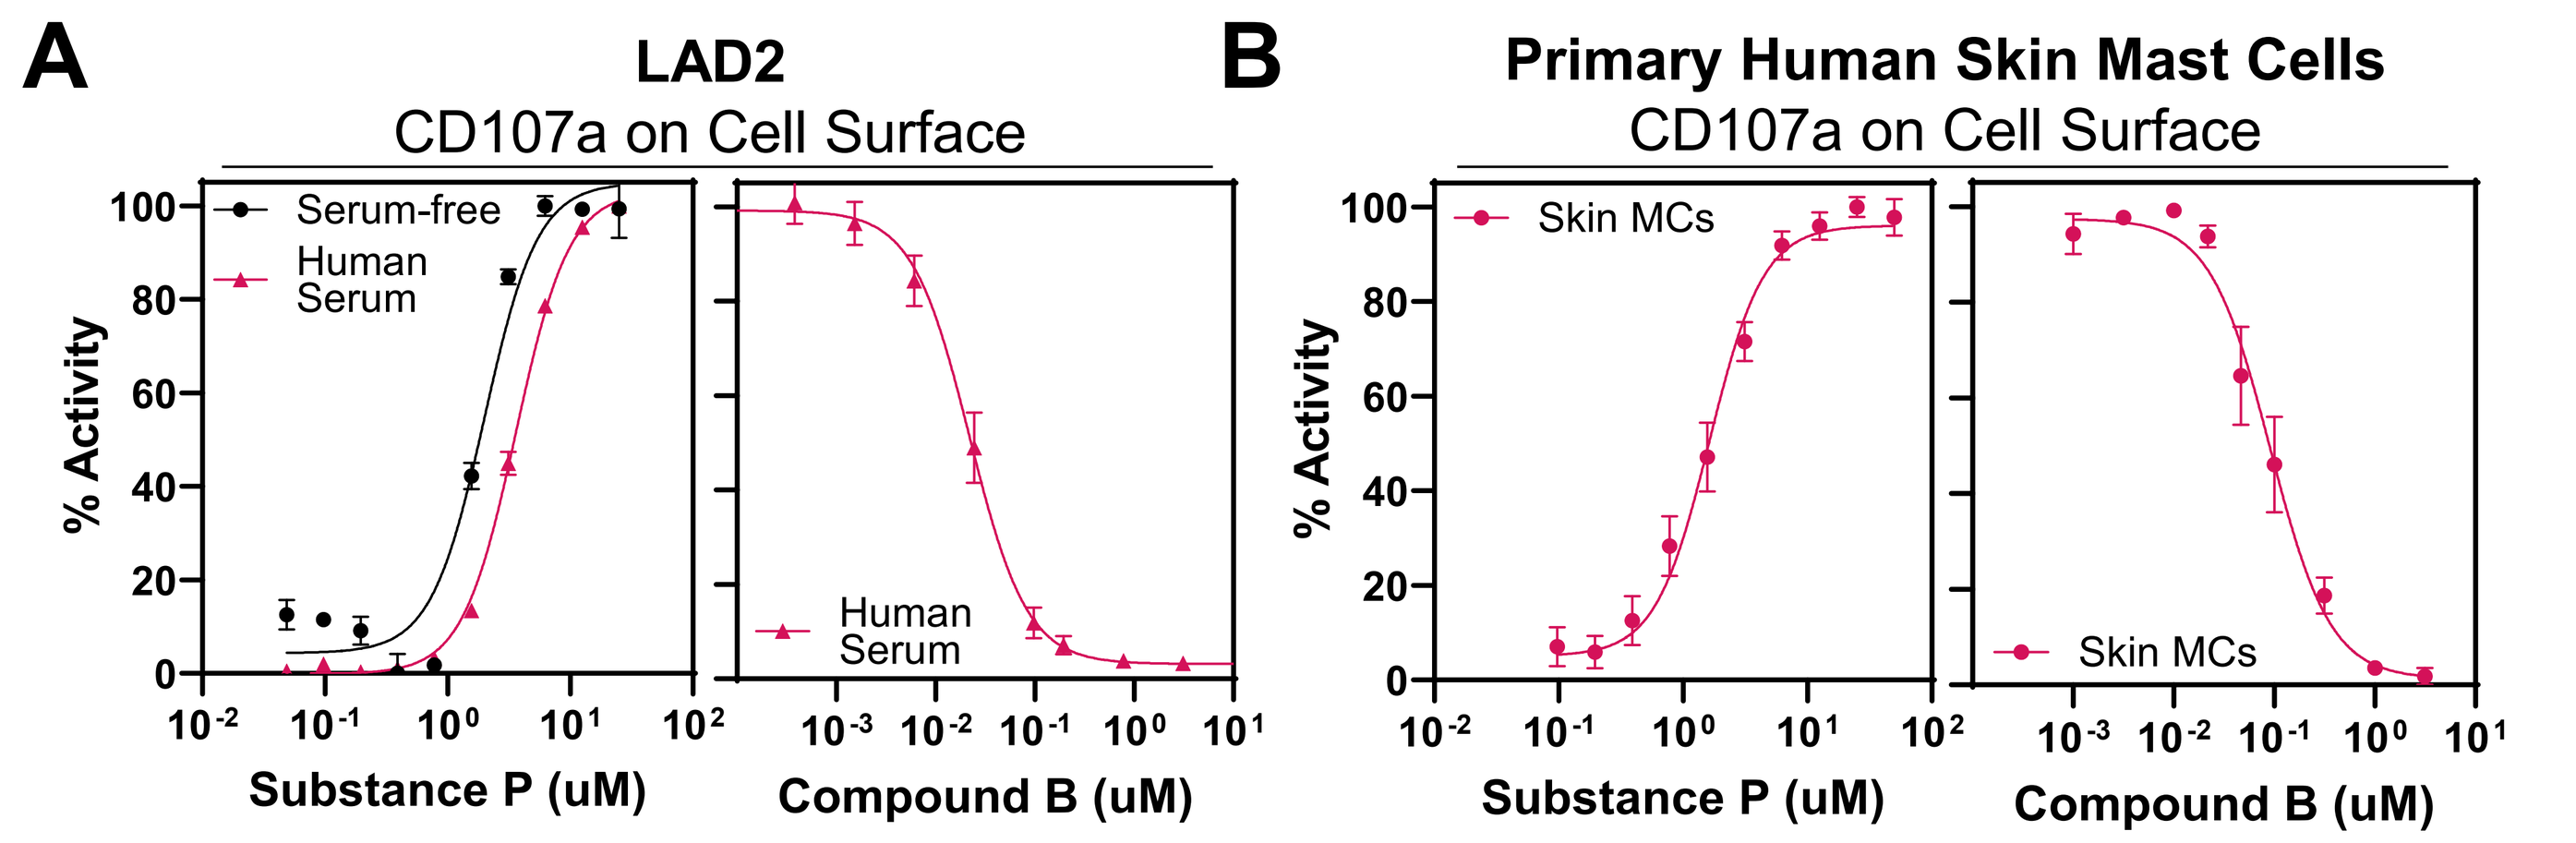
A
LAD2
CD107a on Cell Surface
% Activity
Serum-free
Human Serum
Human Serum
Substance P (uM)
Compound B (uM)
B
Primary Human Skin Mast Cells
CD107a on Cell Surface
Skin MCs
Skin MCs
Substance P (uM)
Compound B (uM)

Figure 6 – Arcus MRGPRX2 Antagonist inhibits degranulation of mast cells in other matrices and from primary sources. (A) Left: LAD2 mast cells were resuspended in serum-free medium or in human serum and stimulated with Substance P. Right: LAD2 cells were resuspended in human serum and incubated with Compound B MRGPRX2 Antagonist prior to stimulation with EC_{90} of Substance P. (B) Left: Primary skin mast cells purified from abdominoplasties were stimulated with Substance P. Right: Skin mast cells were preincubated with Compound B before stimulation with EC_{90} of Substance P. All graphs show relative CD107a levels on mast cells as analyzed by flow cytometry (mean +SEM).

Arcus MRGPRX2 Knock-In (KI) Mice Reproduce Human Responses to MRGPRX2 Agonist and Antagonist

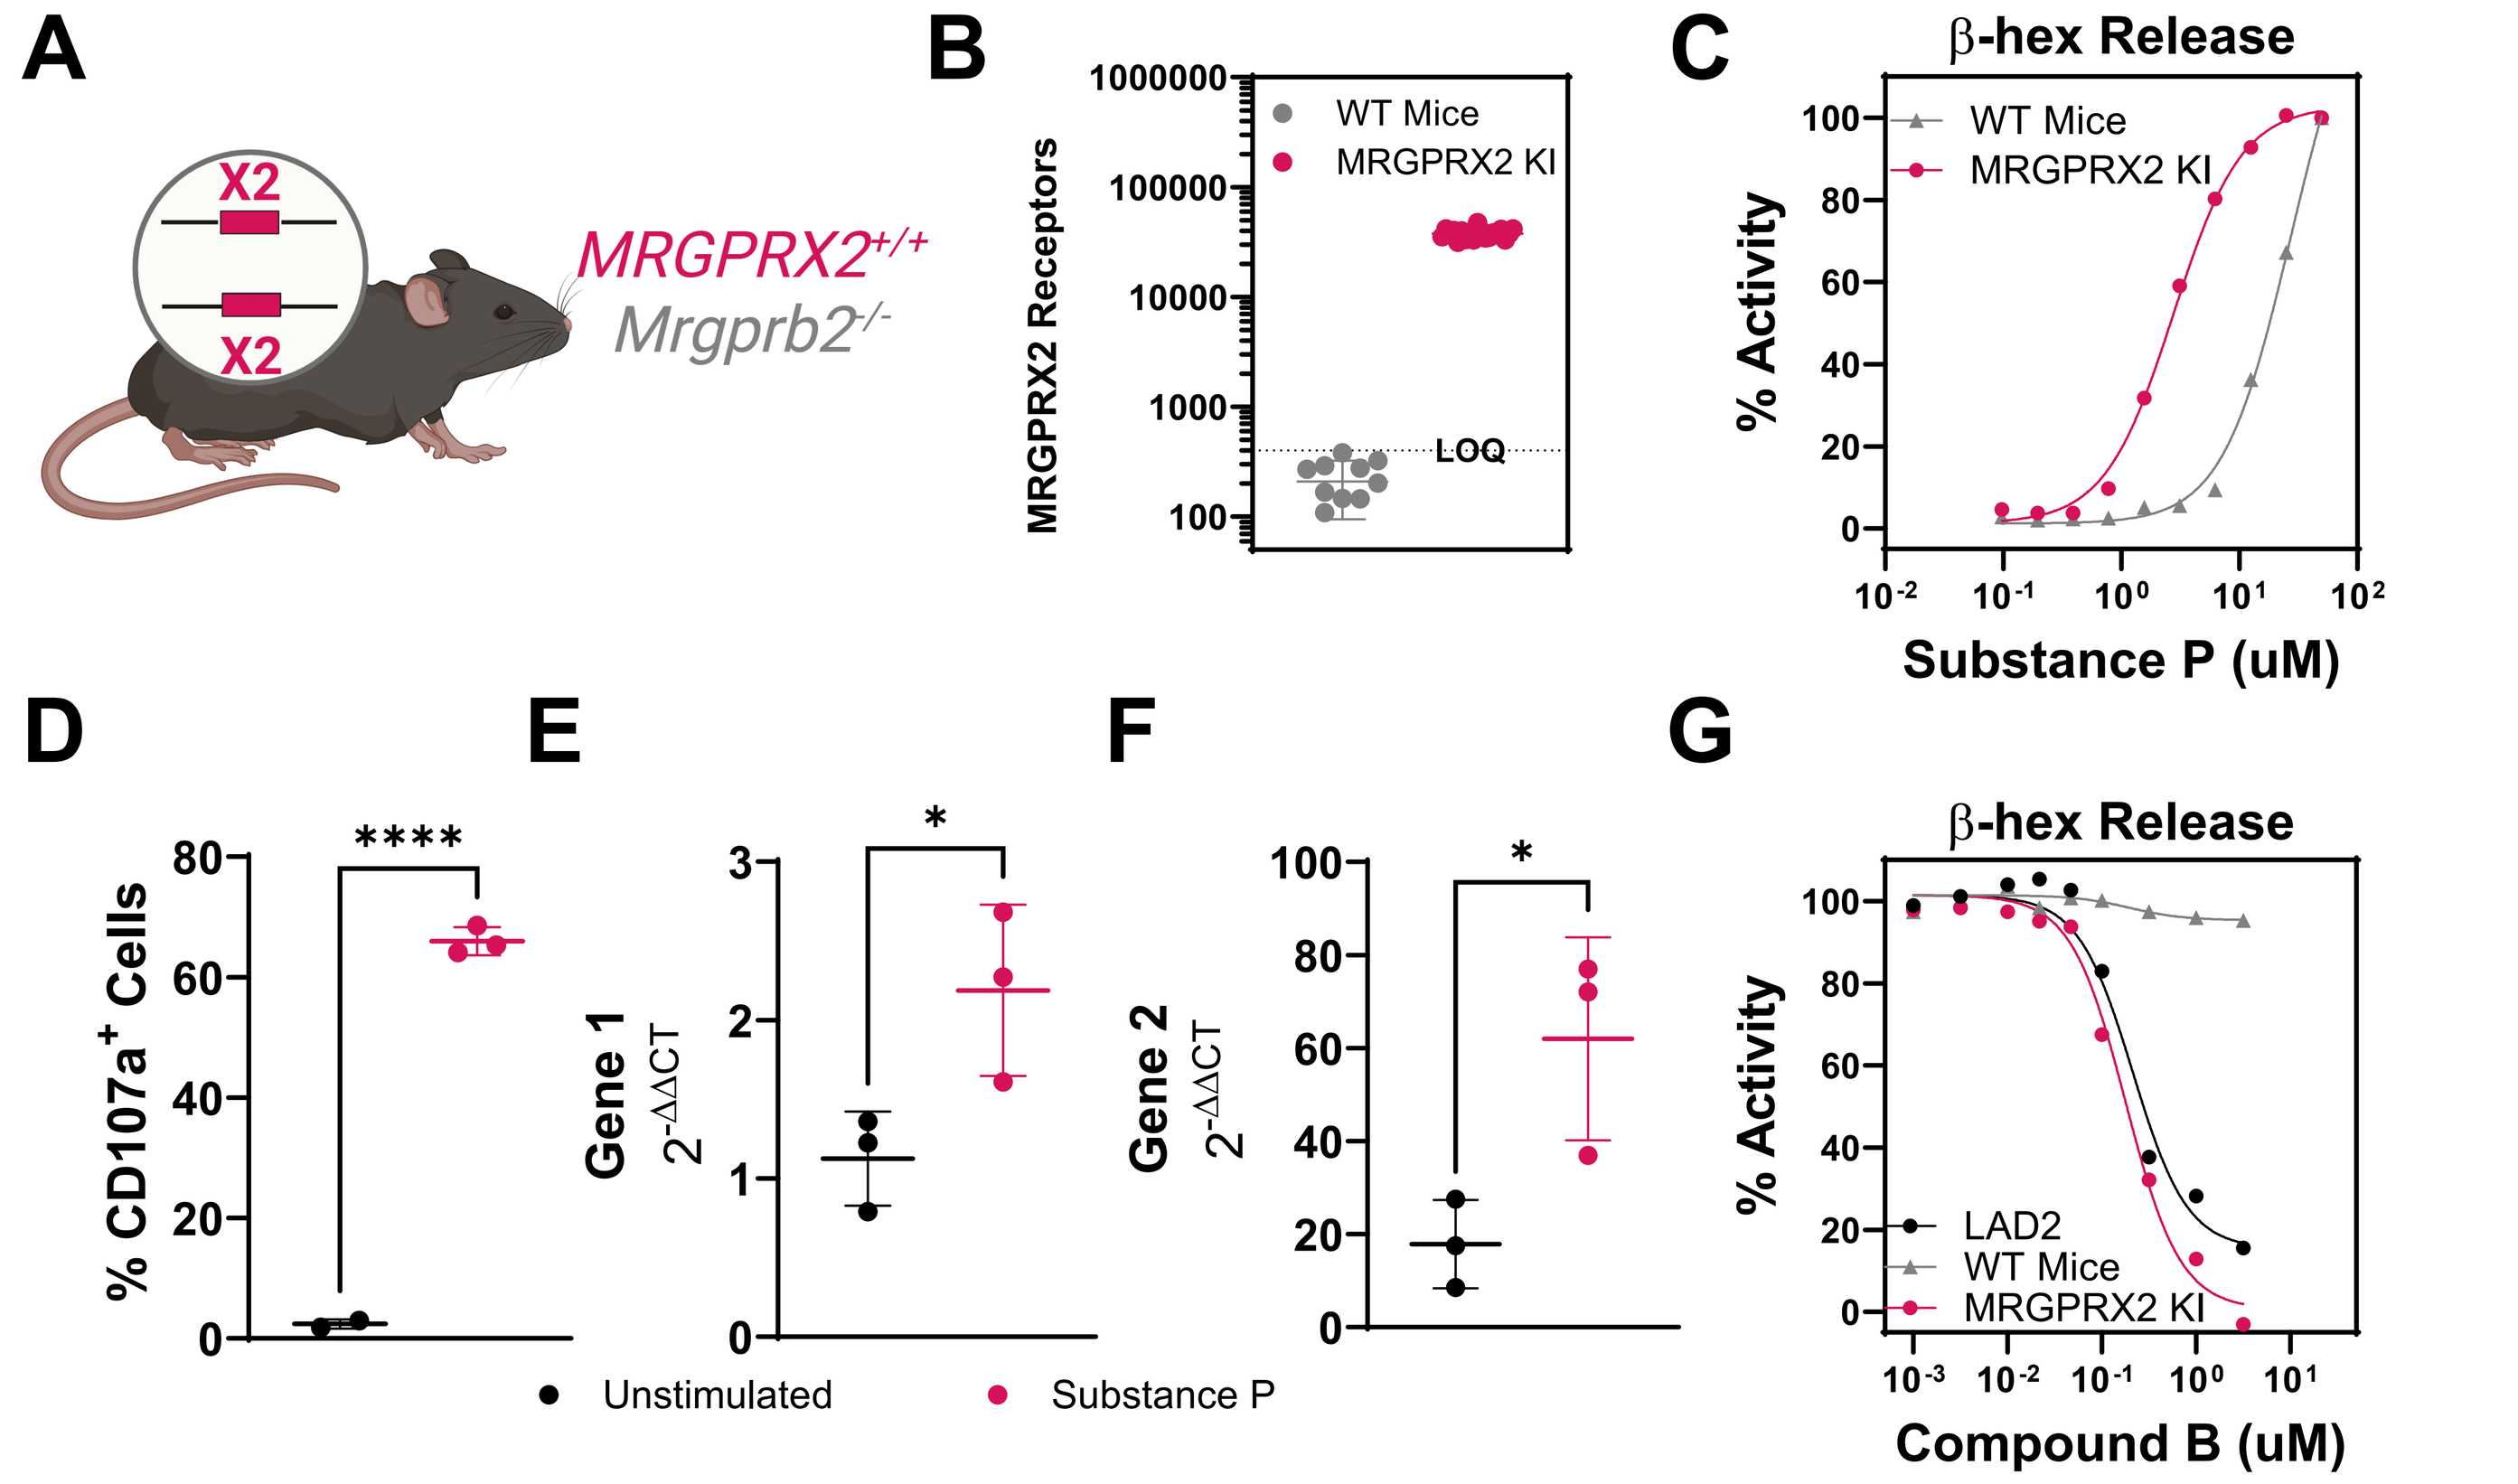
A
X2
X2
MRGPRX2+/+
Mrgprb2-/-
B
MRGPRX2 Receptors
WT Mice
MRGPRX2 KI
LOQ
C
β-hex Release
% Activity
WT Mice
MRGPRX2 KI
Substance P (uM)
D
% CD107a+ Cells

E
Gene 1
2-ΔΔCT
*
F
Gene 2
2-ΔΔCT
*
Unstimulated
Substance P
G
β-hex Release
LAD2
WT Mice
MRGPRX2 KI
Compound B (uM)

Figure 7 – MRGPRX2 KI mice respond to MRPGRX2 agonist and antagonist. (A) MRGPRX2 KI mice were generated by exchanging mouse *Mrgprb2* with human *MRGPRX2*. (B) MRGPRX2 expression as assessed by receptor density methodology on peritoneal lavage mast cells (PMC) from MRGPRX2 KI mice is shown in comparison to receptor levels on WT mice. LOQ, Limit of Quantification. (C) MRGPRX2 KI PMCs response to Substance P is left-shifted in comparison to PMC from WT mice. (D-F) MRGPRX2 KI PMCs upregulate mouse homologs of MRGPRX2 specific signature genes (defined in Figure 4) 24 hours after stimulation with Substance P (G) Arcus MRGPRX2 antagonist Compound B inhibits PMC degranulation triggered with published[1] mouse-specific EC_{50} of Substance P.

Results

MRGPRX2 Antagonist Inhibits Substance P-Induced Paw Swelling

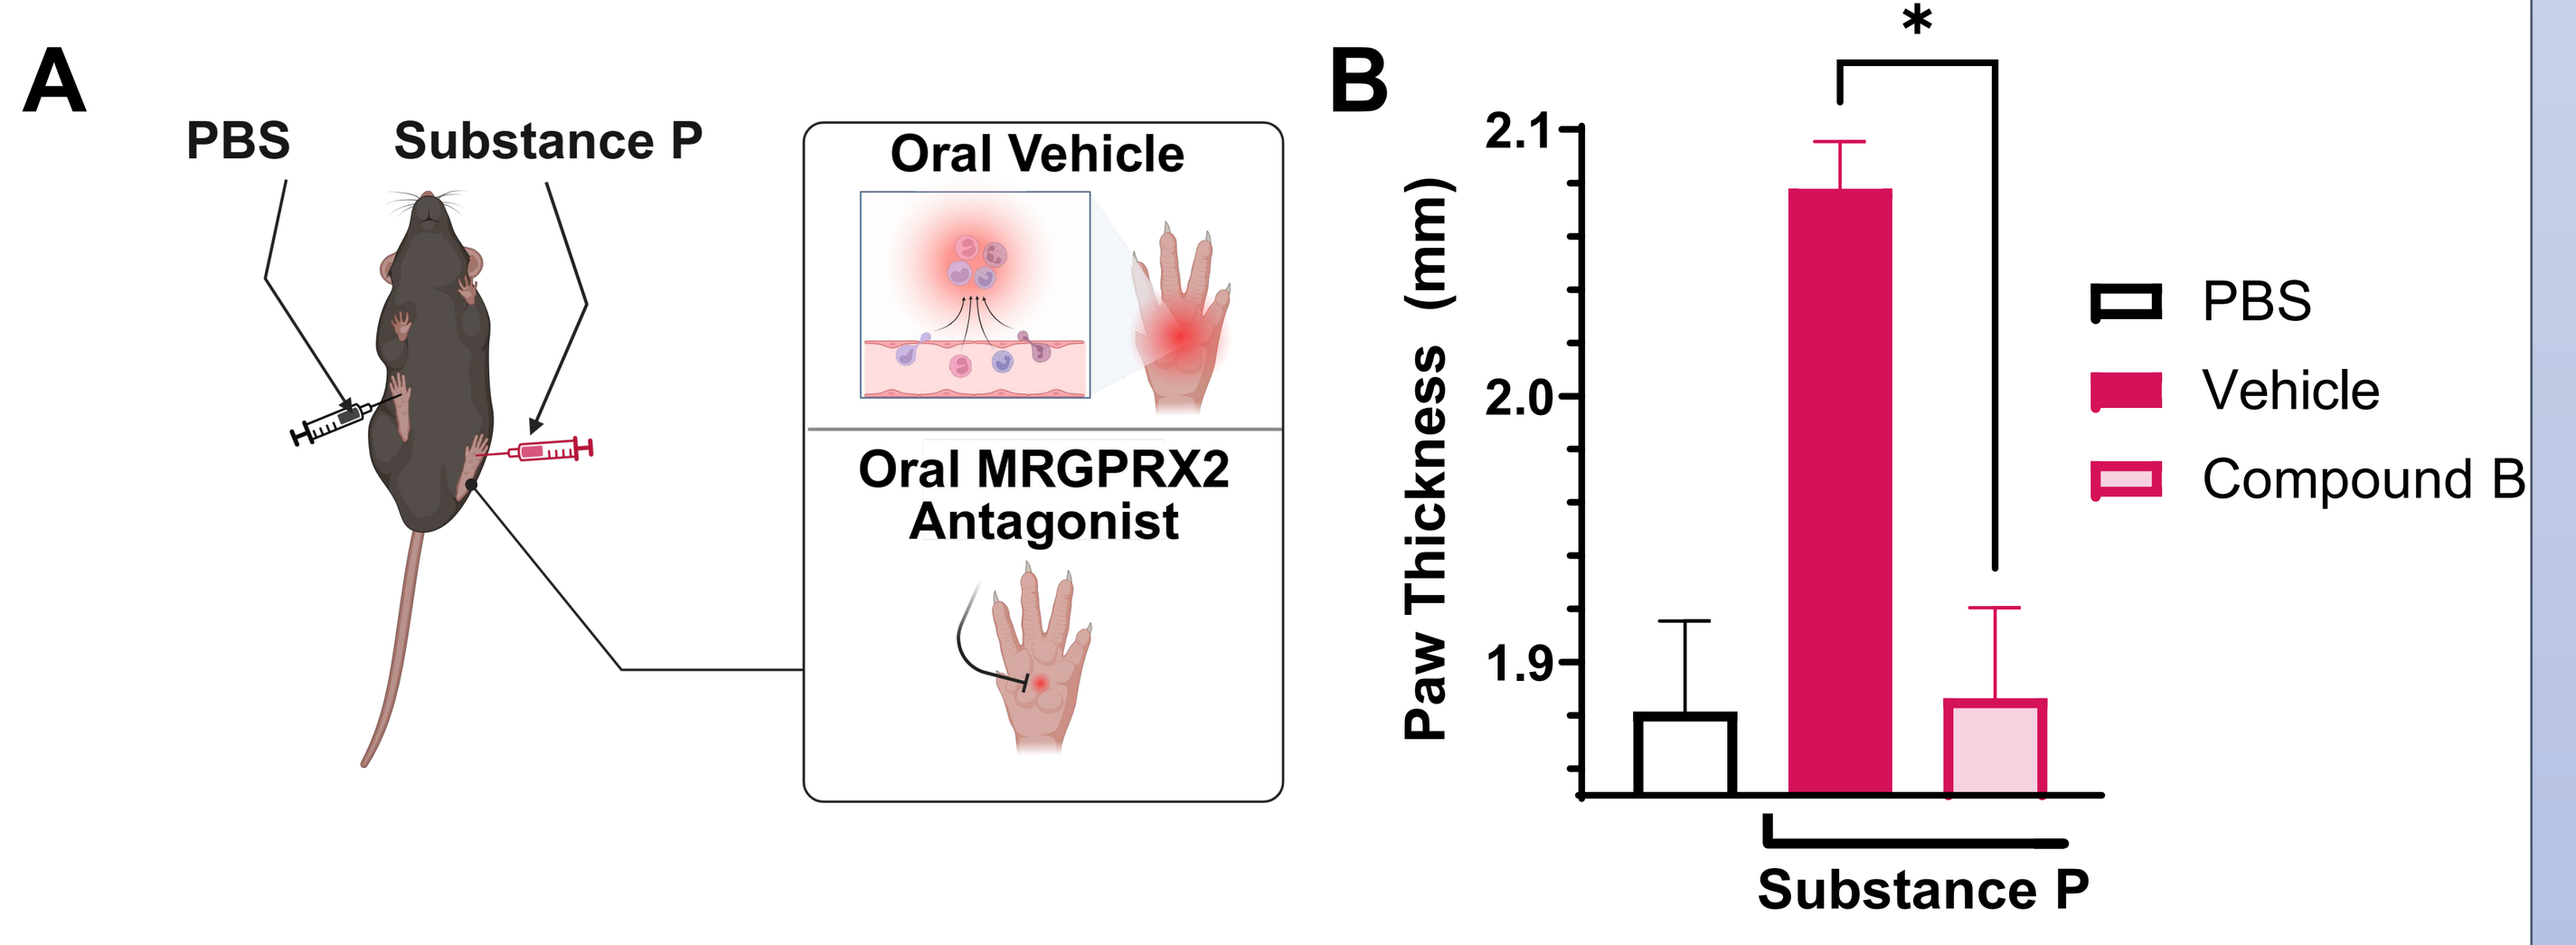
A
PBS
Substance P
Oral Vehicle
Oral MRGPRX2 Antagonist
B
*
Paw Thickness (mm)
PBS
Vehicle
Compound B
Substance P

Figure 8 –MRGPRX2 antagonist Compound B inhibits paw swelling induced with 25 uM Substance P in MRGPRX2 Knock-In mice. (A) Schematic outline of experiments is shown. MRGPRX2 KI mice were dosed orally with 30 mg/kg Arcus MRGPRX2 Antagonist 2 hours before Substance P challenge. (B) Measurements of paw thickness 30 minutes after injection with Substance P are shown, mean baseline prior to any injections is 1.72 mm. Paws from PBS-only injected mice serve as control. Dotted line represents baseline thickness of un-injected paws.

MRGPRX2+ Cells are Significantly Elevated in Atopic Dermatitis

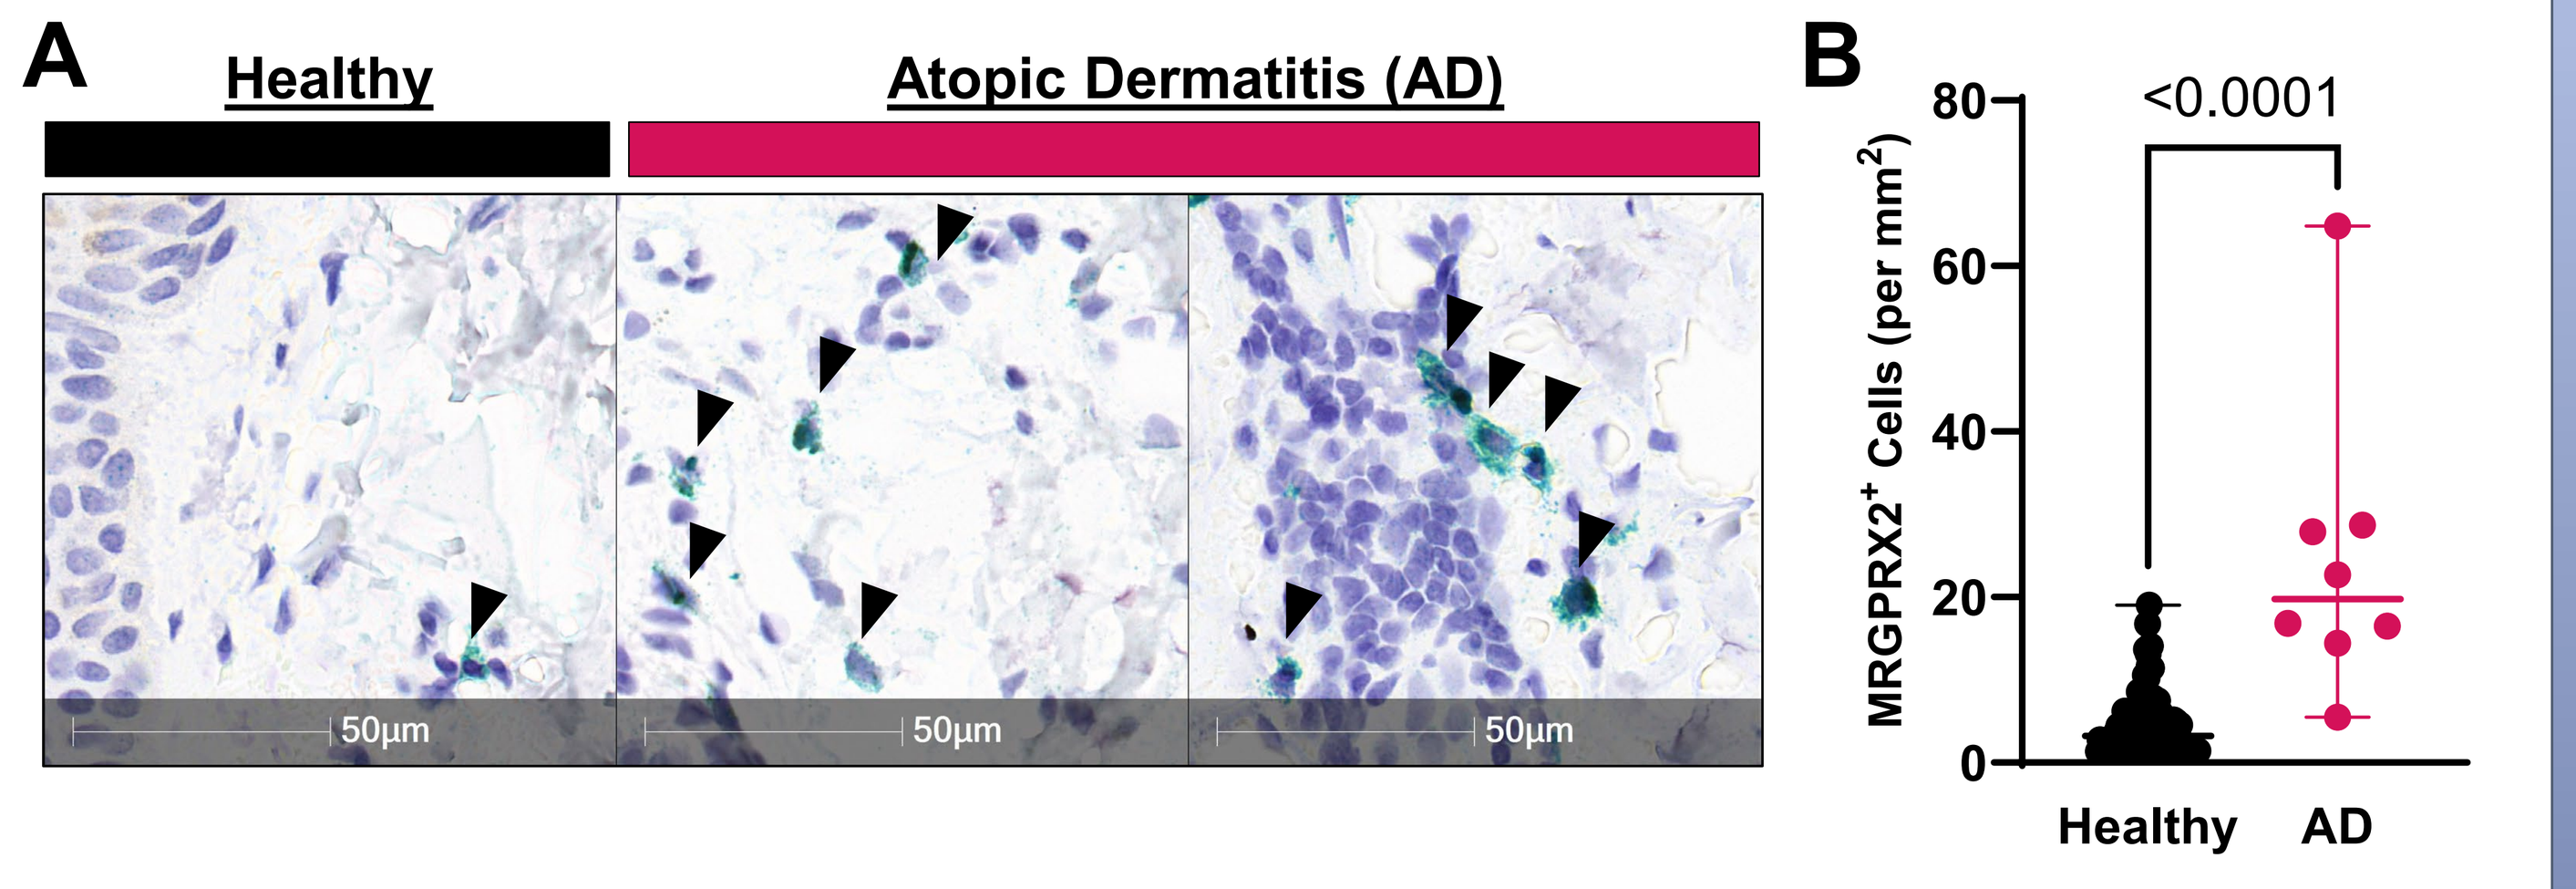
A
Healthy
Atopic Dermatitis (AD)
50μm
50μm
50μm
B
MRGPRX2+ Cells (per mm2)
<0.0001
Healthy
AD

Figure 9 – MRGPRX2 protein was stained on skin biopsies from healthy controls (n=50) or skin biopsies from patients with atopic dermatitis (AD). (A) Representative images taken with 40x magnification. MRGPRX2 is stained with a green chromogen (black triangles). Slides are counterstained with hematoxylin. (B) Quantification of MRGPRX2 positive cells per mm^2 in whole biopsies from healthy controls and AD patients was done with HALO image analysis software from Indica labs.

Summary

- ❖ MRGPRX2 can be activated by various additive agonists (Figures 1-3).
- ❖ MRGPRX2 stimulation combined with IgE receptor (IgER) crosslinking is synergistic, suggesting potentiation of mast cell pathology in atopic settings (Figure 3).
- ❖ MRGPRX2, complement, and IgER activation result in distinct gene expression signatures, suggesting unique biology (Figure 4).
- ❖ MRGPRX2-dependent mast cell degranulation can be inhibited in primary murine and human cells in multiple matrices (Figures 5-7).
- ❖ MRGPRX2 antagonist can fully inhibit Substance P-induced paw swelling in a wheal-and-flare model in MRGPRX2 KI mice. (Figure 8).
- ❖ Lesional skin from atopic dermatitis patients contains significantly more MRGPRX2+ mast cells (Figure 9).

In conclusion, MRGPRX2 is highly expressed and drives unique mast cell degranulation biology that is potentiated in atopic settings and can be fully inhibited with a novel and potent Arcus antagonist.

Some images made with BioRender. References: 1. Mc Neil BD et al. (2015) *Nature*; 2. Gour N et al. (2024) *Immunity*; 3. Thapaliya M et al. (2021) *Curr Allergy Asthma Rep.*; 4. Shtessel M et al. (2021) *J Invest Dermatol.*; 5. Metz M et al. (2014) *J Invest Dermatol.*; 6. Fujisawa D et al. (2014) *J Allergy Clin Immunol.*; 7. Jia T et al. (2024) *J Invest Dermatol.* LAD2 human mast cells were kindly provided by A. Kirshenbaum and D. Metcalfe (NIH, USA).